P5-05-04

SAN ANTONIO BREAST CANCER SYMPOSIUM®
December 6-10, 2022

Estrogen receptor 1 (*ESR1*) mutations in circulating tumor DNA (ctDNA) from patients with ER+/HER2- metastatic breast cancer (mBC) treated with lasofoxifene or fulvestrant in the ELAINE 1 study

Matthew P Goetz[1]; Einav Nili Gal-Yam[2]; Daniel G Stover[3]; Sarah L Sammons[4]; Stephanie L Graff[5]; Grace Wang[6]; Massimo A Cristofanilli[7]; Gary Riordan[8]; Hillary S Sloane[9]; Dominic Carroll[10]; Paul V Plourde[10]; David J Portman[10]

[1]Mayo Clinic Rochester, Rochester, MN; [2]Breast Oncology Institute Sheba Medical Center, Ramat Gan, Israel; [3]Ohio State University, Columbus, OH; [4]Dana Farber Cancer Institute, Harvard Medical School, Boston, MA; [5]Lifespan Cancer Institute/Legorreta Cancer Center at Brown University, Providence, RI; [6]Miami Cancer Institute at Baptist Health, Inc, Miami, FL; [7]Weill Cornell Medicine, New York, NY; [8]Reg Pro1, LLC, Tucson, AZ; [9]Sysmex Inostics, Baltimore, MD; [10]Sermonix Pharmaceuticals, Columbus, OH

Introduction

- Acquired *ESR1* mutations (*mESR1*) during endocrine therapy drive treatment resistance, metastasis, and poor prognosis in ER+/HER2- mBC patients[1-4]
- ELAINE 1 (NCT03781063) is a phase 2, randomized trial of lasofoxifene (LAS), a SERM, versus fulvestrant (Fulv), a SERD, in *mESR1* mBC patients with disease progression on an AI plus a CDK4/6i
 - Preliminary median PFS was 6.04 months (LAS) vs 4.04 months (Fulv; *P*=0.138), with a CBR of 36.5% vs 21.6% (*P*=0.12), respectively, and a favorable safety profile[5]
 - Numerically more commonly detected *mESR1* variants decreased with LAS vs Fulv, regardless of baseline *ESR1* mutant allele fraction (MAF; percentage of cell-free DNA that contains the mutant allele), in preliminary analyses[6]
- Changes in MAF from ctDNA may correlate with clinical response to targeted therapies[7,8]

Objectives

- To evaluate the association between changes in ctDNA *ESR1* MAF with clinical responses to LAS or Fulv in ELAINE 1
- To assess the Y537S ctDNA kinetics and its association with clinical outcomes in both treatment arms

Methods

- Women with ER+/HER2- mBC and *mESR1* ctDNA whose disease progressed on prior AI plus CDK4/6i (≥12 mos) were randomized to oral LAS 5 mg daily or IM Fulv 500 mg on days 1, 15, and 29, then every 4 weeks, until disease progression or severe toxicity
- Efficacy measures were PFS (primary endpoint) and CBR (secondary endpoint; clinical benefit: CR, PR, or SD for ≥24 weeks). Patients were stratified by visceral disease (y/n) and the Y537S mutation (y/n).
- ctDNA was analyzed at baseline and week 8 using the Sysmex Inostics OncoBeam or SafeSeq assays, which detect clone-specific *mESR1* at low MAFs (limit of detection 0.05%)
- *ESR1* MAF changes from baseline were characterized as **decreased** (decreased or cleared [*mESR1* not detected]), **increased** (increased), or **equivocal** (some increased and decreased MAF trends in polyclonal patients [>1 *mESR1*])
 - Patients with equivocal MAF changes were excluded from analyses of associations between MAF changes and clinical benefit

Results

- 103 patients were randomized to LAS (n=52) or Fulv (n=51) in the ITT population
- As of October 2022, 4 patients (LAS vs Fulv: 2 vs 2) remain in the study
 - 86 discontinuations due to disease progressions (45 vs 41);
 - 13 discontinuations due to consent withdrawal (2 vs 5), relocation (0 vs 1), investigator decision (2 vs 1), adverse event (1 vs 0), and others (0 vs 1)
- Patient demographics and characteristics are shown in Table 1
- Most commonly detected *mESR1* variants (>10%) were D538G (56%), Y537S (39%), Y537N (29%), E380Q (22%), and Y537C (11%)
- 61 patients (59%) had evaluable baseline and week 8 ctDNA and unequivocal *ESR1* MAF changes

Table 1. Baseline demographics and characteristics (ITT population)

	Lasofoxifene (n=52)	Fulvestrant (n=51)
Mean age (range), yrs	**61.6 (33–84)**	**60.1 (38–82)**
Race		
White	**43 (82.7)**	**42 (82.4)**
Black or African American	**6 (11.5)**	**5 (9.8)**
Asian	**3 (5.8)**	**4 (7.8)**
Measurable disease, n (%)	38 (73.1)	33 (64.7)
Visceral disease, n (%)	**35 (67.3)**	**33 (64.7)**
Chemotherapy in mBC, n (%)	3 (5.8)	3 (5.9)
AI/CDK4/6i, n (%)	52 (100)	51 (100)
Mean duration on AI/CDK4/6i, yrs	**2.5**	**2.2**
ESR1 mutation, n (%)	52 (100)	51 (100)
D538G	34 (65.4)	24 (47.1)
Y537S	18 (34.6)	22 (43.1)
Y537N	14 (26.9)	16 (31.4)
E380Q	9 (17.3)	14 (27.5)
Y537C	7 (13.5)	4 (7.8)
Polyclonal ESR1 mutation, n (%)	**29 (55.8)**	**27 (52.9)**

Table 2. Change from baseline to week 8 in *ESR1* MAF

ESR1 MAF	Lasofoxifene (n=35)	Fulvestrant (n=26)
Cleared, n (%)	11 (31)	6 (23)
Decreased only (excluding cleared), n (%)	18 (51)	10 (38)
Increased, n (%)	6 (17)	10 (38)

- *mESR1* MAF decreased in numerically more patients with LAS vs Fulv (Table 2, Figure 1)
- In patients with the Y537S variant, complete clearance of Y537S was observed in 5/15 (33%) with LAS vs 1/18 (6%) with Fulv
- Median percent changes in *mESR1* MAF were substantially different between LAS and Fulv for all variants and Y537S (Figure 2)
- PFS was numerically greater with LAS than with Fulv in patients with increased and decreased *ESR1* MAF (Table 3)
- CBR was 55% (16/29) on LAS and 25% (4/16) on Fulv in patients with decreased *ESR1* MAF (Table 3)
- In patients with *mESR1* clearance, 10 of 11 on LAS experienced clinical benefit (PPV 90.9%), in contrast to 2 of 6 on Fulv (PPV 33.3%)
- Figure 3 shows changes in *mESR1* variants for individual patients

Figure 1. Proportion of patients with decreased *ESR1* variants from baseline to week 8

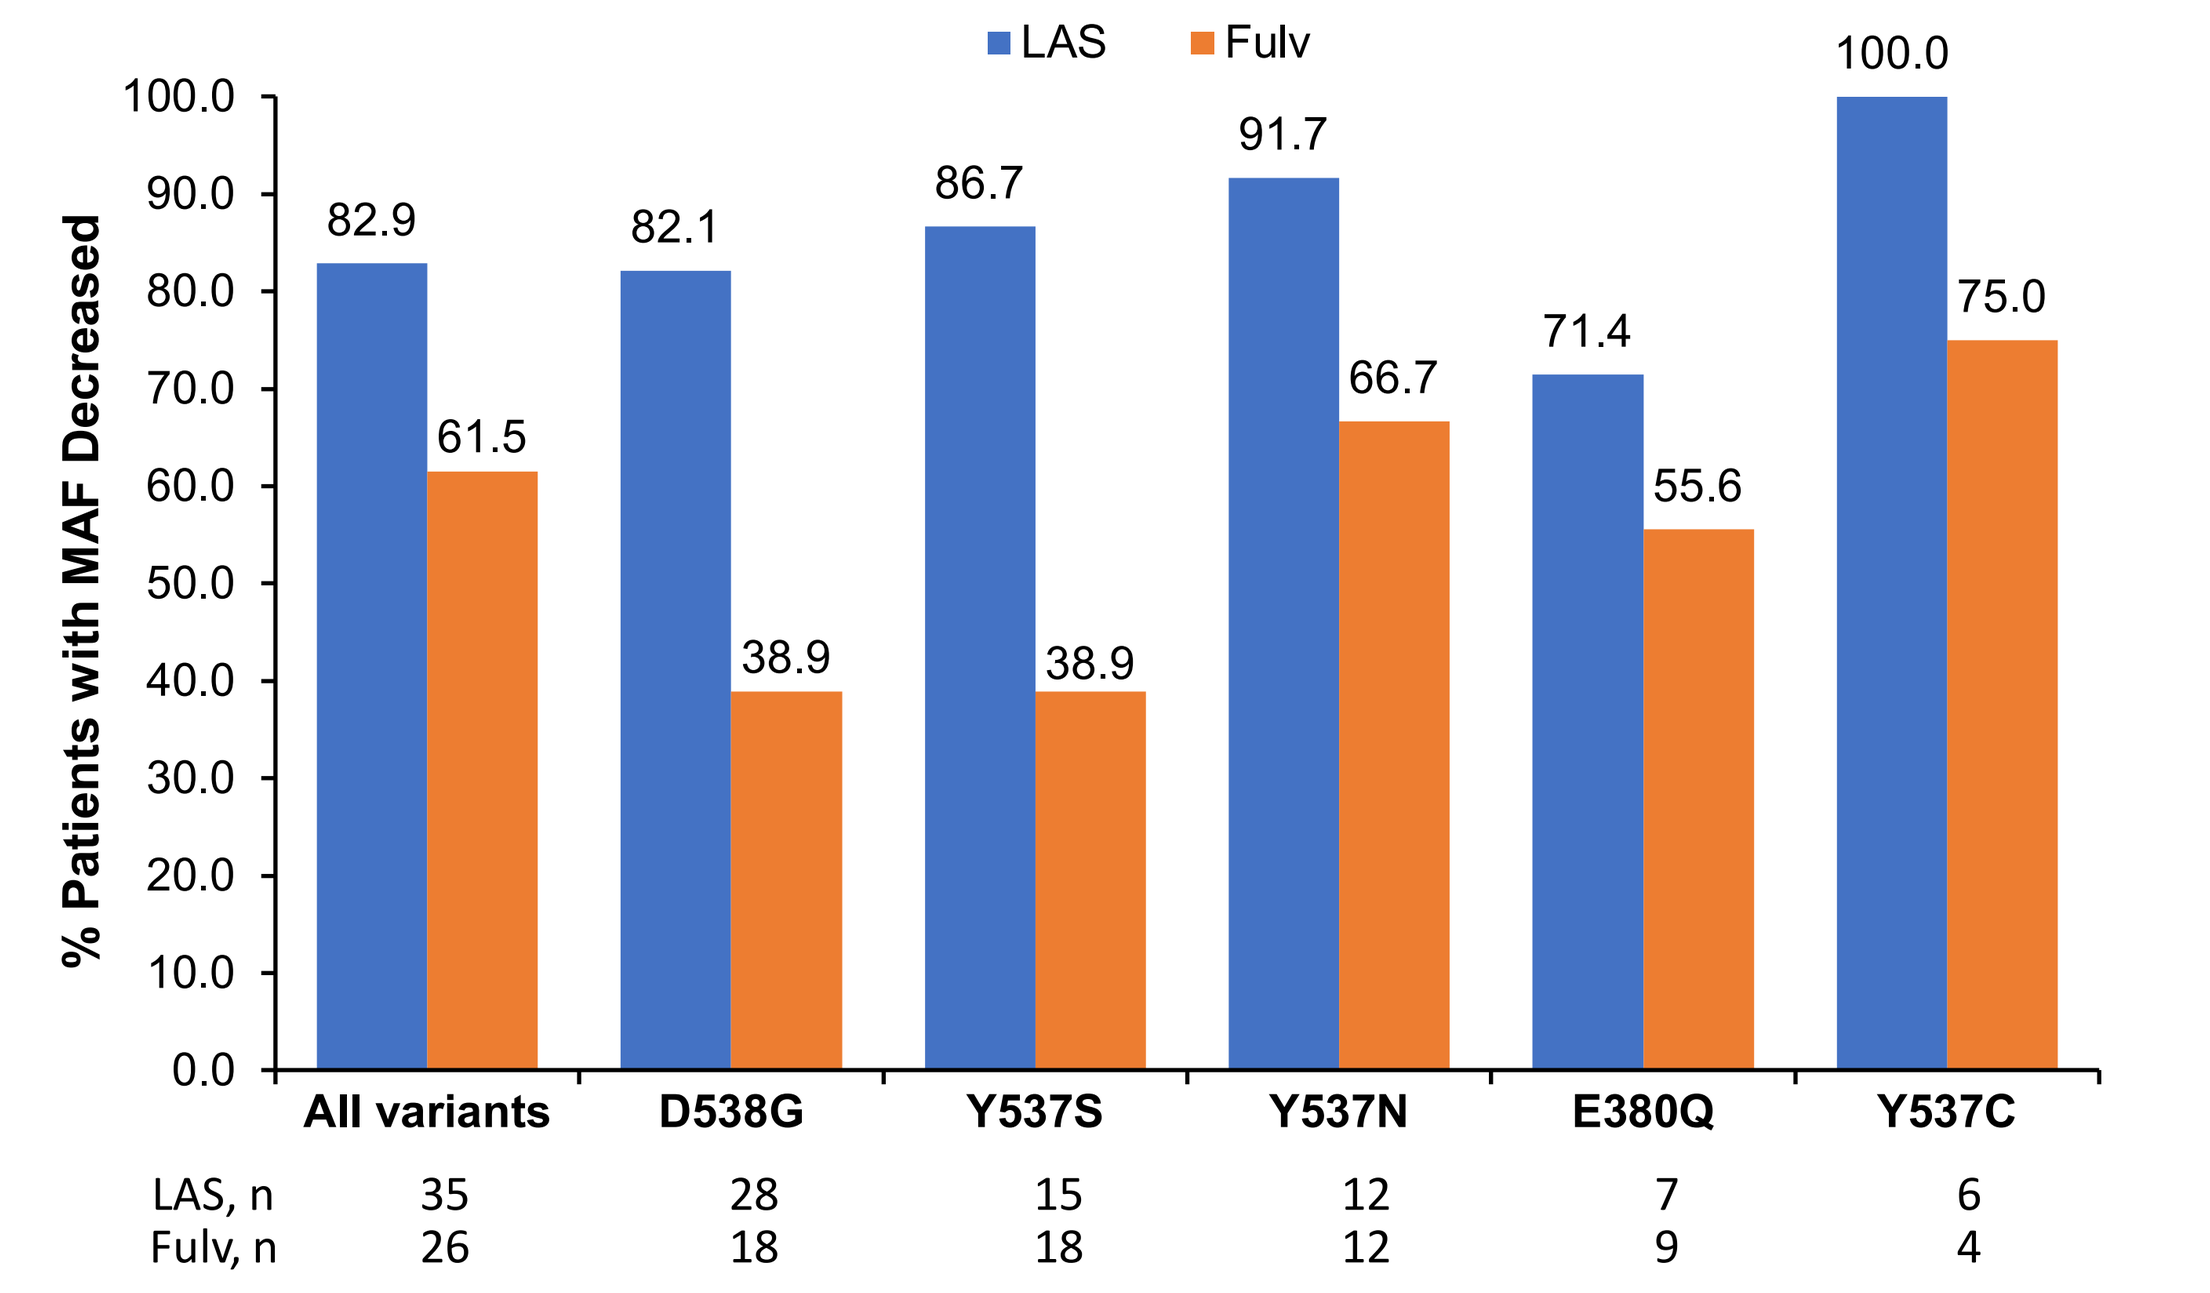
LAS
Fulv
% Patients with MAF Decreased
82.9
61.5
82.1
38.9
86.7
38.9
91.7
66.7
71.4
55.6
100.0
75.0
All variants
D538G
Y537S
Y537N
E380Q
Y537C
LAS, n 35 28 15 12 7 6
Fulv, n 26 18 18 12 9 4

n: number of patients having evaluable baseline and week-8 ctDNA results and respective *ESR1* mutation.

Figure 2. Median % changes in *ESR1* MAF from baseline to week 8

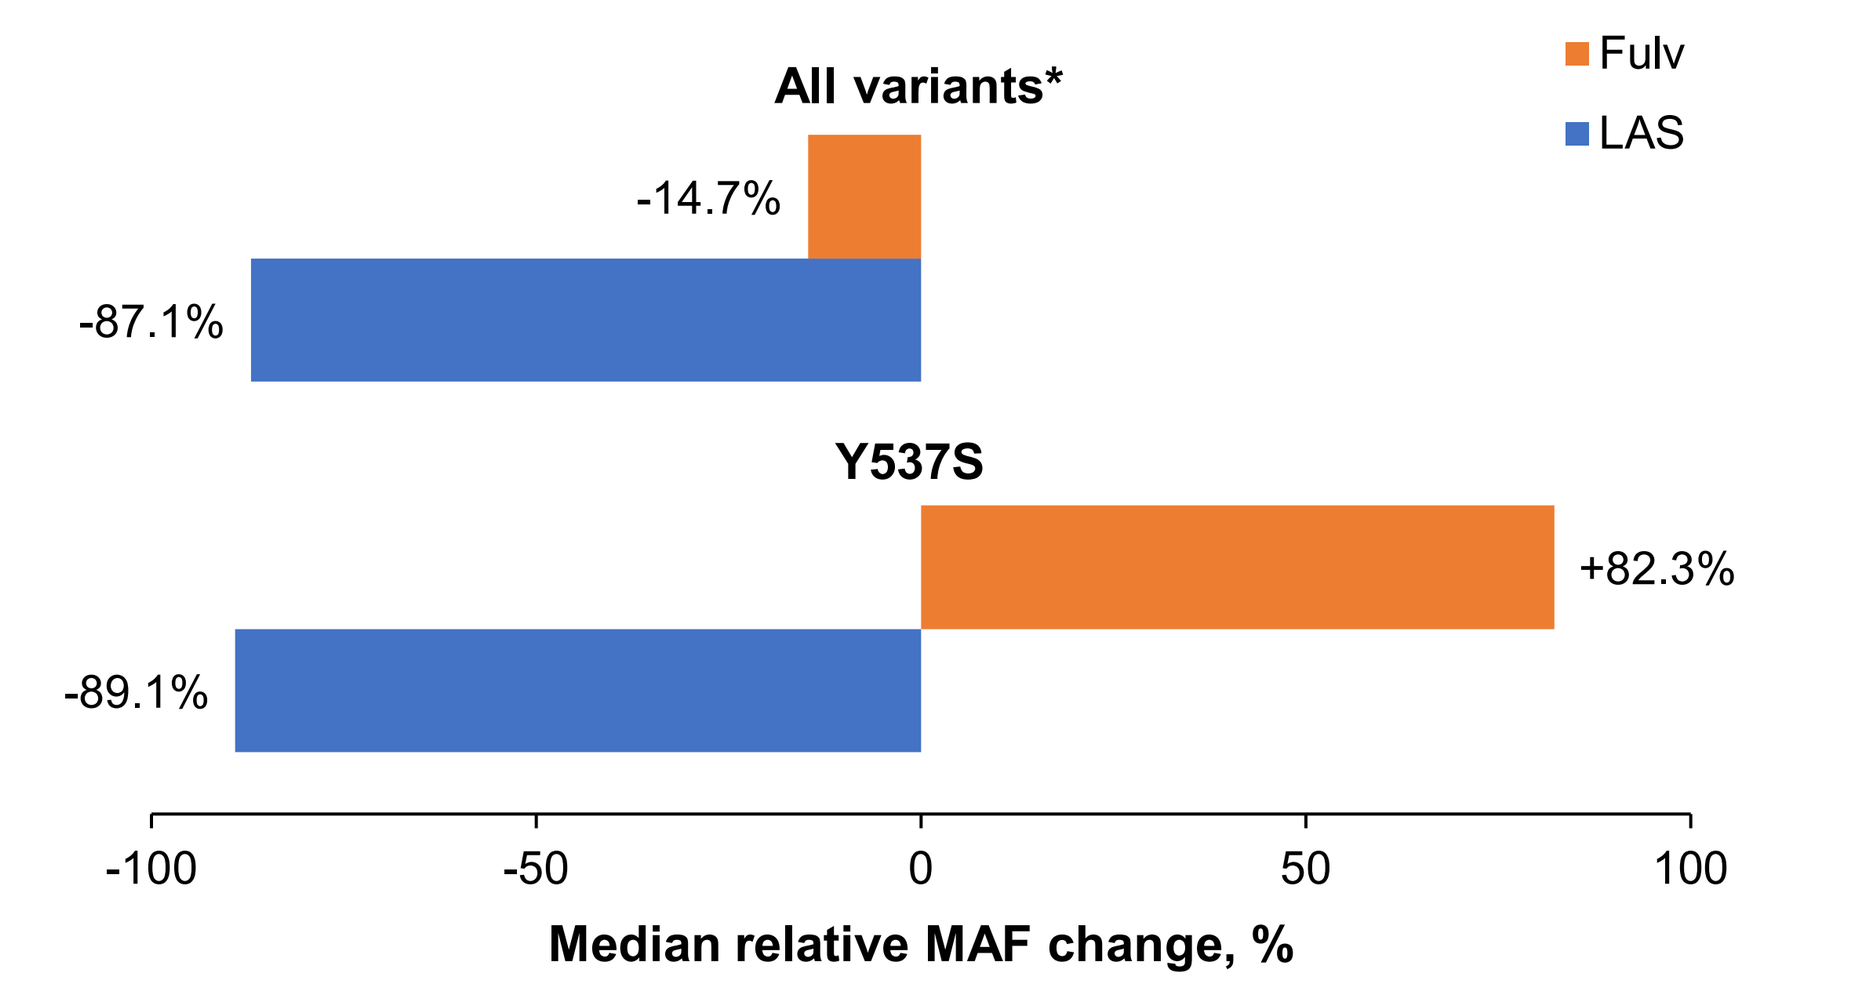
Fulv
LAS
All variants*
-14.7%
-87.1%
Y537S
+82.3%
-89.1%
Median relative MAF change, %

*In polyclonal patients, % change was calculated for aggregate *ESR1* MAF

Table 3. Change from baseline to week 8 in *ESR1* MAF and efficacy outcomes

	Lasofoxifene (n=35)		Fulvestrant (n=26)	
	Decreased/ND MAF	Increased MAF	Decreased/ND MAF	Increased MAF
Median PFS (95% CI), mos	8 (2–11)	4 (2–16.5)	4.5 (3–10)	2.8 (0.5–6)
Clinical benefit at week 24				
Yes, n (%)	16 (45.7)	1 (2.9)	4 (15.4)	1 (3.9)
No, n (%)	13 (37.1)	5 (14.3)	12 (46.2)	9 (34.6)
Sensitivity (95% CI), %	94.1 (73.0–99.0)		80.0 (29.9–98.9)	
Specificity (95% CI), %	27.8 (11.8–48.8)		42.9 (22.6–65.6)	

Clinical benefit: CR, PR, or ≥24 weeks of SD or non-CR/Non-PD as the patient's best overall response during the study

Figure 3. Individual MAF kinetics for the most commonly detected *mESR1* variants

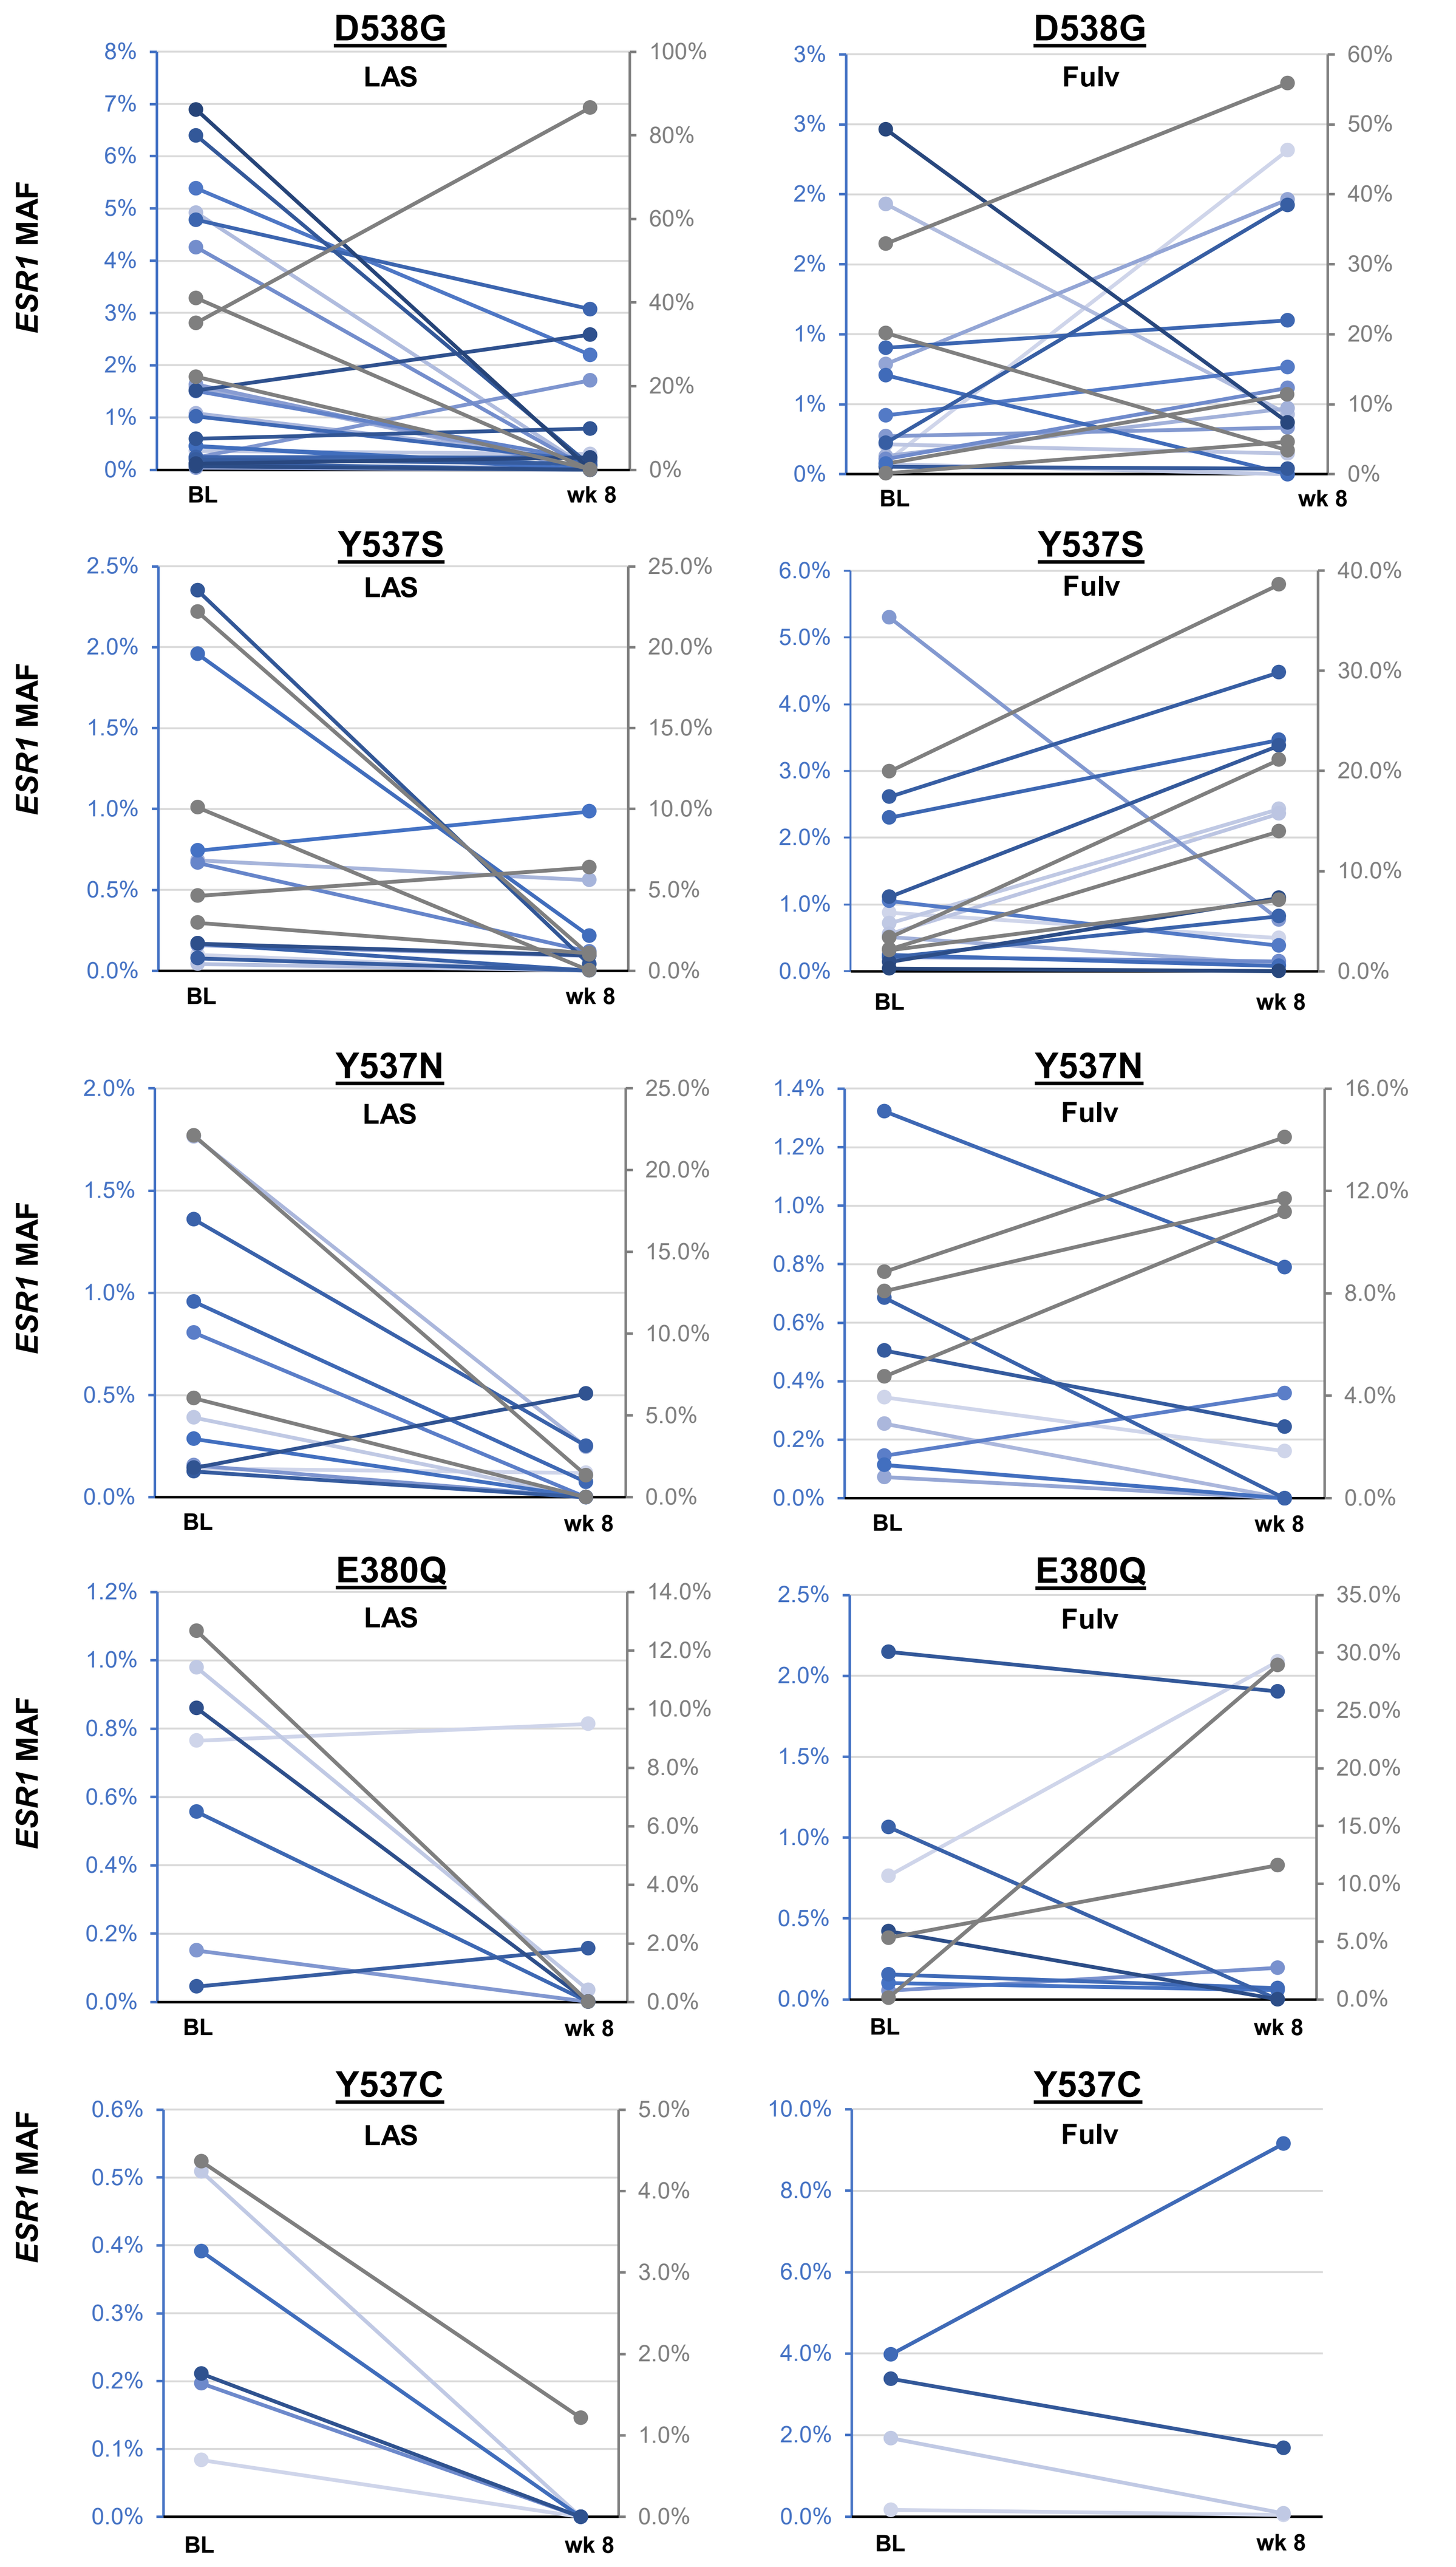
D538G
LAS
D538G
Fulv
Y537S
LAS
Y537S
Fulv
Y537N
LAS
Y537N
Fulv
E380Q
LAS
E380Q
Fulv
Y537C
LAS
Y537C
Fulv
ESR1 MAF
BL
wk 8

Variants with low MAF (any blue) utilize the left vertical axis and those with high MAF (gray) utilize the right vertical axis.

Abbreviations

AI: aromatase inhibitor; BL: baseline; CBR: clinical benefit rate; CDK4/6i: cyclin-dependent kinase 4/6 inhibitor; CR: complete response; ctDNA: circulating tumor DNA; Fulv: fulvestrant; ITT: intent to treat; LAS: lasofoxifene; MAF: mutant allele fraction; *mESR1*: *ESR1* mutation; ND: none detected (clearance); PFS: progression-free survival; PPV, positive predictive value; PR: partial response; SERM: selective estrogen receptor modulator; SERD: selective estrogen receptor degrader; SD: stable disease.

References

1. Fan P, et al. *Cancer Drug Resist*. 2019;2:198-209. **2.** De Santo I, et al. *Cancers (Basel)*. 2019;11:1894. **3.** Brett JO, et al. *Breast Cancer Res*. 2021;23:85. **4.** Herzog SK, et al. *Br J Cancer*. 2022;126:174-186. **5.** Goetz MP, et al. *Annals of Oncology*. 2022;33:S1387-S1388. **6.** Cristofanilli MA, et al. Poster presented at *the 4th Congress of the International Society of Liquid Biopsy*; Miami, FL. Poster PP. 38. **7.** Gerratana L, et al. *JCO Precis Oncol*. 2021;5:943-952. **8.** Garcia-Saenz JA, et al. *BMC Cancer*. 2017;17:210.

Key Takeaways

- Treatment with LAS effectively decreased or cleared *mESR1* from ctDNA, including the difficult-to-treat Y537S variant
- Decreased/cleared *mESR1* was associated with numerically prolonged PFS and higher CBR in the LAS arm but not the Fulv arm

Conclusions

- Our ctDNA data show that *mESR1* levels decreased or cleared in numerically more patients treated with LAS than with Fulv in women with ER+/HER-, *mESR1* mBC, including the Y537S mutant, which is known to be resistant to Fulv[3]
- Decreased/cleared *ESR1* MAF was associated with a prolonged PFS and higher CBR in the LAS arm but not the Fulv arm
- Our results demonstrate target engagement of LAS with *mESR1*
- These data suggest that *ESR1* MAF could potentially be a liquid biomarker for predicting response to LAS monotherapy in *mESR1*, endocrine-resistant mBC patients previously treated with an AI plus CDK4/6i
- The association between ctDNA *ESR1* MAF changes and clinical response to LAS plus abemaciclib was also explored in the ELAINE 2 study (see poster **P5-05-02**)

Disclosures

- **MPG** has CME activities from Research to Practice, Clinical Education Alliance, and Medscape; was a panelist for Total Health Conferencing; a moderator for Curio Science; has done consulting for AstraZeneca, Biovica, Biotheranostics, Blueprint Medicines, Eagle Pharmaceuticals, Eli Lilly, Novartis, Pfizer, Sanofi Genzyme, and Sermonix; has received research support from Eli Lilly, Pfizer, and Sermonix. **ENG** has received honoraria from Eli Lilly, Novartis, Pfizer, Roche, AstraZeneca, and MSD. **DGS** is on the advisory board for Novartis. **SLS** has received research funding (paid to institution) from Astra Zeneca, Abbvie, Bristol Myers Squibb, Eli Lilly, Seagen, and Sermonix; consults for Foundation Medicine, AstraZeneca, Daichii Sankyo, Eli Lilly, Pfizer, Sermonix, and Novartis. **SLG** is an advisor for Daiichi Sankyo, AstraZeneca, Novartis, Pfizer, Genentech, Seagen, Eli Lilly and Gilead Sciences. **GW** has received research support from Daiichi Sankyo, Eisai, Eli Lilly, Gilead Sciences, Pfizer, and Seagen. **MAC** has done consulting for Menarini, Eli Lilly, AstraZeneca, Ellipses, Celcuity, Olaris, and Sermonix; has received research support from Pfizer, Eli Lilly and AstraZeneca; and honoraria from Pfizer, Eli Lilly, Foundation Medicine, and Guardant. **GR** is a consultant and stock holder of Sermonix, NuProbe, and Arcadia Medicine. **HSS** is a former employee of Sysmex Inostics. **DC** consults with Sermonix. **PVP** and **DJP** are employees and stockholders of Sermonix.
- Sermonix Pharmaceuticals sponsored the study and provided support for the medical writing assistance of Hui Zhang, PhD (Precise Publications, LLC).

This presentation is the intellectual property of the Sermonix Pharmaceuticals. Contact David J Portman at Dportman@sermonixpharma.com for permission to reprint and/or distribute.