PO2-14-09

SAN ANTONIO BREAST CANCER SYMPOSIUM®

December 5-9, 2023

Baseline Genomic Alterations and the Activity of Lasofoxifene (LAS) Plus Abemaciclib (Abema) in Patients with ER+/HER2- Metastatic Breast Cancer (mBC): The ELAINE 2 Study

Senthil Damodaran,[1] Massimo A Cristofanilli,[2] Matthew P Goetz,[3] Ciara C O'Sullivan,[3] Gary Riordan,[4] Hope S Rugo,[5] Daniel G Stover,[6] Seth A Wander,[7] Dominic Carroll,[8] Paul V Plourde,[8] David J Portman,[8] and Einav Gal-Yam[9]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Weill Cornell Medicine, New York, NY; [3]Mayo Clinic, Rochester, MN; [4]Reg Pro1, LLC, Tuscon, AZ; [5]University of California San Francisco, San Francisco, CA; [6]The Ohio State University, Columbus, OH; [7]Massachusetts General Hospital, Boston, MA; [8]Sermonix Pharmaceuticals, Columbus, OH; [9]Sheba Medical Center, Ramat Gan, Israel

Introduction

- Estrogen receptor-positive (ER+)/HER2-negative (HER2-) breast cancers treated with endocrine therapy (ET) often acquire constitutively active mutations in the ERα-encoding gene (*ESR1*), resulting in ET resistance[1,2]
- Alterations or expression changes in other genes (eg, *RB1*, *FGFR1*, *CCND1*, *ERBB2*, *TP53*, *CCNE1*) are also implicated in ET or cyclin-dependent kinase 4/6 inhibitor (CDK4/6i) resistance[3-5]
- Novel therapies are needed for *ESR1*-mutated, metastatic breast cancer (mBC), especially after progression on a CDK4/6i
- Lasofoxifene (LAS), a next-generation selective ER modulator (SERM) and potent, breast ER antagonist, prolonged progression-free survival (PFS) vs fulvestrant in the phase 2, ELAINE 1 study in women with *ESR1*-mutated, ER+/HER2- mBC who had disease progression on an aromatase inhibitor plus a CDK4/6i (median PFS 5.6 vs 3.7 months; P=0.138)[6]
- In the single-arm, phase 2, ELAINE 2 study, LAS combined with abemaciclib (Abema) resulted in a median PFS of 56.0 weeks (95% CI, 31.9 weeks–NE; ~13 months), clinical benefit rate (CBR) of 65.5%, and objective response rate of 56% in women with *ESR1*-mutated, ER+/HER2- mBC that progressed on ET and CDK4/6is[7]
 - Changes in *ESR1* mutant allele fraction (MAF) appear predictive of response in ELAINE 2; the positive predictive value (PPV) for clinical benefit after 4 weeks of treatment was 81% when *ESR1* MAF decreased and 93% when cleared[8]

Objective

To describe baseline genomic alterations co-occurring with *ESR1* mutations in the ELAINE 2 study and treatment responses to LAS plus Abema in patients with these genomic co-alterations

Methods

- In ELAINE 2 (NCT04432454), women (age ≥18 yrs) with ER+/HER2- mBC that progressed after prior ET and CDK4/6i and an *ESR1* mutation detected using the Sysmex-Inostics SafeSeq circulating tumor DNA (ctDNA) test received oral LAS 5 mg/day and Abema (provided by Eli Lilly and Co) 150 mg BID until disease progression, death, unacceptable toxicity, or withdrawal
- Baseline genomic alterations in ctDNA (including *ESR1* mutations) were identified with the Guardant360 CDx test
- Median PFS and CBR were assessed in patients with *ESR1* mutations and co-existing baseline genomic alterations
 - For patients who withdrew before disease progression, their last observation time was used as the progression time for PFS estimation
- Data were summarized descriptively with no formal hypothesis testing

Results

- In 26 of the 29 patients (median age 60 yrs) enrolled, *ESR1* mutations were identified at baseline by the Guardant360 CDx test; all 26 (100%) received prior CDK4/6i, 20 (76.9%) prior fulvestrant, 2 (7.7%) prior PIK3CA inhibitor, and 12 (46.2%) prior chemotherapy for mBC (**Figure 1**)
 - In the biomarker subset (n=26), the median PFS and CBR of LAS plus Abema were 12.9 mos and 73.1%, respectively, similar to the overall parent trial (12.9 mos and 65.5%) (**Table**)
- Alterations in 39 genes co-occurred with *ESR1* mutations at baseline
 - *TP53* mutations were identified in 11 (42.3%) patients, *PIK3CA* mutations in 8 (30.8%), *CCND1* amplifications in 6 (23.1%), and *FGFR1* amplifications in 5 (19.2%); 3 (11.5%) patients had both *CCND1* and *FGFR1* amplifications (**Figure 2**; **Table**)
- In patients who had co-occurring *TP53* mutations, the CBR was 64% and median PFS was 8.3 mos; corresponding values for patients with co-occurring *PIK3CA* mutations were 63% and 7.8 mos, respectively (**Table**)
- All patients in the *CCND1* and *FGFR1* amplification subgroups achieved clinical benefit, with a median PFS of 16.6 mos in both subgroups (**Table**)
- For patients with co-alterations in ≥2 other genes of interest, the CBR was 83.3% and median PFS was 14.7 mos (**Table**)

Table. Treatment response by genomic alterations of interest that were concurrently detected with *ESR1* mutations at baseline (n=26)

Genomic alteration	Baseline n (%)	mPFS,[a] mos	CBR, %
ESR1 mutation	26 (100)	12.9	73.1
+ *TP53* mutation	11 (42.3)	8.3	63.6
+ *PIK3CA* mutation	8 (30.8)	7.8	62.5
+ *CCND1* amplification	6 (23.1)	16.6	100
+ *FGFR1* amplification	5 (19.2)	16.6	100
+ *ERBB2* mutation	3 (11.5)	12.9	66.7
+ *CCNE1* amplification or mutation	3 (11.5)	23.0	100
+ *RB1* mutation	1 (3.8)	11.0	100
+ *CCND1* and *FGFR1* amplification	3 (11.5)	7.6	100
+ ≥2 co-alterations in other genes of interest	12 (46.2)	13.8	83.3

[a]For patients who withdrew before disease progression, the last observation time was used as the progression time for PFS estimation.
[b]Other genes of interest include *PIK3CA*, *FGFR1*, *CCND1*, *TP53*, *ERBB2*, *CCNE1*, or *RB1*.
CBR, clinical benefit rate; mPFS, median progression-free survival.

Figure 1. Time on treatment and response to treatment in ELAINE 2

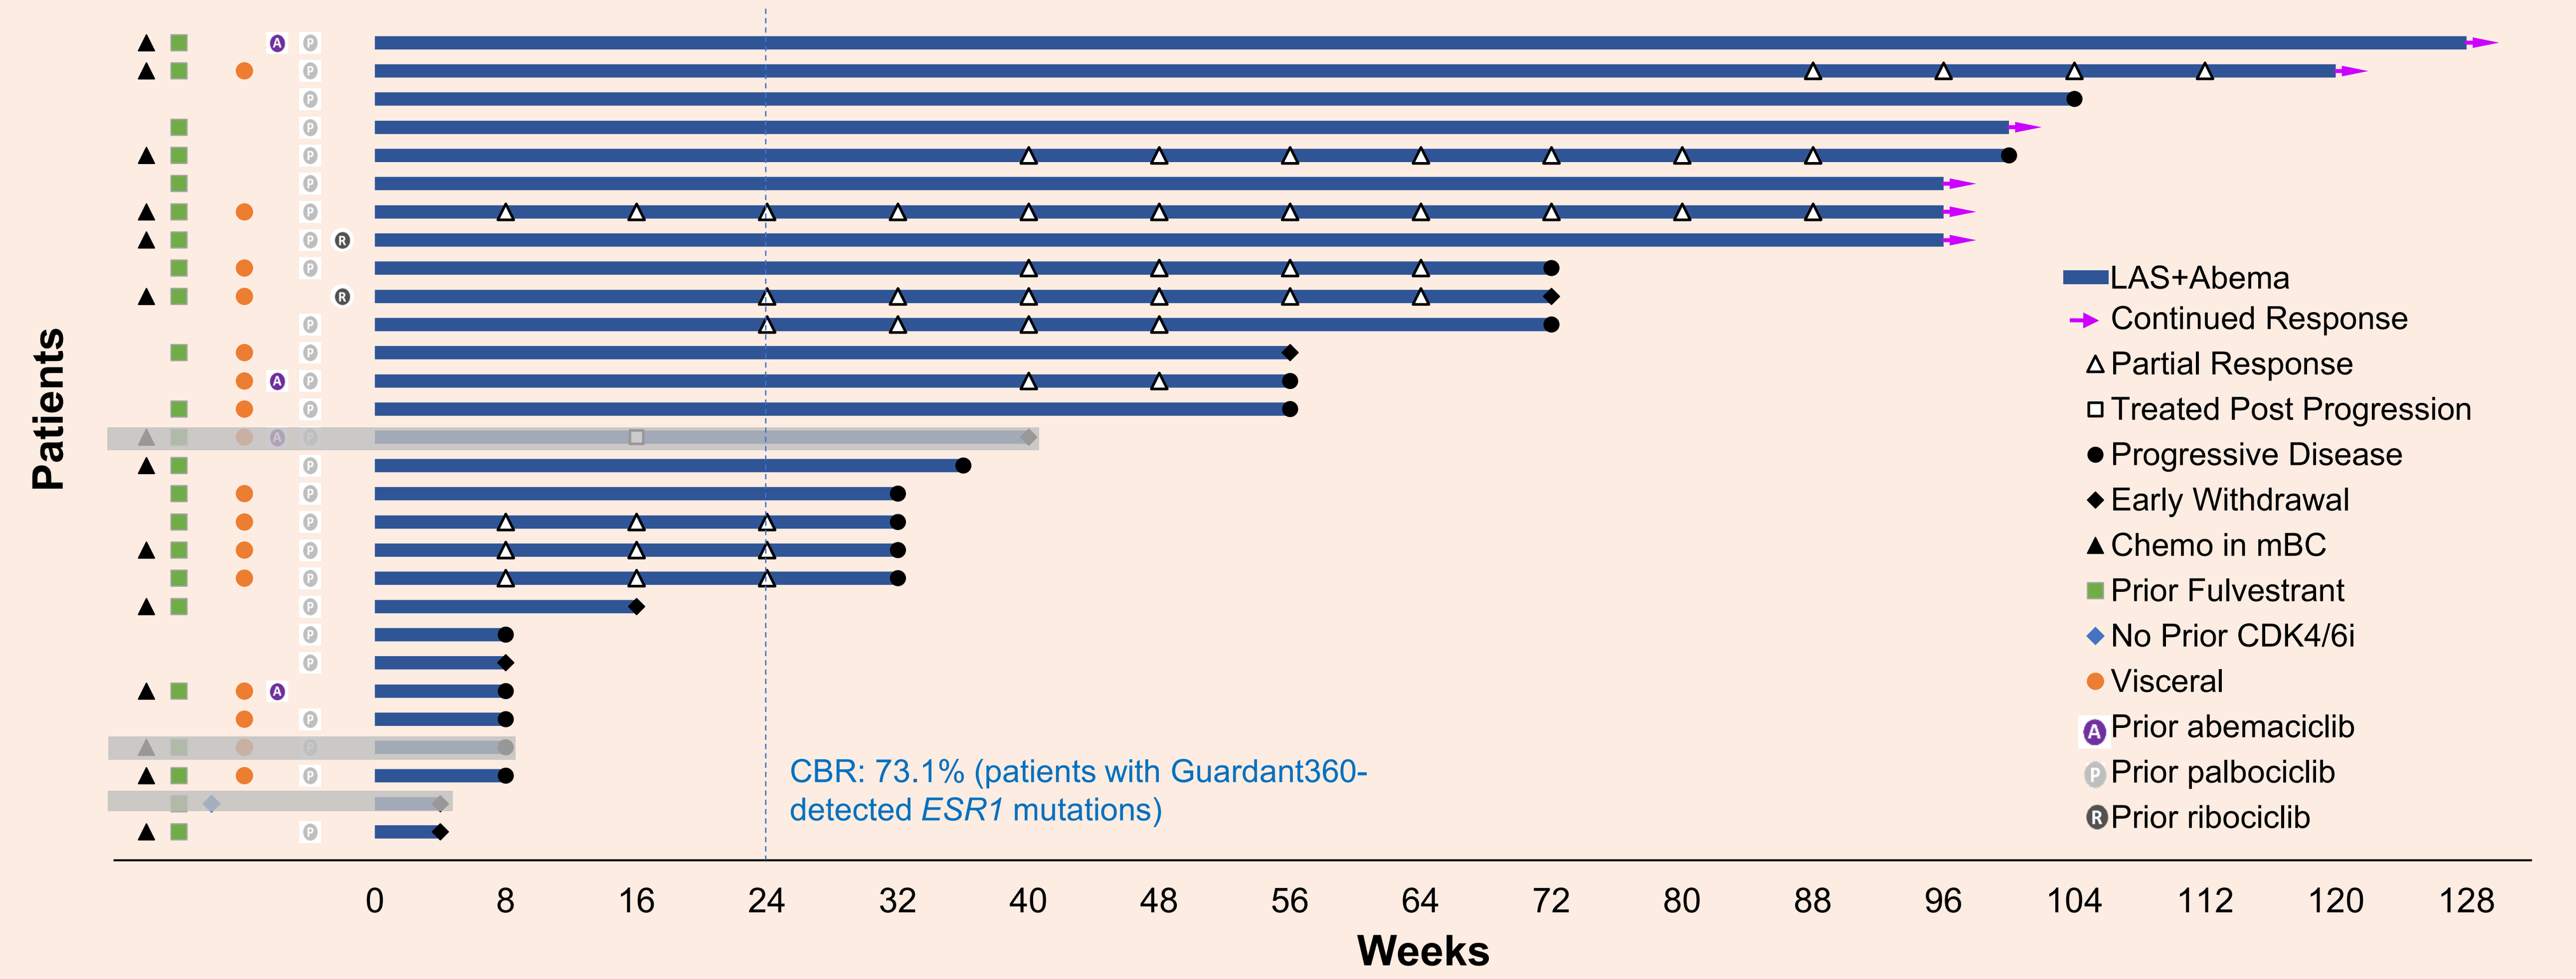
Patients
Weeks
0
8
16
24
32
40
48
56
64
72
80
88
96
104
112
120
128
CBR: 73.1% (patients with Guardant360-detected *ESR1* mutations)
LAS+Abema
Continued Response
Partial Response
Treated Post Progression
Progressive Disease
Early Withdrawal
Chemo in mBC
Prior Fulvestrant
No Prior CDK4/6i
Visceral
Prior abemaciclib
Prior palbociclib
Prior ribociclib

Data as of April 2023. Bars shaded in gray represent patients with *ESR1* mutations detected in the Sysmex-Inostics SafeSeq ctDNA test but not in the Guardant360 CDx test, who were excluded from the analysis of treatment response with genomic co-alterations.
Abema, abemaciclib; CBR, clinical benefit rate; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; LAS, lasofoxifene.

Figure 2. Distribution of genomic alterations detected by the Guardant360 CDx test

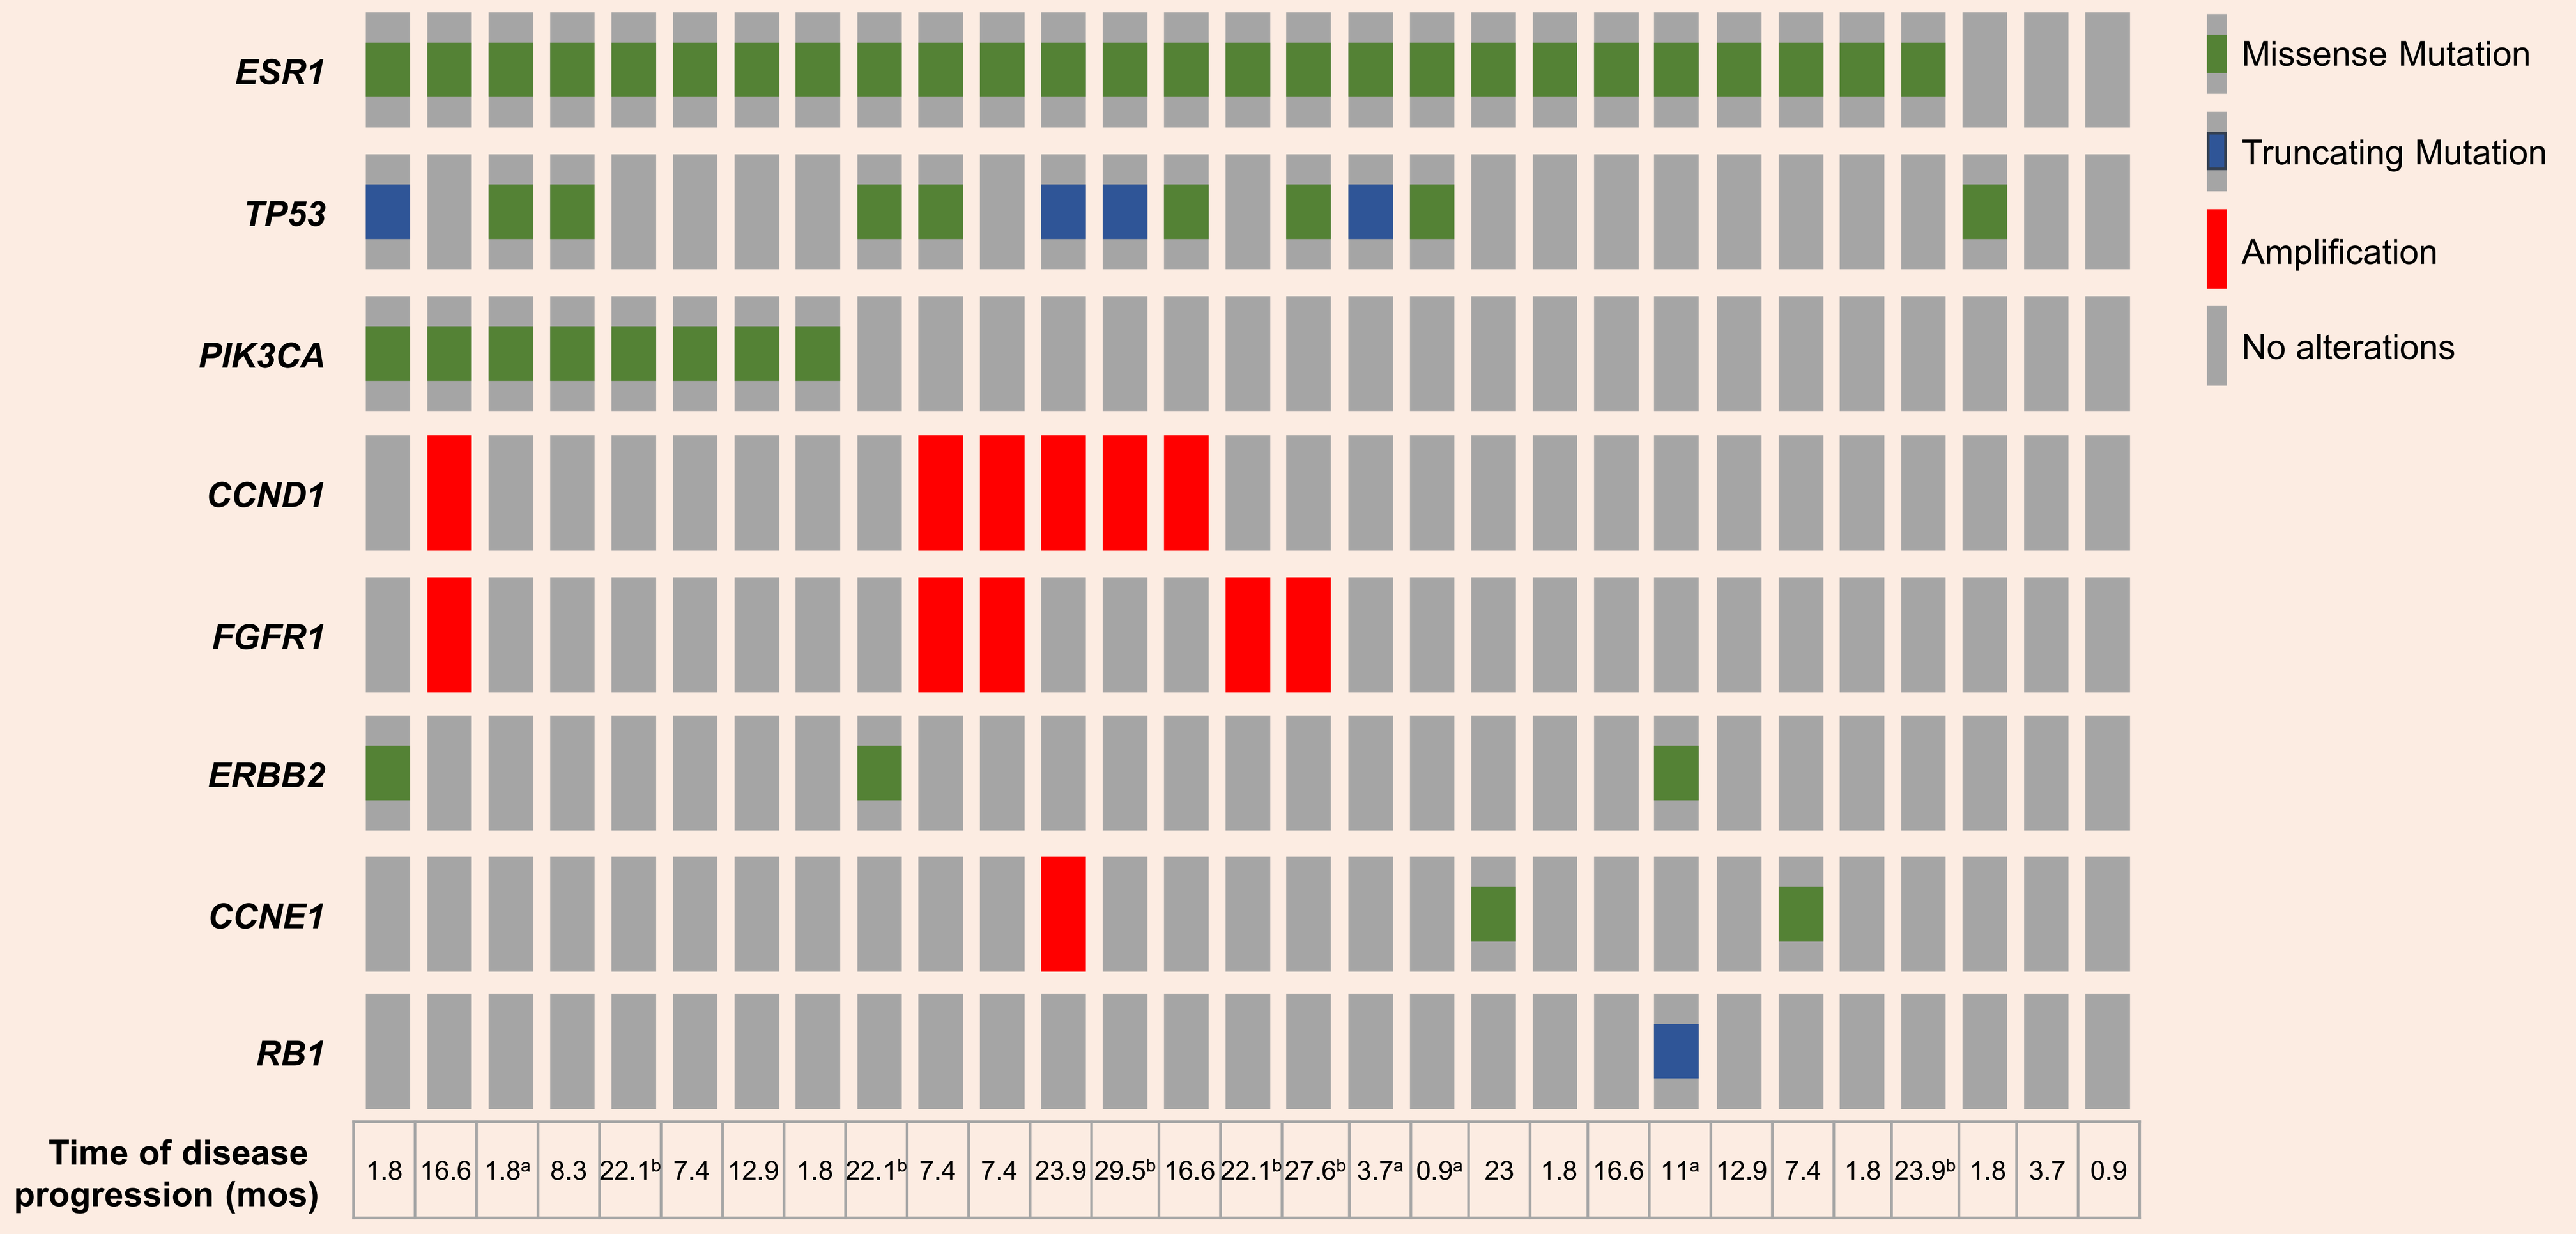
ESR1
TP53
PIK3CA
CCND1
FGFR1
ERBB2
CCNE1
RB1
Time of disease progression (mos)
1.8
16.6
1.8[a]
8.3
22.1[b]
7.4
12.9
1.8
22.1[b]
7.4
7.4
23.9
29.5[b]
16.6
22.1[b]
27.6[b]
3.7[a]
0.9[a]
23
1.8
16.6
11[a]
12.9
7.4
1.8
23.9[b]
1.8
3.7
0.9
Missense Mutation
Truncating Mutation
Amplification
No alterations

Three patients with *ESR1* mutations found by Sysmex-Inostics SafeSeq but not Guardant360 were excluded from the treatment response with gene co-alteration analysis.
[a]Early withdrawal, time of last assessment or withdrawal. [b]Continued response, time of last assessment.

Key Takeaways

- Genomic alterations concurrent with *ESR1* mutations were frequent in mBC that has progressed on ET and CDK4/6is
- Co-alterations in *ESR1* and other genes associated with treatment resistance did not appear to compromise the efficacy of LAS plus Abema in ELAINE 2

Conclusions

- Guardant360 ctDNA profiling from patients in ELAINE 2 demonstrates that other genomic alterations are frequently detected concurrently with *ESR1* mutations in the endocrine-resistant setting, in line with previous findings among patients with hormone receptor-positive mBC[3,5]
- While mutations in *TP53* and *PIK3CA* and amplifications in *CCND1* and *FGFR1* are associated with poor prognosis and CDK4/6i resistance,[4,9,10] the efficacy of LAS plus Abema in ELAINE 2 appeared similar in those with or without their co-occurrence with *ESR1* mutations
- The current analysis is limited by its exploratory nature and the small sample size of the study; results should be interpreted with caution
- With the ELAINE 2 data suggesting the potential of LAS plus Abema for treating *ESR1*-mutated, ER+/HER2- mBC in the post-CDK4/6i setting, the ongoing, registrational, phase 3, ELAINE 3 study (NCT05696626) will further evaluate the safety and efficacy of LAS plus Abema compared with fulvestrant plus Abema in a larger patient population

References

1. Brett JO, et al. *Breast Cancer Res*. 2021;23:85. **2.** Toy W, et al. *Nat Genet*. 2013;45:1439-1445. **3.** Wander SA, et al. *Cancer Discov*. 2020;10:1174-1193. **4.** Chien TJ. *Am J Cancer Res*. 2021;11:3813-3831. **5.** Rao X, et al. *Clin Cancer Res*. 2023;29:3372-3383. **6.** Goetz MP, et al. *Ann Oncol*. 2023;In Press. **7.** Damodaran S, et al. *Ann Oncol*. 2023;In Press. **8.** Damodaran S, et al. *Cancer Research*. 2023;83:P5-05-02. **9.** Meric-Bernstam F, et al. *JCO Precis Oncol*. 2018;2018. **10.** Fillbrunn M, et al. *BMC Cancer*. 2022;22:1002.

Disclosures

- **SD** has received research funding from EMD Serono, Guardant Health, Taiho Pharmaceuticals, Novartis, and Sermonix. **MAC** has received research support from Pfizer, Eli Lilly, and AstraZeneca; honoraria from Pfizer, Eli Lilly, Foundation Medicine, and Guardant; and has consulted for Menarini, Eli Lilly, AstraZeneca, Ellipses, Celcuity, Olaris, and Sermonix. **MPG** has CME activities from Research to Practice, Clinical Education Alliance, and Medscape; was a panelist for Total Health Conferencing and a moderator for Curio Science; has done consulting for AstraZeneca, Biovica, Biotheranostics, Blueprint Medicines, Eagle, Eli Lilly, Novartis, Pfizer, Sanofi Genzyme, and Sermonix; and has received research support from Eli Lilly, Pfizer, and Sermonix. **CCOS** has received research funding (paid to institution) from Eli Lilly, Sermonix, AstraZeneca, SeaGen Inc, Bavarian Nordic, Minnemarita Therapeutics, Biovica, and nFerence. **GR** is a consultant and stockholder of Sermonix and Arcadia Medicine, Inc. **HSR** has received research support from Pfizer, Merck, Novartis, Eli Lilly, Roche, Daiichi Sankyo, Seattle Genetics, Macrogenics, Sermonix, Boehringer Ingelheim, Polyphor, AstraZeneca, Ayala, Astellas, and Gilead; and honoraria from Puma, Samsung, and NAPO. **DGS** is on an advisory board for Novartis. **SAW** has received consulting/advisory board income from Foundation Medicine, Veracyte, Eli Lilly, Novartis, Pfizer, Biovica, AstraZeneca, and Puma Biotechnology; educational/speaking activities with Eli Lilly, Guardant Health, and 2ndMD; and institutional research support from Genentech, Pfizer, Nuvation Bio, Regor Therapeutics, and Sermonix. **DC** consults with Sermonix. **PVP** and **DJP** are employees and stockholders of Sermonix. **EG-Y** has received honoraria from Eli Lilly, Novartis, Pfizer, Roche, AstraZeneca, and MSD.
- Sermonix Pharmaceuticals sponsored the study and provided support for the medical writing assistance of Hui Zhang, PhD and Kathleen Ohleth, PhD (Precise Publications, LLC). Eli Lilly and Company provided abemaciclib for the study.

This presentation is the intellectual property of Sermonix Pharmaceuticals. Contact David J Portman at dportman@sermonixpharma.com for permission to reprint and/or distribute.