PO3-18-05

SAN ANTONIO BREAST CANCER SYMPOSIUM®
December 5-9, 2023

Pharmacokinetics (PK) of Lasofoxifene (LAS) Monotherapy and Combined with Abemaciclib (Abema)

Senthil Damodaran,[1] Paul V Plourde,[2] Simon N Jenkins,[2] Phil Mayer, David J Portman,[2] Matthew P Goetz[3]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Sermonix Pharmaceuticals, Columbus, OH; [3]Mayo Clinic, Rochester, MN

Introduction

- Lasofoxifene (LAS), a next-generation selective estrogen receptor (ER) modulator (SERM) and potent, breast ER antagonist, was shown to:
 - Reduce fracture risk in postmenopausal women with osteoporosis[1]
 - Improve vaginal atrophy in postmenopausal women[2]
 - Exhibit antitumor activity in phase 2 studies of women with (ER+)/HER2-, *ESR1*-mutated, metastatic breast cancer (mBC) that progressed on endocrine therapy (ET) and cyclin-dependent kinase 4/6 inhibitors (CDK4/6is)[3,4]
 - In the ELAINE 1 study, LAS alone prolonged progression-free survival (PFS) vs. fulvestrant (median ~5.6 vs 3.7 mos; *P*=0.138)[3]
 - In the ELAINE 2 study, LAS plus abemaciclib (Abema) was associated with a median PFS of ~13 months[4]
- Linear pharmacokinetics (PK) were seen with oral LAS following single (up to 100 mg) and multiple doses (up to 20 mg QD for 14d) in healthy volunteers; no clinically significant age or race differences were found[5,6]
 - LAS PK profiles between Caucasian and Japanese patients were similar[7,8]
- Here, we report select PK data of oral LAS (5 mg/day) from women with mBC and an *ESR1* mutation in the phase 2, ELAINE 1 and 2 studies

Objectives

To examine PK data of LAS 5 mg/day and investigate a possible drug-drug interaction between LAS and Abema

Methods

- Women with *ESR1*-mutated, ER+/HER2- mBC that progressed on endocrine therapy (ET) and CDK4/6 received oral LAS 5 mg/day as monotherapy (ELAINE 1) or combined with Abema (provided by Eli Lilly and Co) 150 mg BID (ELAINE 2)
- Blood was collected prior to daily dosing at baseline and weeks 2, 4, 8, and then every 4 weeks up to the final visit in ELAINE 1, and at baseline, weeks 2, 4, 6, and 8 in ELAINE 2
- LAS was assayed using validated high-performance liquid chromatography/tandem mass spectrometry (LC/MS/MS; linear range, 0.025 to 25.0 ng/mL); levels below the lower limit of quantification (LLOQ) were reported as 0 ng/mL
- Abema and its 2 major, active metabolites (M2 and M20) were measured by validated Turbo Ion Spray LC/MS/MS (linear range, 1.00 to 500 ng/mL); values below the LLOQ were reported as <LLOQ ng/mL
- Mean plasma steady-state minimum concentrations (C_{min}) at each visit were calculated for LAS (ELAINE 1 and 2), and Abema and its M2 and M20 metabolites (ELAINE 2)
- LAS levels from ELAINE 1 and 2 were directly compared, while Abema, M2 and M20 plasma levels in ELAINE 2 were compared with previously reported levels; the relationship of LAS concentrations with PFS was also explored

Results

Figure 1. Plasma concentrations of lasofoxifene over time in (A) ELAINE 1 and (B) ELAINE 2

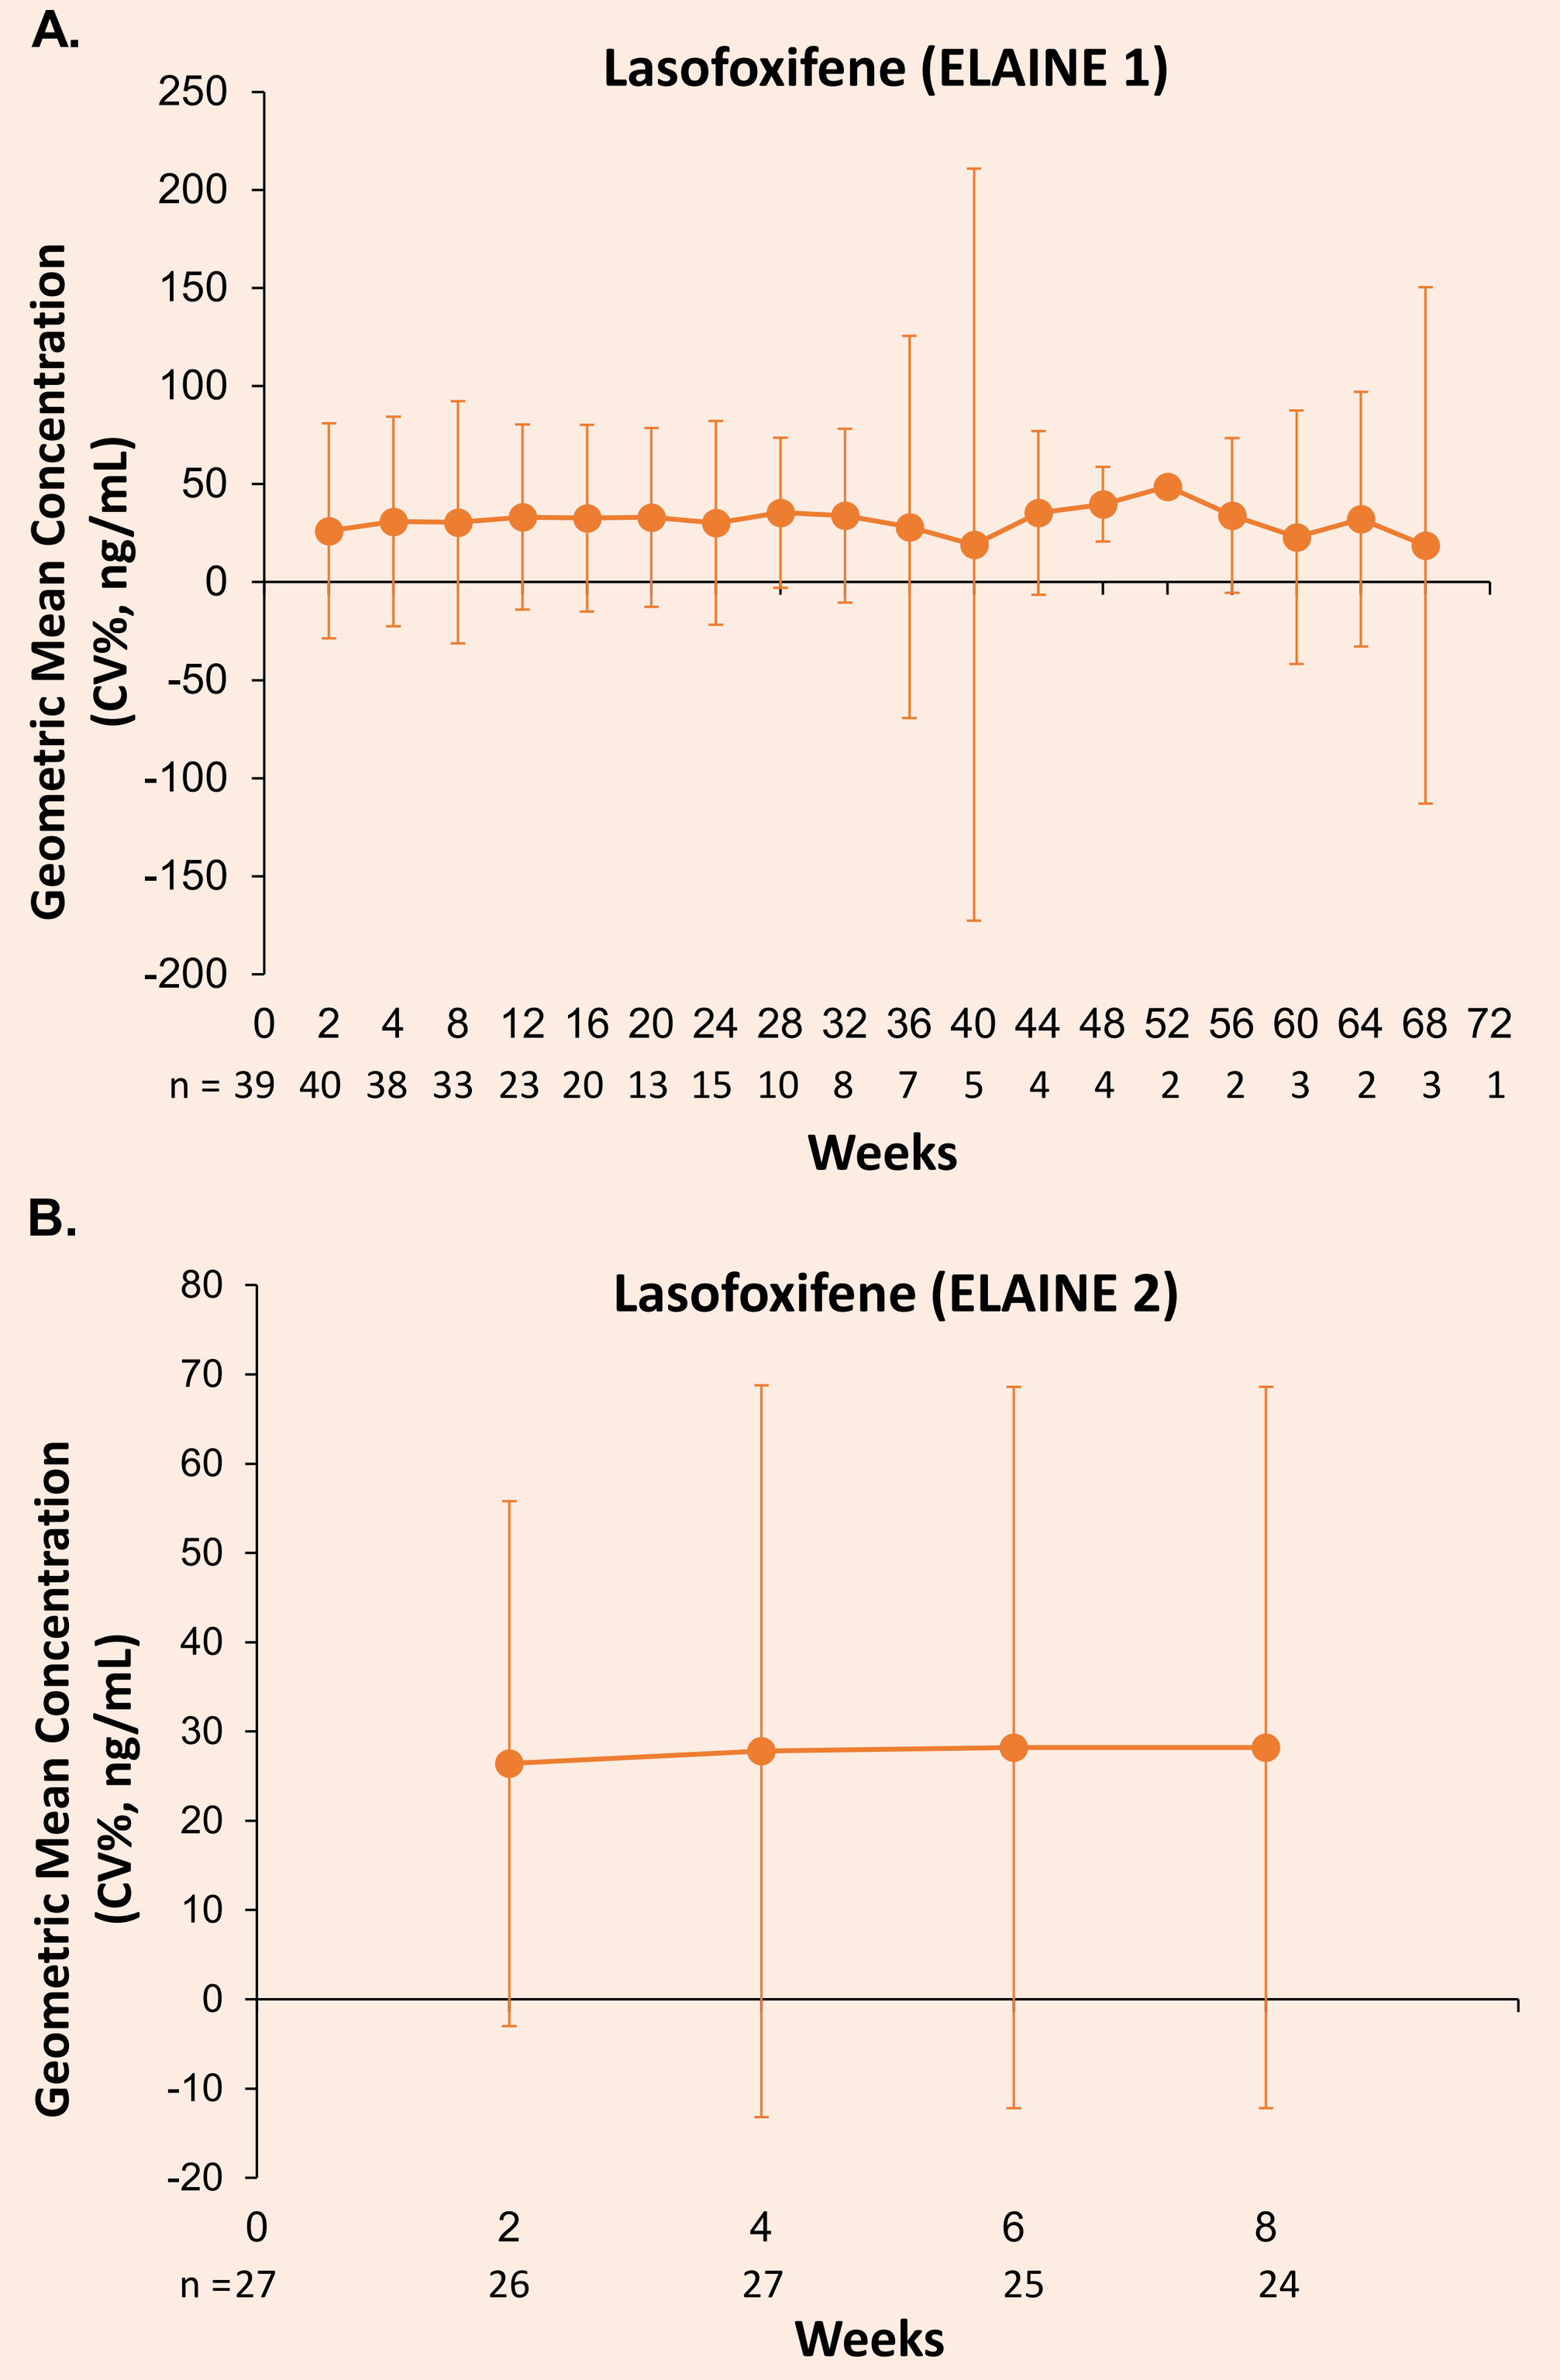
A.
Lasofoxifene (ELAINE 1)
Geometric Mean Concentration (CV%, ng/mL)
0 2 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72
n = 39 40 38 33 23 20 13 15 10 8 7 5 4 4 2 2 3 2 3 1
Weeks
B.
Lasofoxifene (ELAINE 2)
Geometric Mean Concentration (CV%, ng/mL)
0 2 4 6 8
n =27 26 27 25 24
Weeks

Table 1. Steady-state minimum concentrations (C_{min} [CV%]) for abemaciclib and M2 and M20 metabolites reported in the EMA EPAR[9] and in ELAINE 2

	C_{min} (CV%), ng/mL		
	Abemaciclib	M2	M20
European Medicines Agency[a]	143 (110)	62.6 (67)	113 (58)
ELAINE 2[b]	113.8 (179.3)	55.6 (91.5)	90.7 (119.7)

[a]European Medicines Agency Review of Pharmacokinetics for Abemaciclib (Verzenios).
[b]Subjects having no abemaciclib dose reduction; 4 subjects had a dose reduction.
C_{min}, steady-state minimum concentration; CV, coefficient of variation; EMA, European medicine agency; EPAR, European public assessment report.

Figure 2. Plasma concentrations of (A) abemaciclib, (B) M2 metabolite, and (C) M20 metabolite over time in ELAINE 2

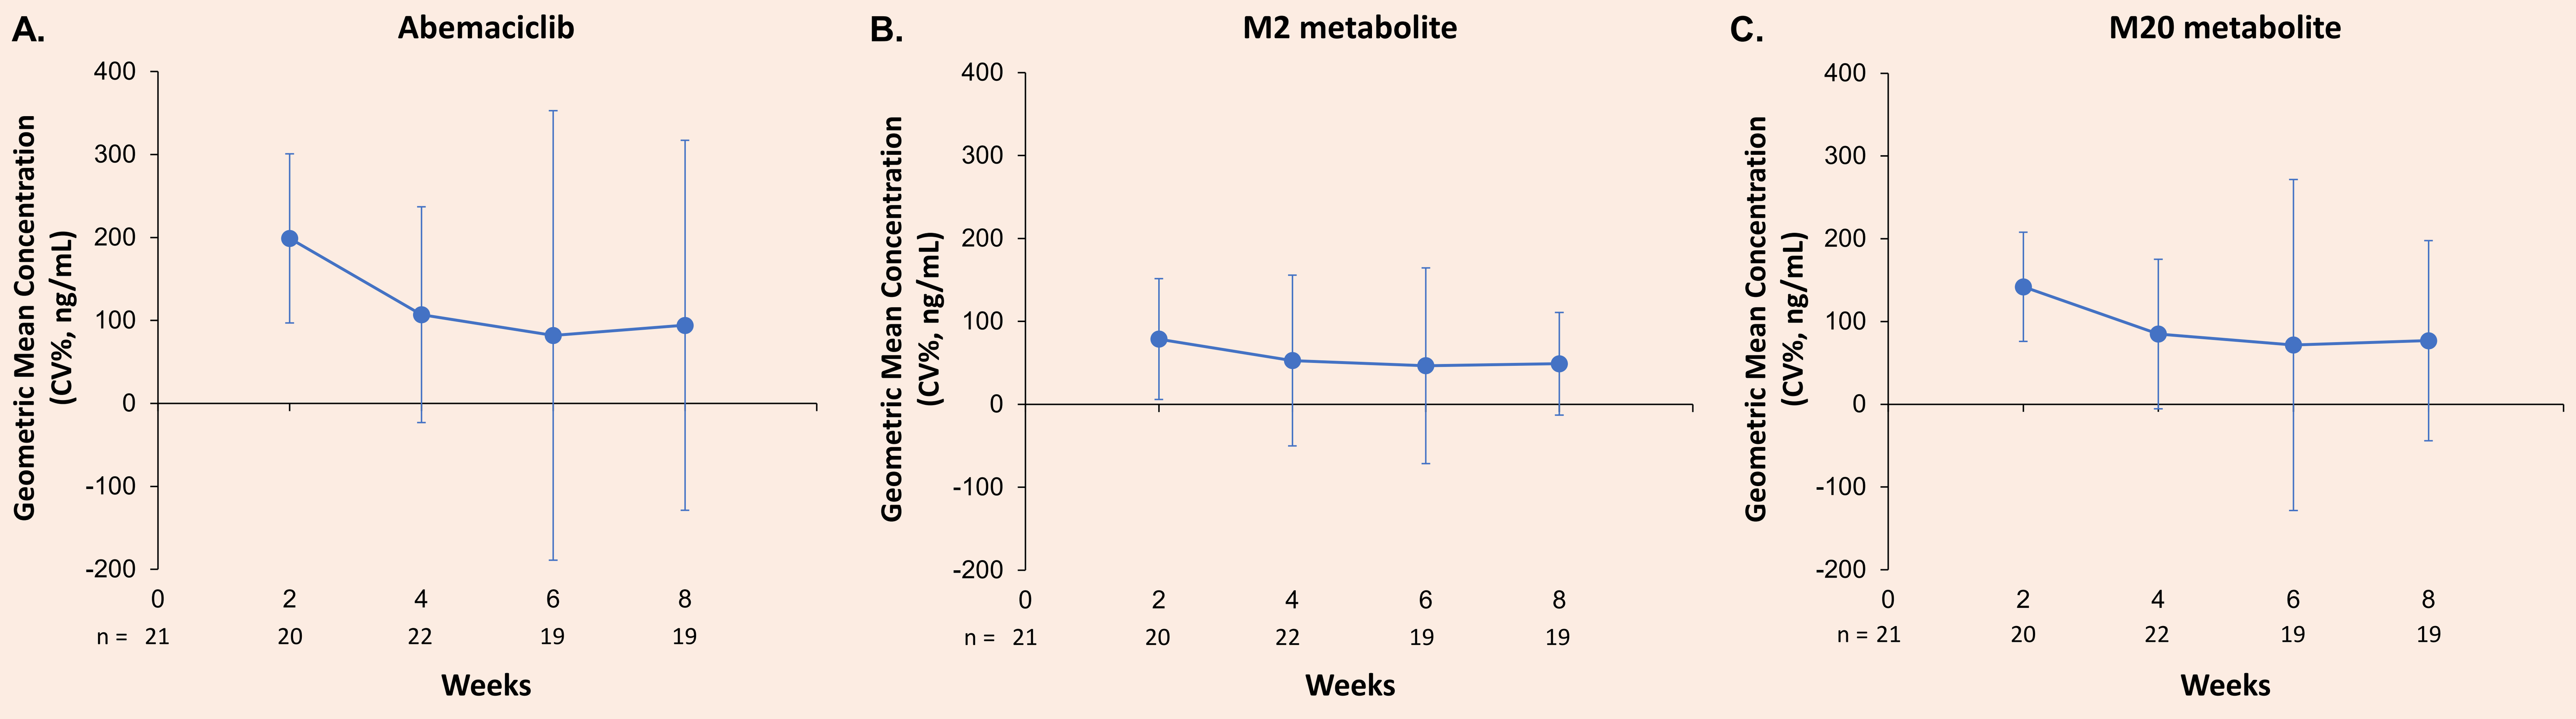
A.
Abemaciclib
B.
M2 metabolite
C.
M20 metabolite
Geometric Mean Concentration (CV%, ng/mL)
0 2 4 6 8
n = 21 20 22 19 19
Weeks

Data from patients who received oral LAS (5 mg/day) plus Abema (150 mg BID).

- Levels of LAS were similar between the ELAINE 1 and 2 studies and were consistent across visits with no evidence of longer-term or unexpected accumulation (**Figure 1**); across all time points for subjects who had PK samples assessed, the geometric mean C_{min} of LAS was 30.4 ng/mL (CV%, 56.9) in ELAINE 1 and 27.9 ng/mL (CV%, 36.1) in ELAINE 2
- Inter-subject variability was observed in plasma Abema concentrations in ELAINE 2; the mean concentrations of Abema and its M2 and M20 metabolites were comparable across all timepoints assessed among patients who did not have Abema dose reductions (**Figure 2**)
- The mean C_{min} for abemaciclib, M2 and M20 are similar to the C_{min} previously reported in the European public assessment report for Abema[9] (**Table 1**)
- Length of PFS was not related to LAS in the ELAINE 1 trial (shown descriptively in **Figure 3**) or LAS or Abema levels in the ELAINE 2 trial

Figure 3. Mean C_{min} of lasofoxifene across visits for each subject and PFS in ELAINE 1

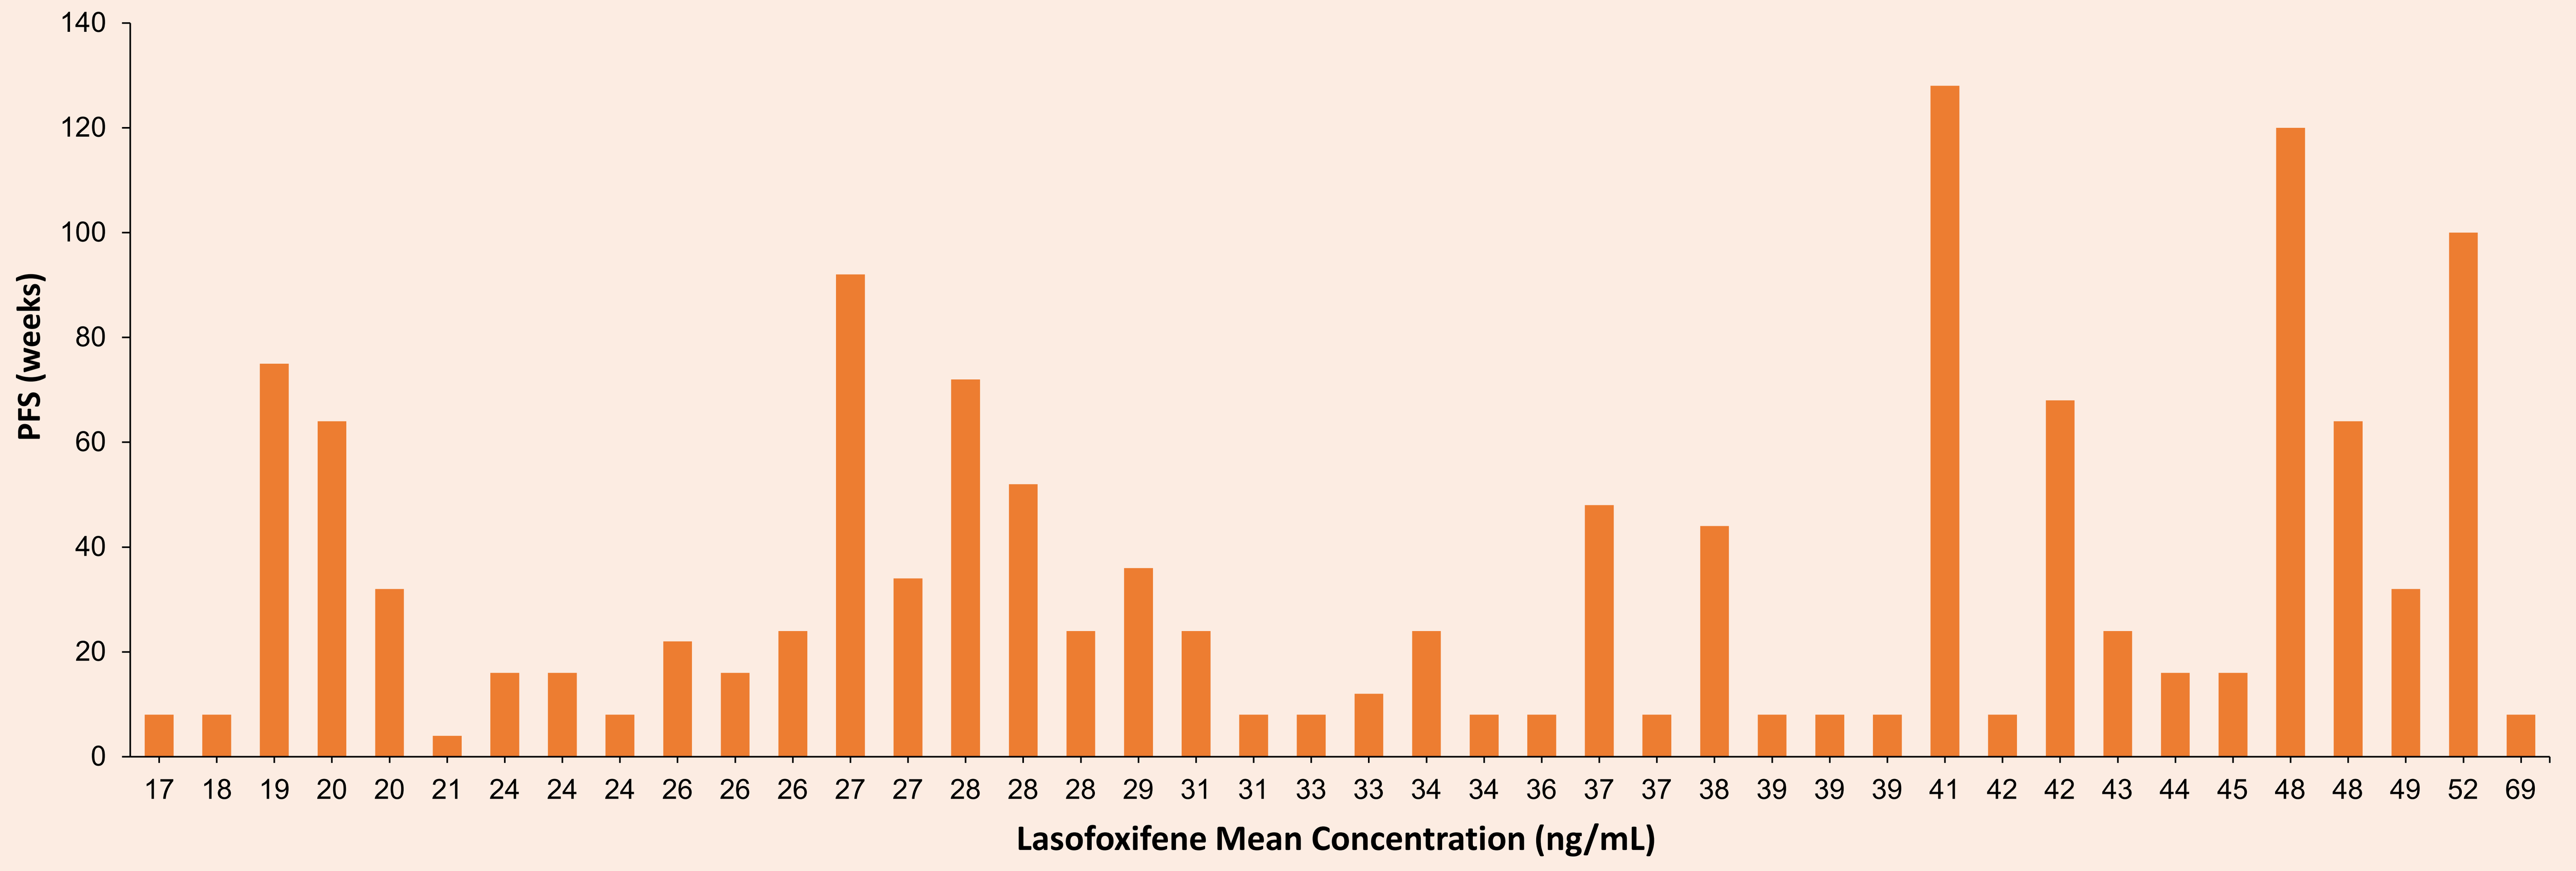
PFS (weeks)
17 18 19 20 20 21 24 24 24 26 26 26 27 27 28 28 28 29 31 31 33 33 34 34 36 37 37 38 39 39 39 41 42 42 43 44 45 48 48 49 52 69
Lasofoxifene Mean Concentration (ng/mL)

PFS, progression-free survival. Mean concentrations for LAS and Abema were calculated for each individual based on values obtained across patient visits.

Key Takeaways

- PK data for LAS 5 mg/day were consistent with previous clinical trials, resulting in similar steady-state concentrations in patients across several studies
- Addition of Abema to LAS did not appear to alter the PK of LAS in ELAINE 2 compared with monotherapy in ELAINE 1, suggesting little to no drug-drug interaction
- Abema concentrations in ELAINE 2 were consistent with previous monotherapy data, suggesting no impact of LAS on Abema PK

Conclusions

- The C_{min} for LAS from both ELAINE studies was consistent with that predicted for 5 mg/day LAS from the non-oncologic program (26 ng/mL) and an ethno-bridging study (Japanese subjects: 30 ng/mL, Caucasians: 33 ng/mL),[6] suggesting no significant effect of Abema on the PK of LAS
- Abema PK data from ELAINE 2 were consistent with published data;[9] little to no interactions between LAS and the metabolism of Abema or its M2 and M20 metabolites were observed
 - This is not unexpected, as lasofoxifene does not have a significant inhibitory or induction effect on CYP3A4,[6] the major metabolic pathway of abemaciclib[10]
- These results support the continued development of LAS alone and combined with Abema for treating ER+/HER2-, *ESR1*-mutated mBC that progressed on ET and CDK4/6is

References

1. Cummings SR, et al. *N Engl J Med*. 2010;362:686-696. **2.** Portman DJ, et al. *Menopause*. 2015;22:1385-1386. **3.** Goetz MP, et al. *Ann Oncol*. 2023;In Press. **4.** Damodaran S, et al. *Ann Oncol*. 2023;In Press. **5.** Gardner M, et al. *J Clin Pharmacol*. 2006;46:52-58. **6.** Data on file. Sermonix Pharmaceuticals. **7.** Fountaine RJ, et al. *J Clin Pharmacol*. 2006;46:693-699. **8.** Jhee S, et al. 12th AACR-JCA Joint Conference: Breakthroughs in Cancer Research. 2022:Abstract A83. **9.** Assessment report: Verzenios (International non-proprietary name: abemaciclib). European Medicines Agency. Procedure No. EMEA/H/C/004302/II/0013. 2022. **10.** Verzenios (abemaciclib) 50, 100, 150 mg film-coated tablets Summary of Product Characteristics. Eli Lilly and Company. Indianapolis, IN.

Disclosures

- **SD** has received research funding from EMD Serono, Guardant Health, Taiho Pharmaceuticals, Novartis, and Sermonix. **MPG** has CME activities from Research to Practice, Clinical Education Alliance, and Medscape; was a panelist for Total Health Conferencing and a moderator for Curio Science; has done consulting for AstraZeneca, Biovica, Biotheranostics, Blueprint Medicines, Eagle, Eli Lilly, Novartis, Pfizer, Sanofi Genzyme, and Sermonix; and has received research support from Eli Lilly, Pfizer, and Sermonix. **PM** and **SNJ** are consultants for Sermonix. **DJP** and **PVP** are employees and stockholders of Sermonix.
- Sermonix Pharmaceuticals sponsored the study and provided support for the medical writing assistance of Hui Zhang, PhD and Kathleen Ohleth, PhD (Precise Publications, LLC). Eli Lilly and Company provided abemaciclib for the study.

This presentation is the intellectual property of Sermonix Pharmaceuticals. Contact David J Portman at dportman@sermonixpharma.com for permission to reprint and/or distribute.