Abstract #1057

Lasofoxifene (LAS) Plus Abemaciclib (Abema) for Treating *ESR1*-mutated ER+/HER2- Metastatic Breast Cancer (mBC) after Progression on Prior Therapies: ELAINE 2 Study Update

Senthil Damodaran, MD, PhD;[1] Halle C. F. Moore, MD;[2] Ian C. Anderson, MD;[3] Mathew Cherian, MBBS;[4] Ciara C. O'Sullivan, MB, BCh, BAO;[5] Paul V. Plourde, MD;[6] David J. Portman, MD;[6] and Matthew P. Goetz, MD[5]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Cleveland Clinic, Cleveland, OH; [3]Providence Medical Group, Santa Rosa, CA; [4]The Ohio State University Comprehensive Cancer Center, Columbus, OH; [5]Mayo Clinic, Rochester, MN; [6]Sermonix Pharmaceuticals, Columbus, OH

Introduction

- Endocrine therapy (ET), particularly aromatase inhibitors (AIs), for estrogen receptor (ER)-positive breast cancer can lead to acquired *ESR1* mutations (*mESR1*), which drive endocrine resistance and tumor progression[1-4]
- Current therapies have limited activity for mBC with *mESR1*, especially in the post CDK4/6 inhibitor (CDK4/6i) setting[5,6]
- LAS is a next-generation ET and breast ER antagonist that prolonged progression-free survival (PFS; median 5.6 vs 3.7 mos; *P*=0.138) compared with fulvestrant (Fulv) in patients with *mESR1* mBC that progressed on an AI plus CDK4/6i, with a greater clinical benefit rate (CBR, 37% vs 22%; *P*=0.117) and objective response rate (ORR, 13% vs 3%; *P*=0.124), in the phase 2, ELAINE 1 study[7]
- Results from the phase 2, ELAINE 2 study showed that LAS plus Abema had a median PFS of ~13 mos, ORR of 33%, and CBR of 62%, with a well tolerated safety profile in patients with endocrine-resistant *mESR1* mBC[8]
- Here, we report ELAINE 2 data with longer patient follow up

Objective

The objective of the ELAINE 2 study was to evaluate the safety and efficacy of LAS combined with Abema in patients with *ESR1*-mutated, ER+/HER2- mBC in the post-CDK4/6i setting

Methods

- The open-label, phase 2, single-arm ELAINE 2 study (NCT04432454) enrolled women (≥18 years) with ER+/HER2- mBC and acquired *mESR1* identified in circulating tumor DNA (ctDNA) who had progressed on prior ET for mBC (≤2 lines)
 - Prior CDK4/6i and/or one line of chemotherapy for mBC was allowed
- Patients took oral LAS 5 mg/day and Abema (supplied by Eli Lilly and Co) 150 mg BID until disease progression, death, toxicity, or withdrawal
- **Primary endpoint:** safety and tolerability (AEs)
- **Secondary endpoints:** PFS, CBR, ORR, duration of response, and time to response
 - Staging scans were performed every 8 weeks
- *ESR1* mutant allele fractions (MAF) were analyzed in ctDNA at baseline and wk 4 using Sysmex-Inostics SafeSeq assay
 - Guardant 360 ctDNA analysis was utilized to identify other genomic alterations
- Data were summarized descriptively with no formal hypothesis testing

2023 American Society of Clinical Oncology Annual Meeting • June 2-6 • Chicago, IL

Results

Figure 1. Time on treatment and response in all patients

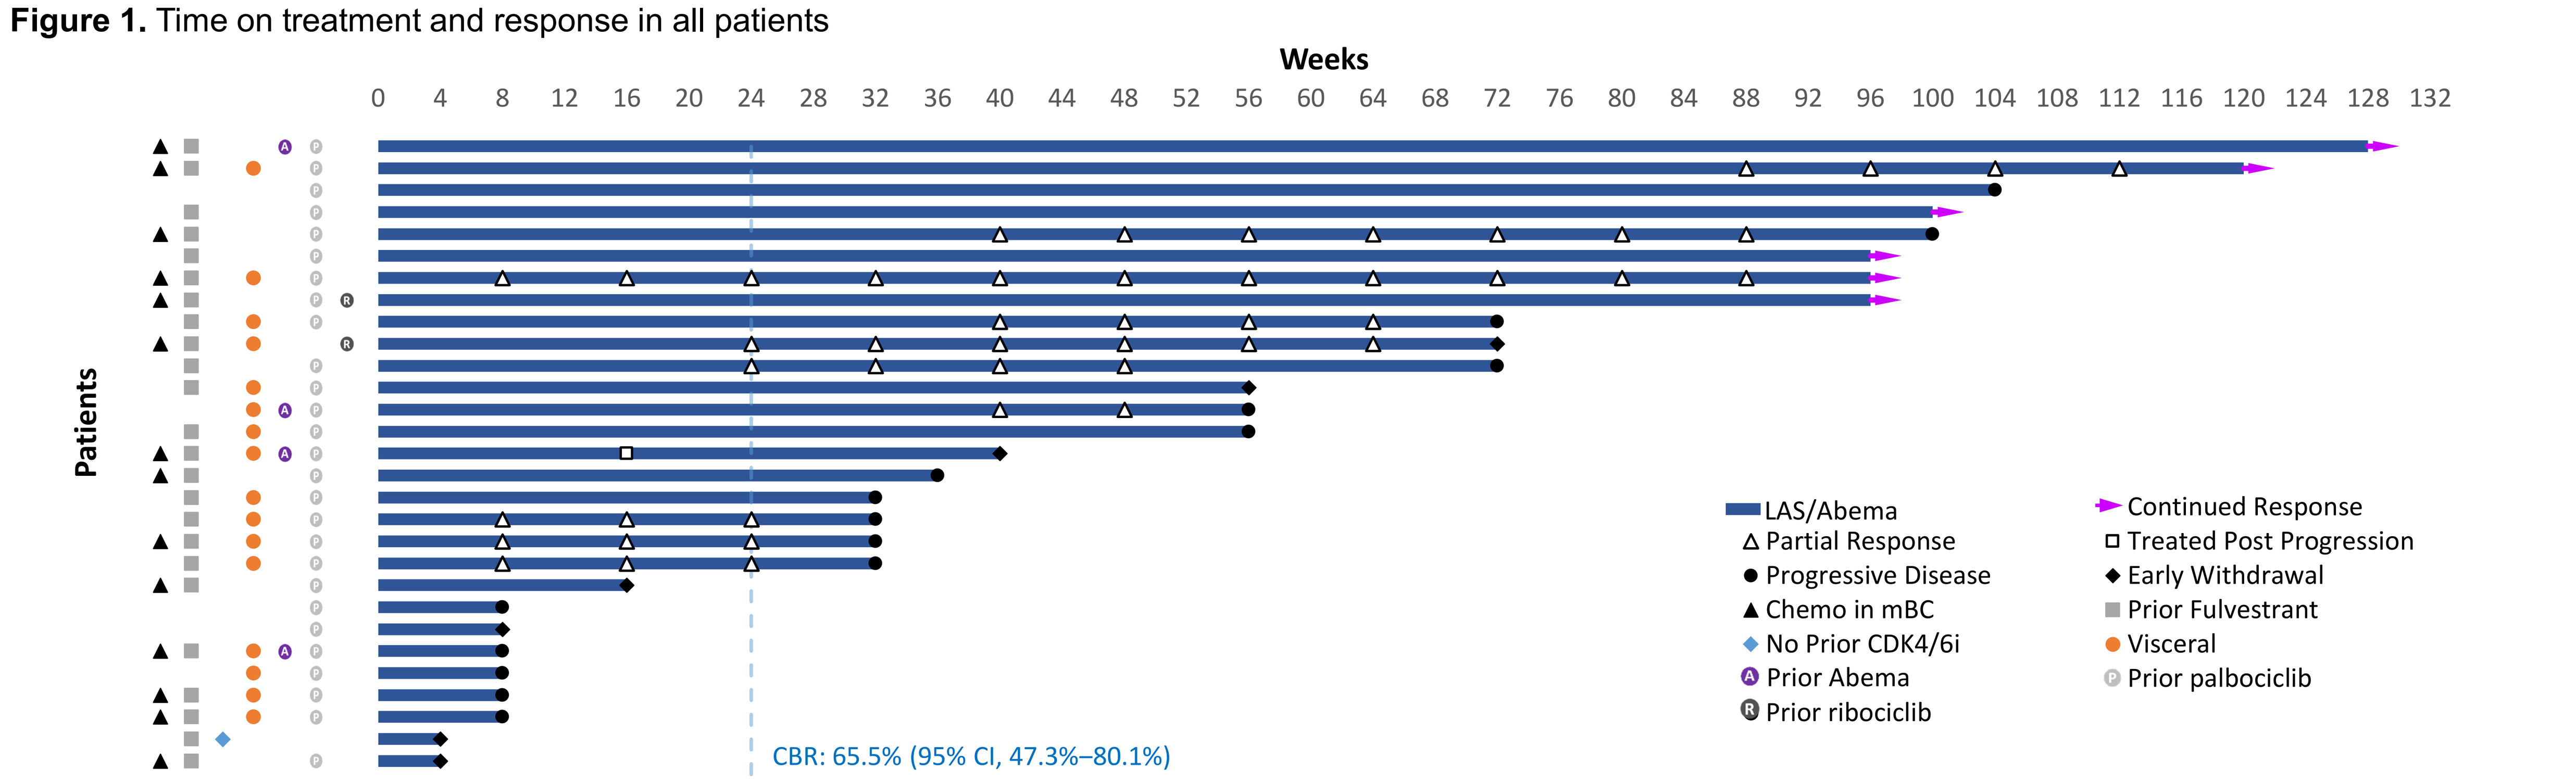
Weeks
0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76 80 84 88 92 96 100 104 108 112 116 120 124 128 132
Patients
LAS/Abema
Partial Response
Progressive Disease
Chemo in mBC
No Prior CDK4/6i
Prior Abema
Prior ribociclib
Continued Response
Treated Post Progression
Early Withdrawal
Prior Fulvestrant
Visceral
Prior palbociclib
CBR: 65.5% (95% CI, 47.3%–80.1%)

Data as of April 2023. Abema, abemaciclib; CBR, clinical benefit rate; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; CI, confidence interval; LAS, lasofoxifene.

Patient disposition and baseline characteristics

- As of April 2023, 6 of 29 enrolled patients were continuing treatment (**Figure 1**); 17 had disease progression, 1 discontinued due to AEs, 3 patient withdrawal, and 2 for other causes
- Patients had a median age of 60 yrs and received a median of 2 prior lines of ET for mBC; 55.2% had visceral disease; all but one (96.6%) received prior CDK4/6i (median duration 2 yrs), 79.3% prior Fulv, and 48.3% prior chemotherapy for mBC
- The most frequently detected *ESR1* mutations at baseline were Y537S (65.5%), D538G (44.8%), Y536N (27.6%), and Y537C (10.3%); 48.3% of patients had polyclonal *ESR1* mutations

Table 1. Most frequently reported TEAEs (in ≥12% of patients)* (N=29)

TEAE, n (%)	Grade 2	Grade 3	Grade 4	All grades
Diarrhea	5 (17.2)	0	0	24 (82.8)
Nausea	5 (17.2)	0	0	15 (51.7)
Fatigue	3 (10.3)	1 (3.4)	0	11 (37.9)
Vomiting	2 (6.9)	1 (3.4)	0	9 (31.0)
Anemia	1 (3.4)	3 (10.3)	0	8 (27.6)
Dyspnea	4 (13.8)	0	0	8 (27.6)
White blood cell count decreased	6 (20.7)	0	0	8 (27.6)
Blood creatinine increased	4 (13.8)	0	0	7 (24.1)
Constipation	0	1 (3.4)	0	6 (20.7)
Cough	1 (3.4)	0	0	6 (20.7)
Decreased appetite	2 (6.9)	0	0	6 (20.7)
Hypokalemia	0	3 (10.3)	0	5 (17.2)
Muscle spasm	0	0	0	5 (17.2)
Alopecia	0	0	0	4 (13.8)
Dehydration	2 (6.9)	0	0	4 (13.8)
Dizziness	2 (6.9)	0	0	4 (13.8)
Fall	1 (3.4)	2 (6.9)	0	4 (13.8)
Hot flush	1 (3.4)	0	0	4 (13.8)
Hypoalbuminemia	0	0	0	4 (13.8)
Myalgia	0	0	0	4 (13.8)
Edema peripheral	0	0	0	4 (13.8)
Pain in extremity	0	0	0	4 (13.8)
Pruritus	1 (3.4)	0	0	4 (13.8)
Stomatitis	1 (3.4)	0	0	4 (13.8)
Urinary tract infection	4 (13.8)	0	0	4 (13.8)

*Patients' maximum grade was counted.
TEAEs, treatment-emergent adverse events, defined as adverse events that started or worsened after the first dose.

Safety

- LAS/Abema was well tolerated with primarily grade 1/2 treatment-emergent AEs (TEAEs), most commonly diarrhea, nausea, fatigue, and vomiting (**Table 1**)
- Three patients (10.3%) had scan-identified VTEs (**Table 2**)
- One patient discontinued treatment due to grade 2 diarrhea
- No deaths on treatment occurred
- Abema dose was reduced to 100 mg BID in 6 (20.7%) patients; LAS dose was not reduced per protocol

Table 2. Patients who experienced thrombotic events

Pt	Prior treatment	VTE event	Action/Outcome
1	1st line: Fulv/RIB for 36 wks; 2nd line: CAPE for 16 wks	Symptomatic spontaneous thrombotic event (patient presenting with shortness of breath) occurred at wk 64 and confirmed with a PE (grade 3) on imaging	Patient received anticoagulation and remained on study, but subsequently developed bleeding complications and was withdrawn at 72 wks, shortly after PE diagnosis Patient had an ongoing confirmed PR with a 50% reduction of 8-cm sum diameter liver metastases of 30 wks duration at that time thrombotic event occurrence
2	1st line: Fulv/PAL for 20 wks; 2nd line: Fulv/ALP for 20 wks	A DVT occurred after an elective knee surgery and an asymptomatic PE noted on surveillance scans at wk 24	Patient's blood successfully anticoagulated, and therapy reinitiated. Clinical benefit achieved Patient was on treatment for 36 wks before disease progression
3	1st line: paclitaxel for 1 yr; 2nd line: Fulv/PAL for 3.5 yrs	An asymptomatic PE noted on surveillance scans at wk 72 and an asymptomatic popliteal DVT diagnosed upon further evaluation	Patient anticoagulated, and therapy reinitiated. Patient was on treatment for 100 wks with a confirmed PR of 60 wks duration before disease progression

ALP, alpelisib; CAPE, capecitabine; DVT, deep vein thrombosis; Fulv, fulvestrant; PAL, palbociclib; PE, pulmonary embolism; PR, partial response; RIB, ribociclib; VTE, venous thromboembolism.

Table 3. Treatment response in patients with co-existing genomic alterations (N=26)

Alteration	Baseline n (%)	CBR	Median PFS
***PIK3CA* mutation**	8 (28%)	63%	36 wks
CCND1* amplification	6 (21%)	100%	72 wks
FGFR1* amplification	5 (17%)	100%	72 wks
***TP53* mutation**	12 (41%)	58%	72 wks

*3 of the 6 patients with baseline CCND1 amplification also had FGFR1 amplification.
Data as of May 2023. CBR, clinical benefit rate; PFS, progression-free survival.

Figure 2. Kaplan-Meier analysis of progression-free survival

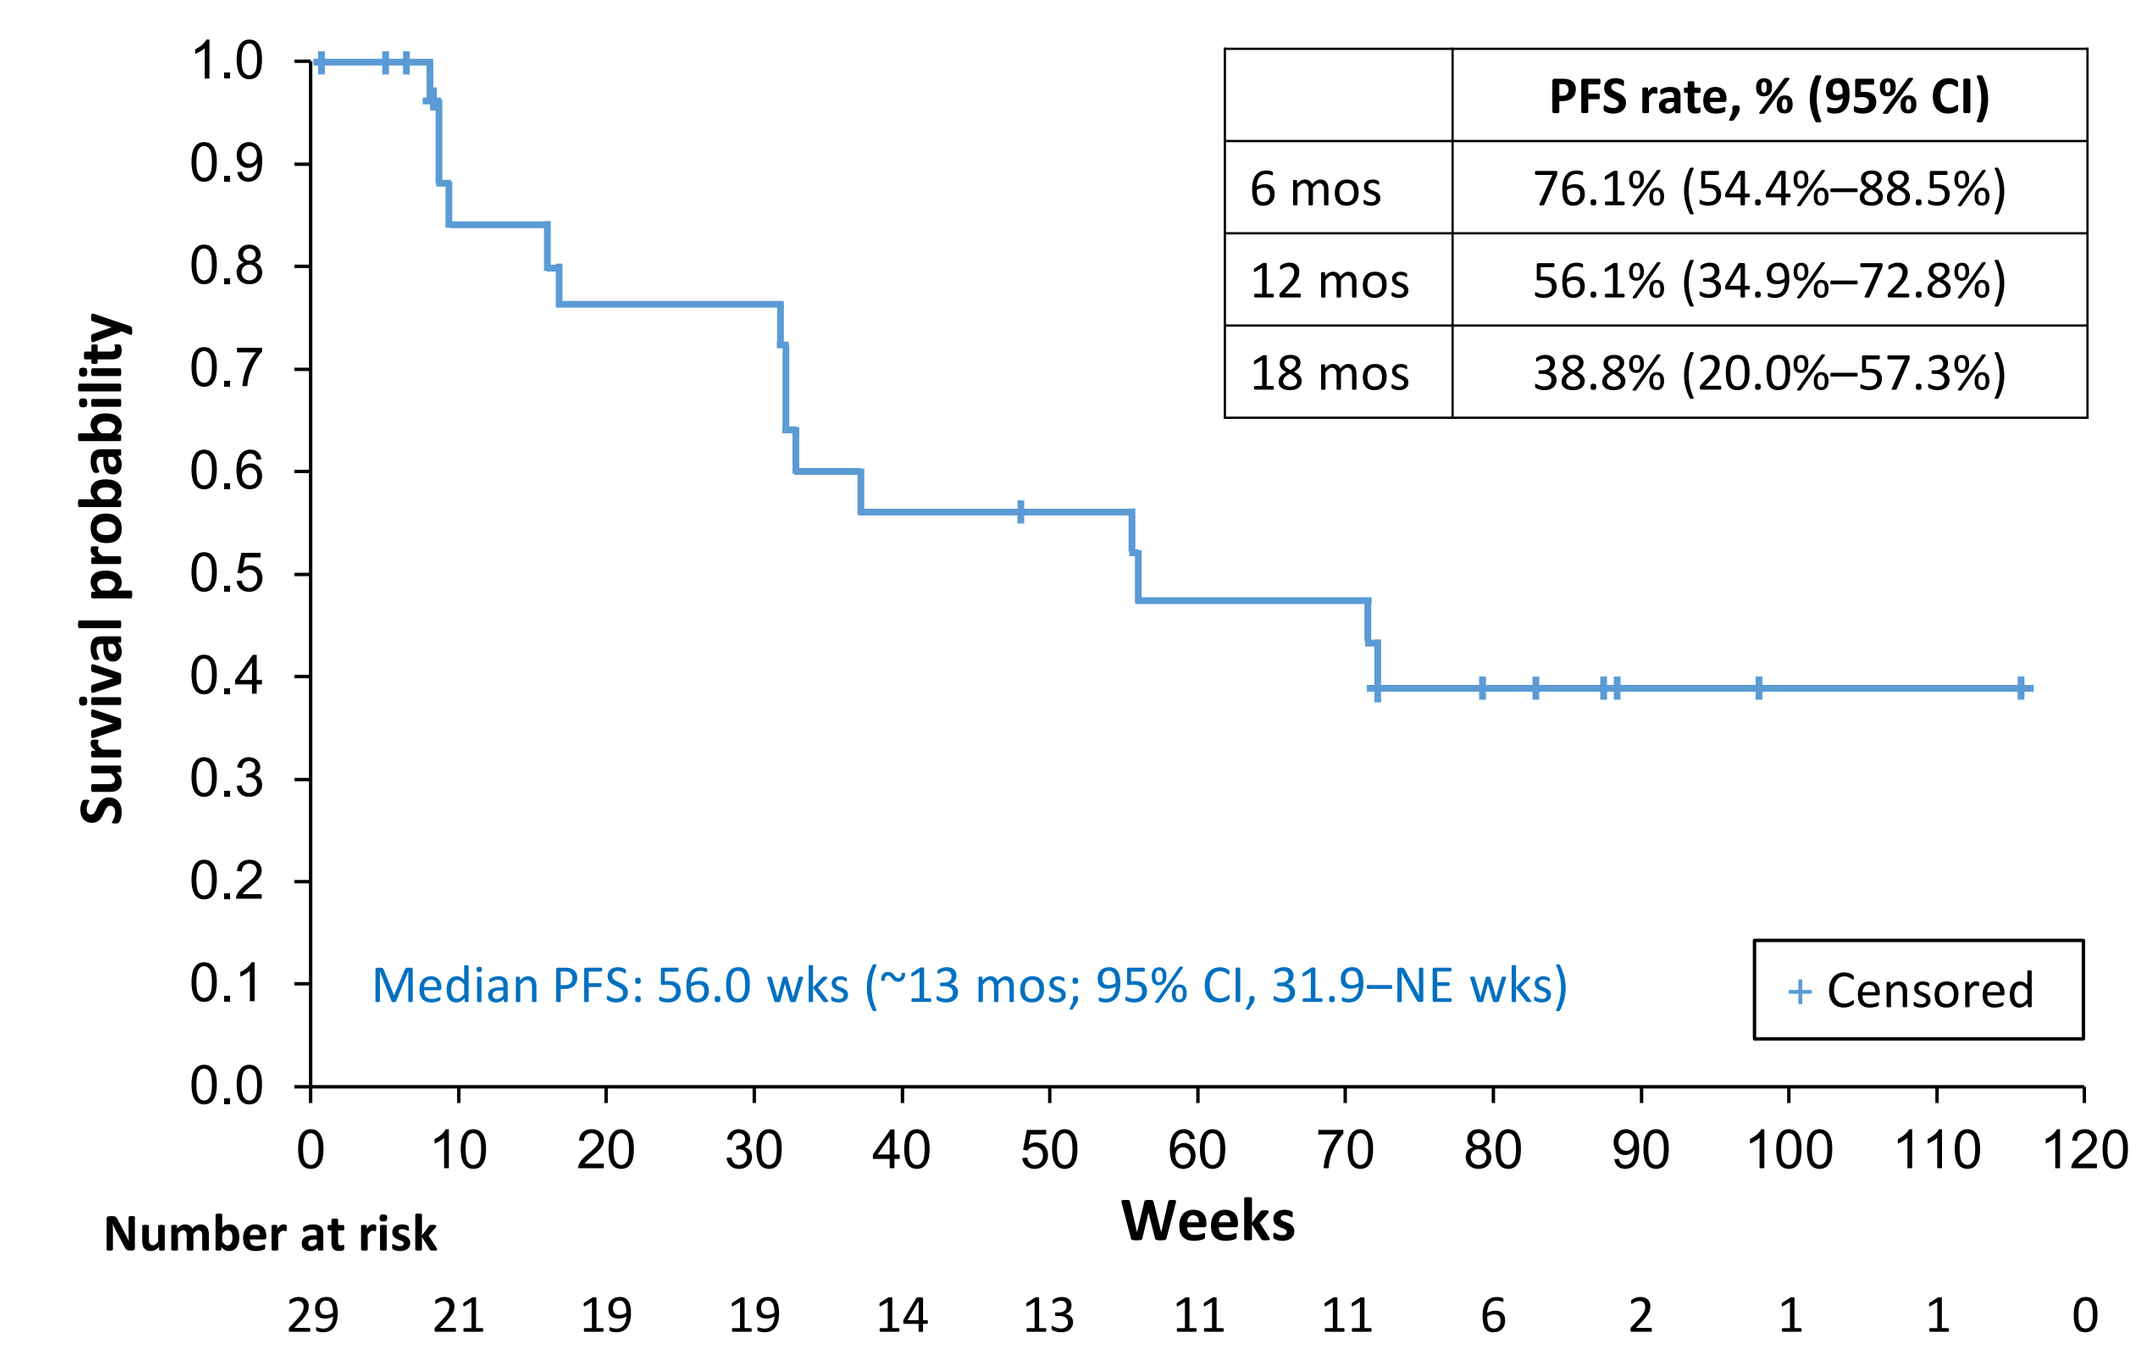
Survival probability
1.0 0.9 0.8 0.7 0.6 0.5 0.4 0.3 0.2 0.1 0.0
PFS rate, % (95% CI)
6 mos 76.1% (54.4%–88.5%)
12 mos 56.1% (34.9%–72.8%)
18 mos 38.8% (20.0%–57.3%)
Median PFS: 56.0 wks (~13 mos; 95% CI, 31.9–NE wks)
+ Censored
0 10 20 30 40 50 60 70 80 90 100 110 120
Weeks
Number at risk
29 21 19 19 14 13 11 11 6 2 1 1 0

Data as of January 2023. CI, confidence interval; NE, not estimable; PFS, progression-free survival.

Figure 3. Best response in patients with measurable lesions (n=18)

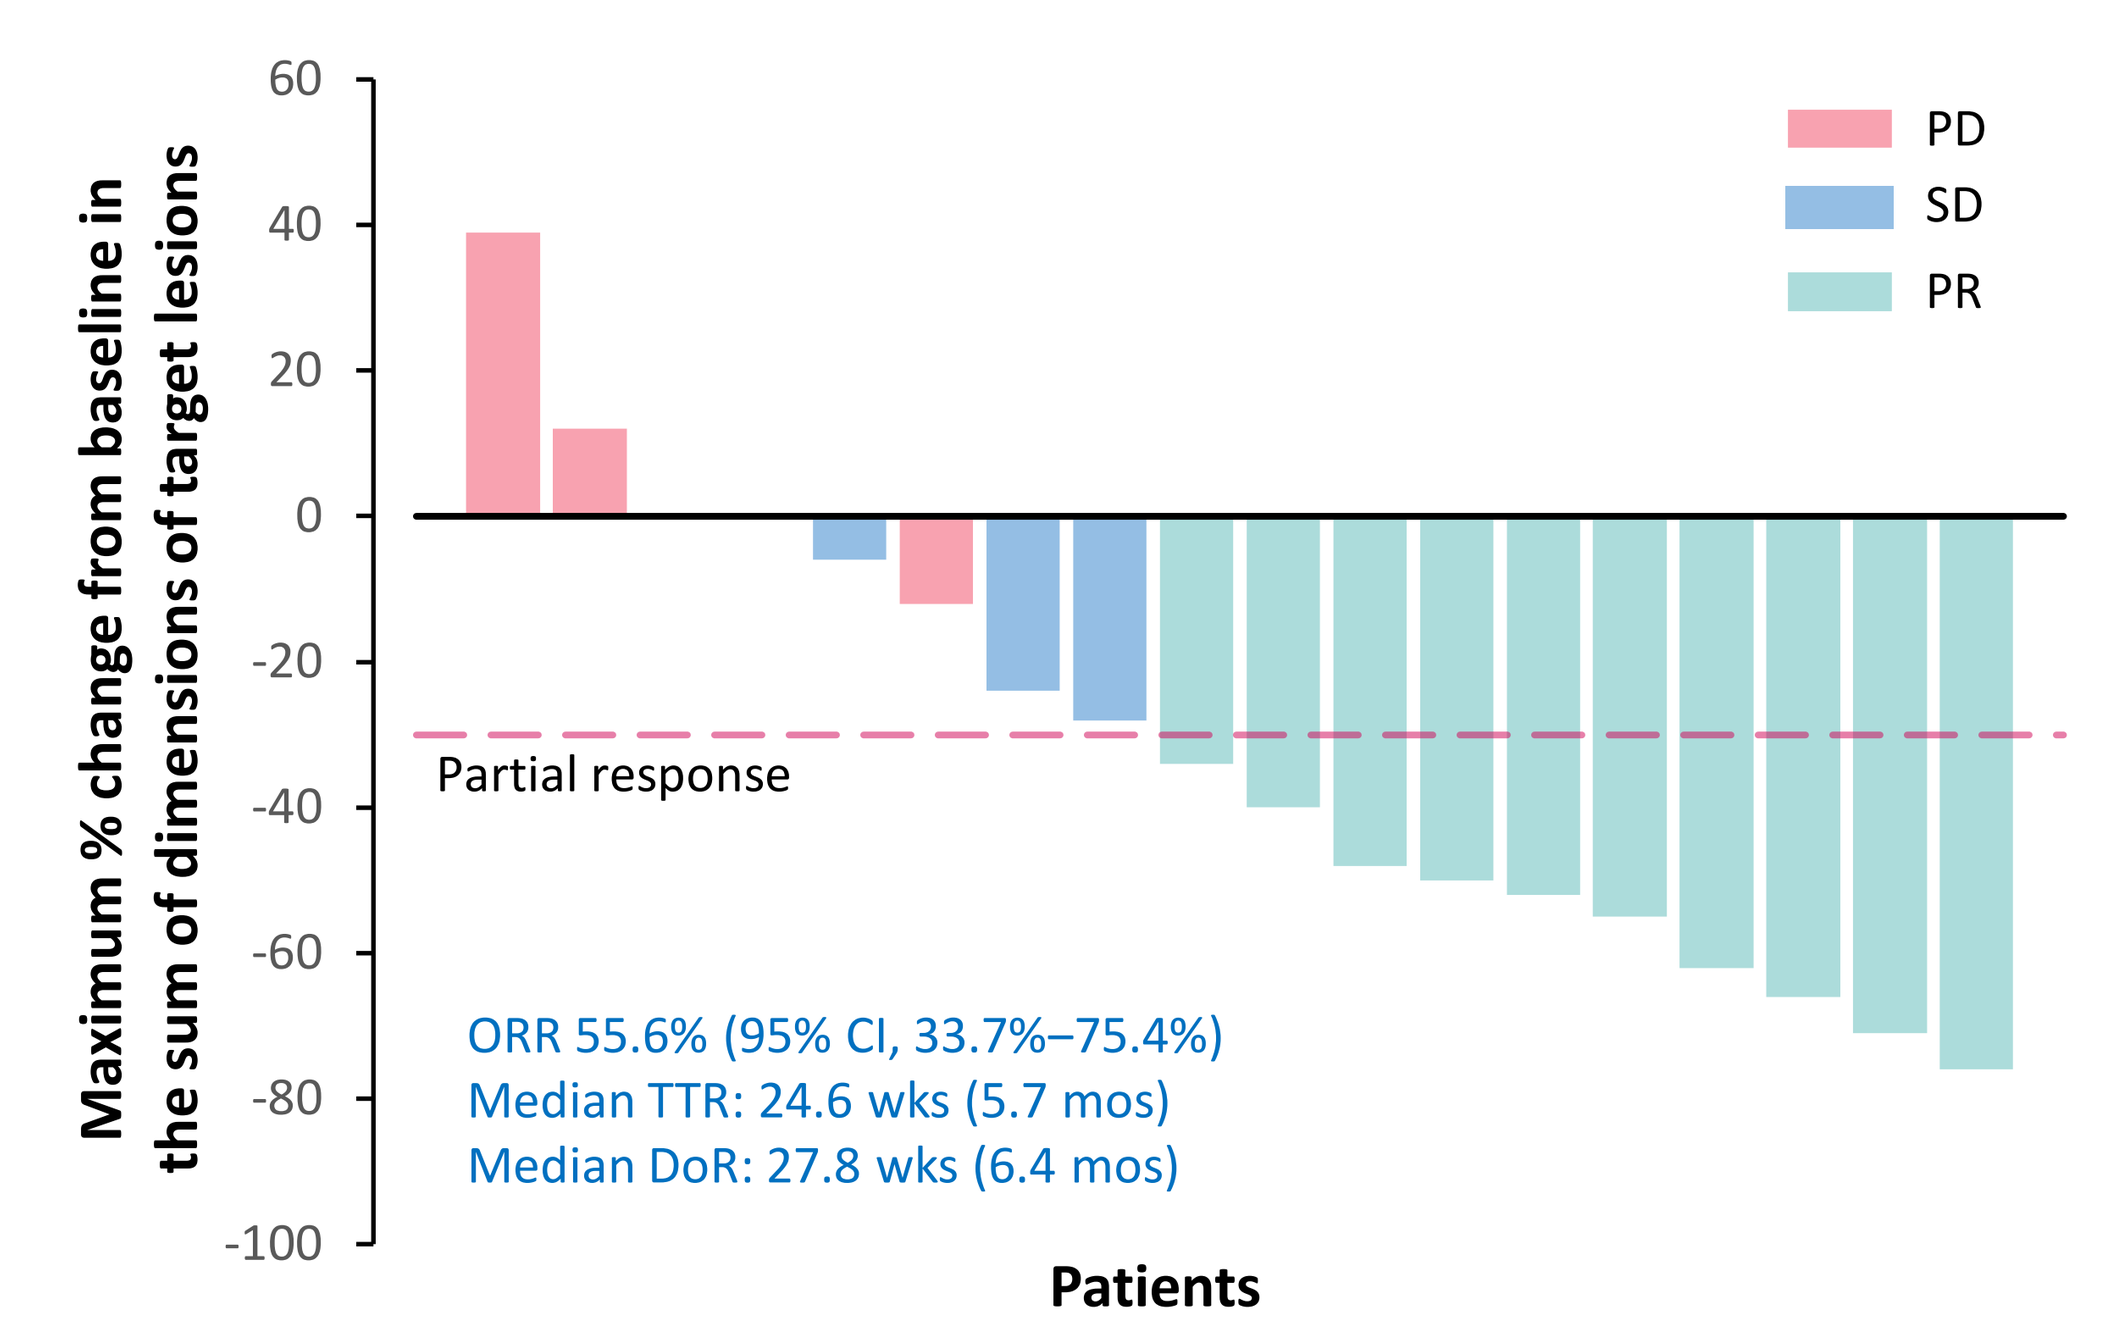
Maximum % change from baseline in the sum of dimensions of target lesions
60 40 20 0 -20 -40 -60 -80 -100
PD
SD
PR
Partial response
ORR 55.6% (95% CI, 33.7%–75.4%)
Median TTR: 24.6 wks (5.7 mos)
Median DoR: 27.8 wks (6.4 mos)
Patients

Data as of April 2023. CI, confidence interval; DoR, duration of response; ORR, objective response rate; PD, progressive disease; PR, partial response; SD, stable disease; TTR, time to response.

Efficacy

- Median PFS was 56.0 wks (~13 mos; **Figure 2**), CBR was 65.5% (**Figure 1**), and ORR was 55.6% (**Figure 3**); median overall survival was not estimable at the time
- PFS rate (95% CI) was 76.1%, 56.1%, and 38.8% at 6, 12, and 18 mos respectively (**Figure 2**); 8 (27.6%) patients achieved PFS over 96 wks
- Of the 4 patients who had prior Abema exposure, two achieved clinical benefit, and one with RECIST progression at wk 16 remained on study with stable disease until wk 40
- In 26 patients with evaluable baseline and wk 4 ctDNA, *ESR1* MAF decreased at wk 4 in 21 (80.8%) patients, including 14 (53.8%) whose *ESR1* MAF was undetectable
- Antitumor activity of LAS/Abmea was not compromised by co-occurring alterations that confer endocrine resistance (**Table 3**)

Key Takeaways

- In ELAINE 2, LAS/Abema was well tolerated with an acceptable safety profile
- LAS/Abema was associated with a clinically meaningful PFS (median ~13 mos), ORR (56%), and CBR (66%) in women with ER+/HER2- mBC and acquired *mESR1*
- *ESR1* MAF decreased with LAS/Abema, consistent with target engagement

Conclusions

- With longer ELAINE 2 follow up, LAS/Abema continued to be well tolerated; no new safety signals were noted
 - Incidence of PE/DVT in ELAINE 2 was within the range previously reported for Abema-based therapies[9-11]
- LAS/Abema exhibited meaningful antitumor activity in patients with *mESR1*, ER+/HER2- mBC that had progressed on ET and CDK4/6is
- Observed decreases in *mESR1* ctDNA suggest target engagement with LAS
- Other co-occurring genetic alterations associated with resistance did not appear to compromise the efficacy of LAS/Abema
- A confirmatory, phase 3 study (ELAINE 3; NCT05696626) will compare LAS/Abema with Fulv/Abema. If ELAINE 2 results are confirmed, the combination of LAS and Abema may provide a practice-changing option for treating *mESR1* mBC

Disclosures

- SD has received research funding from EMD Serono, Guardant Health, Taiho Pharmaceuticals, Novartis, and Sermonix Pharmaceuticals. HCFM has received research funding (paid to institution) from Daiichi Sankyo, Roche, AstraZeneca, and Sermonix Pharmaceuticals, and consulting fees from Myovant. ICA has received research funding (paid to institution) from AbbVie, Apollomics, AstraZeneca, EMD Serono, Hutchison MediPharma, Merck, Seattle Genetics, and Turning Point Therapeutics. CCOS has received research funding (paid to institution) from Eli Lilly, Sermonix, AstraZeneca, Seagen Inc, Bavarian Nordic, Minnemarita Therapeutics, Biovica, and nFerence. DJP and PVP are employees and stockholders of Sermonix Pharmaceuticals. MPG has CME activities from Research to Practice, Clinical Education Alliance, and Medscape; was a panelist for Total Health Conferencing and a moderator for Curio Science; has done consulting for AstraZeneca, Biovica, Biotheranostics, Blueprint Medicines, Eagle, Eli Lilly, Novartis, Pfizer, Sanofi Genzyme, and Sermonix; and has received research support from Eli Lilly, Pfizer, and Sermonix Pharmaceuticals. MAC declares no conflicts of interest.
- Eli Lilly and Co provided abemaciclib for the study. Sermonix Pharmaceuticals sponsored the study and provided support for the medical writing assistance of Hui Zhang, PhD and Kathleen Ohleth, PhD (Precise Publications, LLC).

Corresponding author: David J. Portman, MD (DPortman@sermonixpharma.com)

References: **1.** De Santo I, et al. *Cancers (Basel)*. 2019;11:1894. **2.** Brett JO, et al. *Breast Cancer Res*. 2021;23:85. **3.** Robinson DR, et al. *Nat Genet*. 2013;45:1446. **4.** Toy W, et al. *Nat Genet*. 2013;45:1439. **5.** Bidard FC, et al. *J Clin Oncol*. 2022;40:3246. **6.** Kalinsky K, et al. *J Clin Oncol*. 2022;40:LBA1004. **7.** Goetz MP, et al. *Ann Oncol*. 2022;33:S1387. **8.** Damodaran S, et al. *J Clin Oncol*. 2022;40:1022. **9.** Rugo HS, et al. *Oncologist*. 2021;26:e522. **10.** Hamilton E, et al. *Breast Cancer Res Treat*. 2022;195:55-64. **11.** Wander SA, et al. *J Natl Compr Canc Netw*. 2021:1.