Durable Complete Remission of Metastatic ER+/HER2− Breast Cancer after Lasofoxifene Therapy

Einav Nili Gal-Yam, MD, PhD[1], PhD; David Portman[2]; Keren Levanon, MD, PhD[3]

[1]*Institute of Oncology, Chaim Sheba Medical Center, Ramat Gan, Israel*
[2]*Sermonix Pharmaceuticals, Columbus, Ohio*
[3]*Sackler Faculty of Medicine, Tel-Aviv University, Ramat Aviv, Israel*

Introduction

- Endocrine therapy (ET), particularly with aromatase inhibitors (AIs), reduces estrogen receptor (ER) activity and has been the mainstay for treating ER+ breast cancer
- Long-term ET often leads to treatment resistance caused by acquired *ESR1* mutations[1,2]
 - *ESR1* mutations result in a constitutively active (ligand independent) ER leading to AI resistance, tumor progression, and overall poor prognosis[3,4]
- LAS, a selective estrogen receptor modulator (SERM), has shown potent activity against *ESR1* mutants alone or in combination with CDK4/6 inhibitor (CDK4/6i) compared to fulvestrant ([Fulv], a selective estrogen degrader [SERD]) in metastatic breast cancer (mBC) xenograft models expressing *ESR1* mutations[5,6]
- LAS modifies the constitutive conformation of the mutated *ERα* to an antagonist conformation, thereby inactivating the receptor[6]
- The ELAINE 1 study (NCT03781063) comparing LAS and Fulv in patients progressing after CDK4/6i and AIs has recently been completed (submitted to ESMO 2022)

Case Study Objectives

- To report a clinical case with metastatic ER+/HER2− breast cancer who demonstrated a durable CR after lasofoxifene treatment.
- To provide a comprehensive evaluation of a clinical case of metastatic ER+/HER2− breast cancer with radiographic imaging and physical exams confirming complete regression of MBC including complete clearance of blood ESR1 mutant circulating tumor DNA (ctDNA).

Materials & Methods

- **Patient History**. The patient at age 49 was diagnosed with a stage 1 T1cN0 G3 ER and PR positive, HER2-negative breast cancer in 2007. She was pre-menopausal at the time of diagnosis and had non- remarkable medical history. Her oncotype RS was 30, and she received adjuvant dose dense Doxorubicin plus Cyclophosphamide treatment every 2 weeks for 4 cycles. This was followed by adjuvant radiation to breast and endocrine treatment with Tamoxifen for 5 years. In 2015, due to increasing back pain and elevated tumor markers, a pet-FGD scan was performed which revealed pathologic uptake in pleural thickening in the left hemithorax, para-mediastinal, cardiophrenic and internal mammary lymph nodes, a mass in the lung left upper lobe and an intermediate left pleural effusion. A core needle biopsy from a pleural lesion revealed cores of carcinoma compatible with metastasis of breast origin, positive for ER, PR and negative HER2. She started on Letrozole 2.5 mg plus Palbociclib 125mg daily in January 2016 with excellent clinical and complete radiological response. Palbociclib dose was reduced to 100mg per day after a year of treatment due to hematologic toxicity. At the end of 2020, nearly 5 years after starting treatment, successive scans revealed new extensive left pleural lesions and a pathologic lesion in the right ileum. ESR1 mutation analysis was perform on the patients ctDNA to reveal hot spot Y537C and D538G ESR1 mutations.

- **Laboratory**. ESR1 ctDNA analysis was conducted at Sysmex Inostics, Inc. clinical laboratory in Baltimore, Maryland using their CLIA validated SafeSeq Breast Cancer test. The SafeSEQ Breast Cancer CDx is a next generation sequencing based clinical trial assay for the qualitative detection of somatic mutations in AKT1, ERBB2, ESR1, KRAS, TP53 and PIK3CA genes using circulating tumor DNA (ctDNA) from plasma derived from peripheral whole blood obtained from patients with breast cancer.

Results

Figure 1. ELAINE 1 (NCT03781063): Study Design.

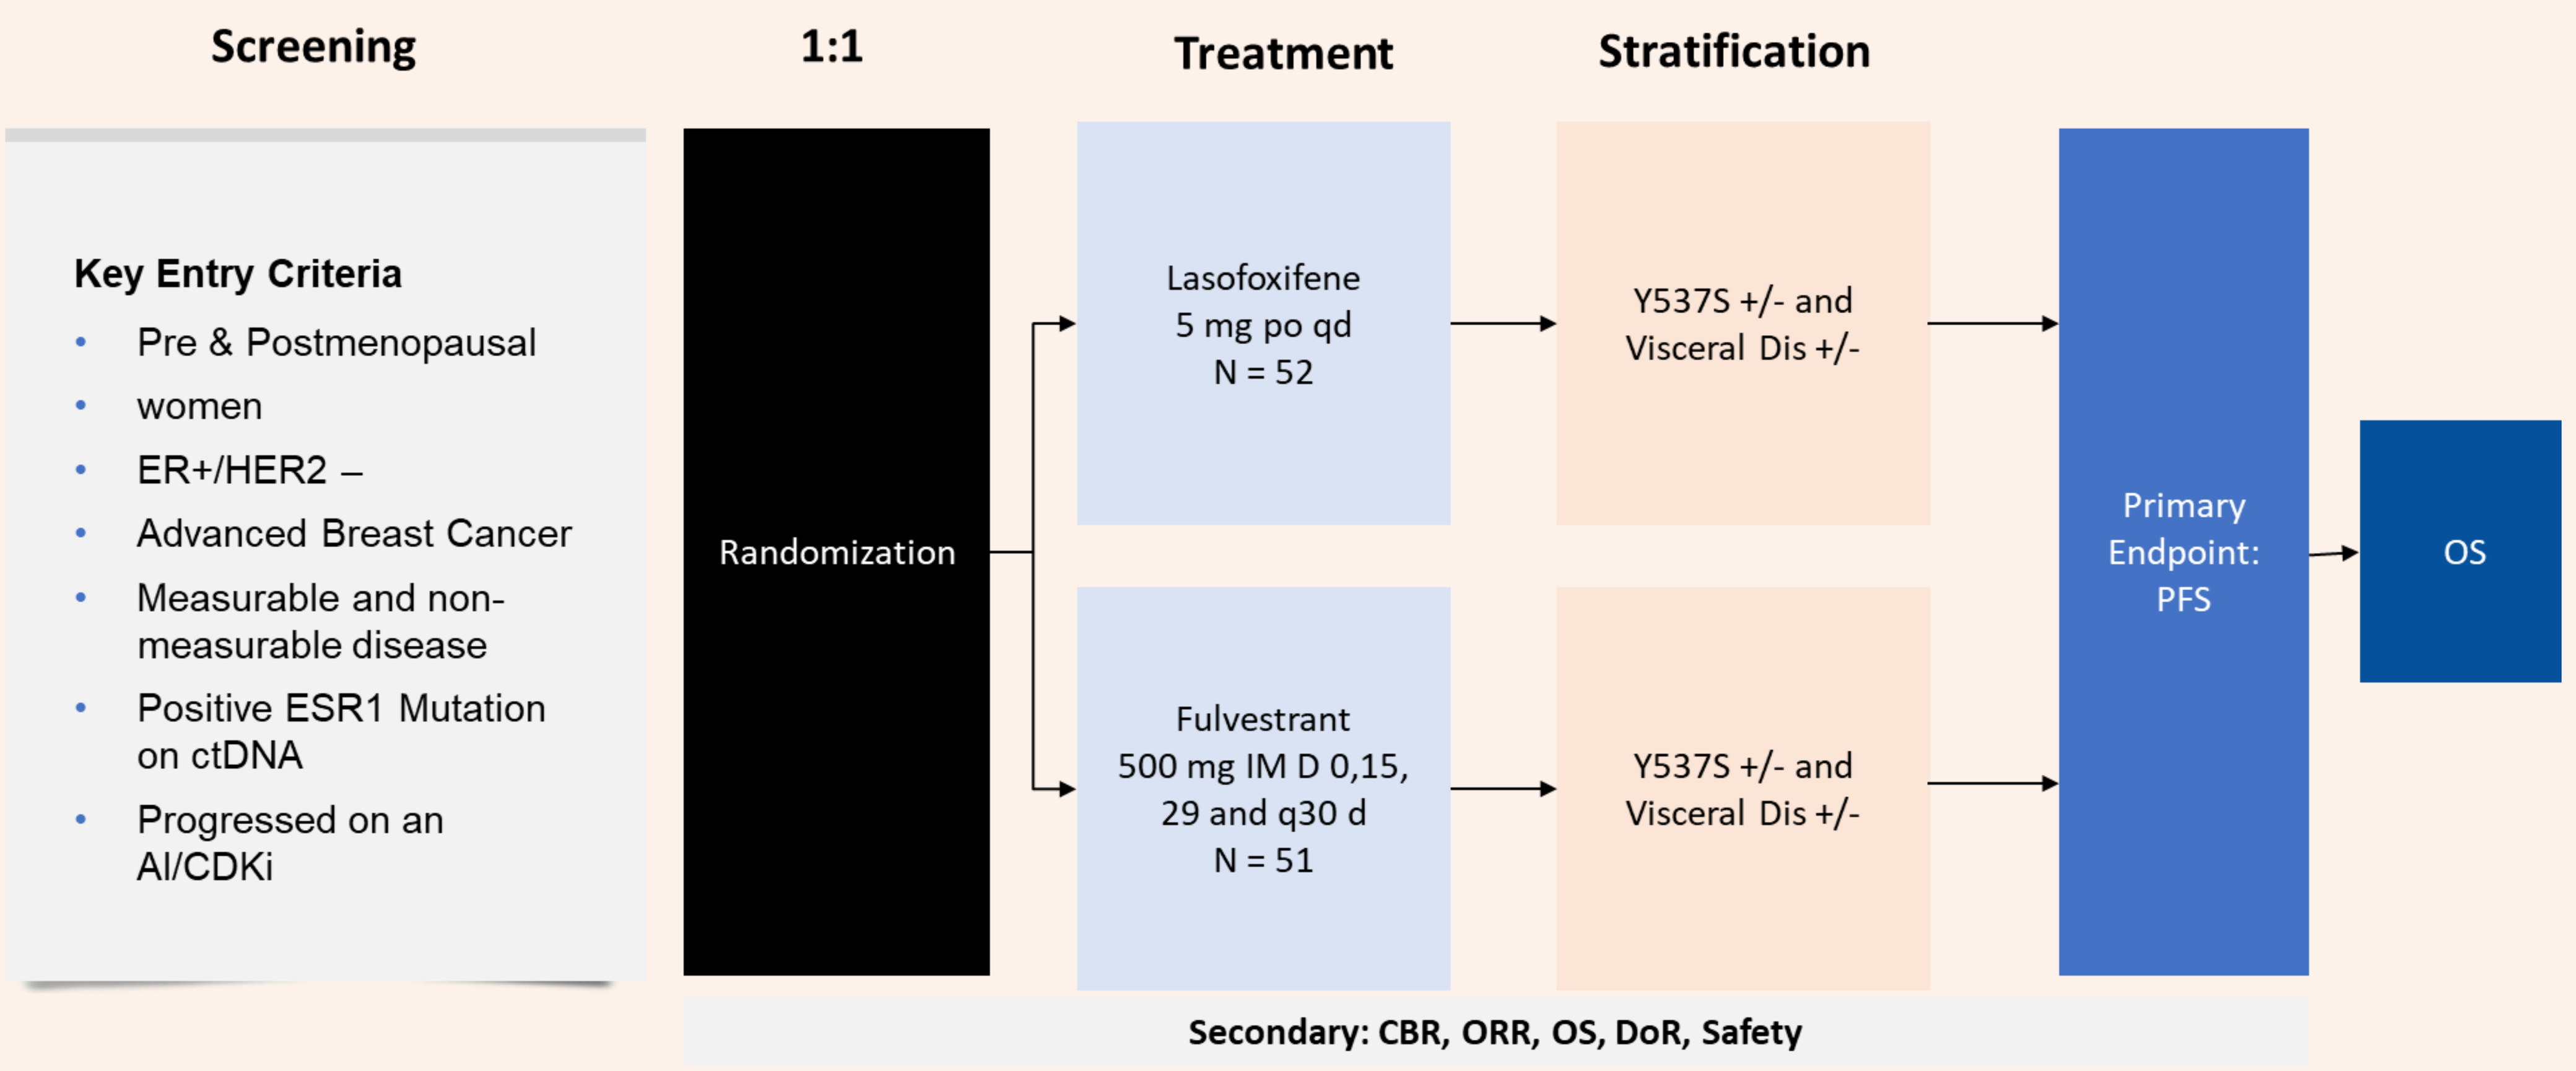
Screening
1:1
Treatment
Stratification
Key Entry Criteria
Pre & Postmenopausal
women
ER+/HER2 –
Advanced Breast Cancer
Measurable and non-measurable disease
Positive ESR1 Mutation on ctDNA
Progressed on an AI/CDKi
Randomization
Lasofoxifene
5 mg po qd
N = 52
Fulvestrant
500 mg IM D 0,15, 29 and q30 d
N = 51
Y537S +/- and Visceral Dis +/-
Y537S +/- and Visceral Dis +/-
Primary Endpoint: PFS
OS
Secondary: CBR, ORR, OS, DoR, Safety

Source: ELAINE I clinical trial (NCT03781063); Accessed at: https://clinicaltrials.gov/ct2/show/NCT03781063.

Clinical Case Patient Characteristics.

63 y.o. female with mBC to pleura and LNs; 4 yrs 11 months on palbo/letrozole then progression with extensive pleural lesions, pleural effusion

Enrolled in Elaine 1 study and randomized to lasofoxifene

Currently at 88 wks with confirmed CR (by blinded independent review) with 72-week duration of ongoing response

- Target pleural lesions sum diameter 30 mm (16mm and 14mm), also non-target pleural lesions; target lesions decreased 55.3% to 14mm (11mm/3mm) at 8 weeks; absent (0/0) at 16 wks, non-target lesions also absent
- ctDNA MAF at baseline:
 - D538G 0.243%
 - Y537C 0.169%
 - None detected at 72 weeks

Figure 2. Documented Complete Response from ELAINE 1 Study.

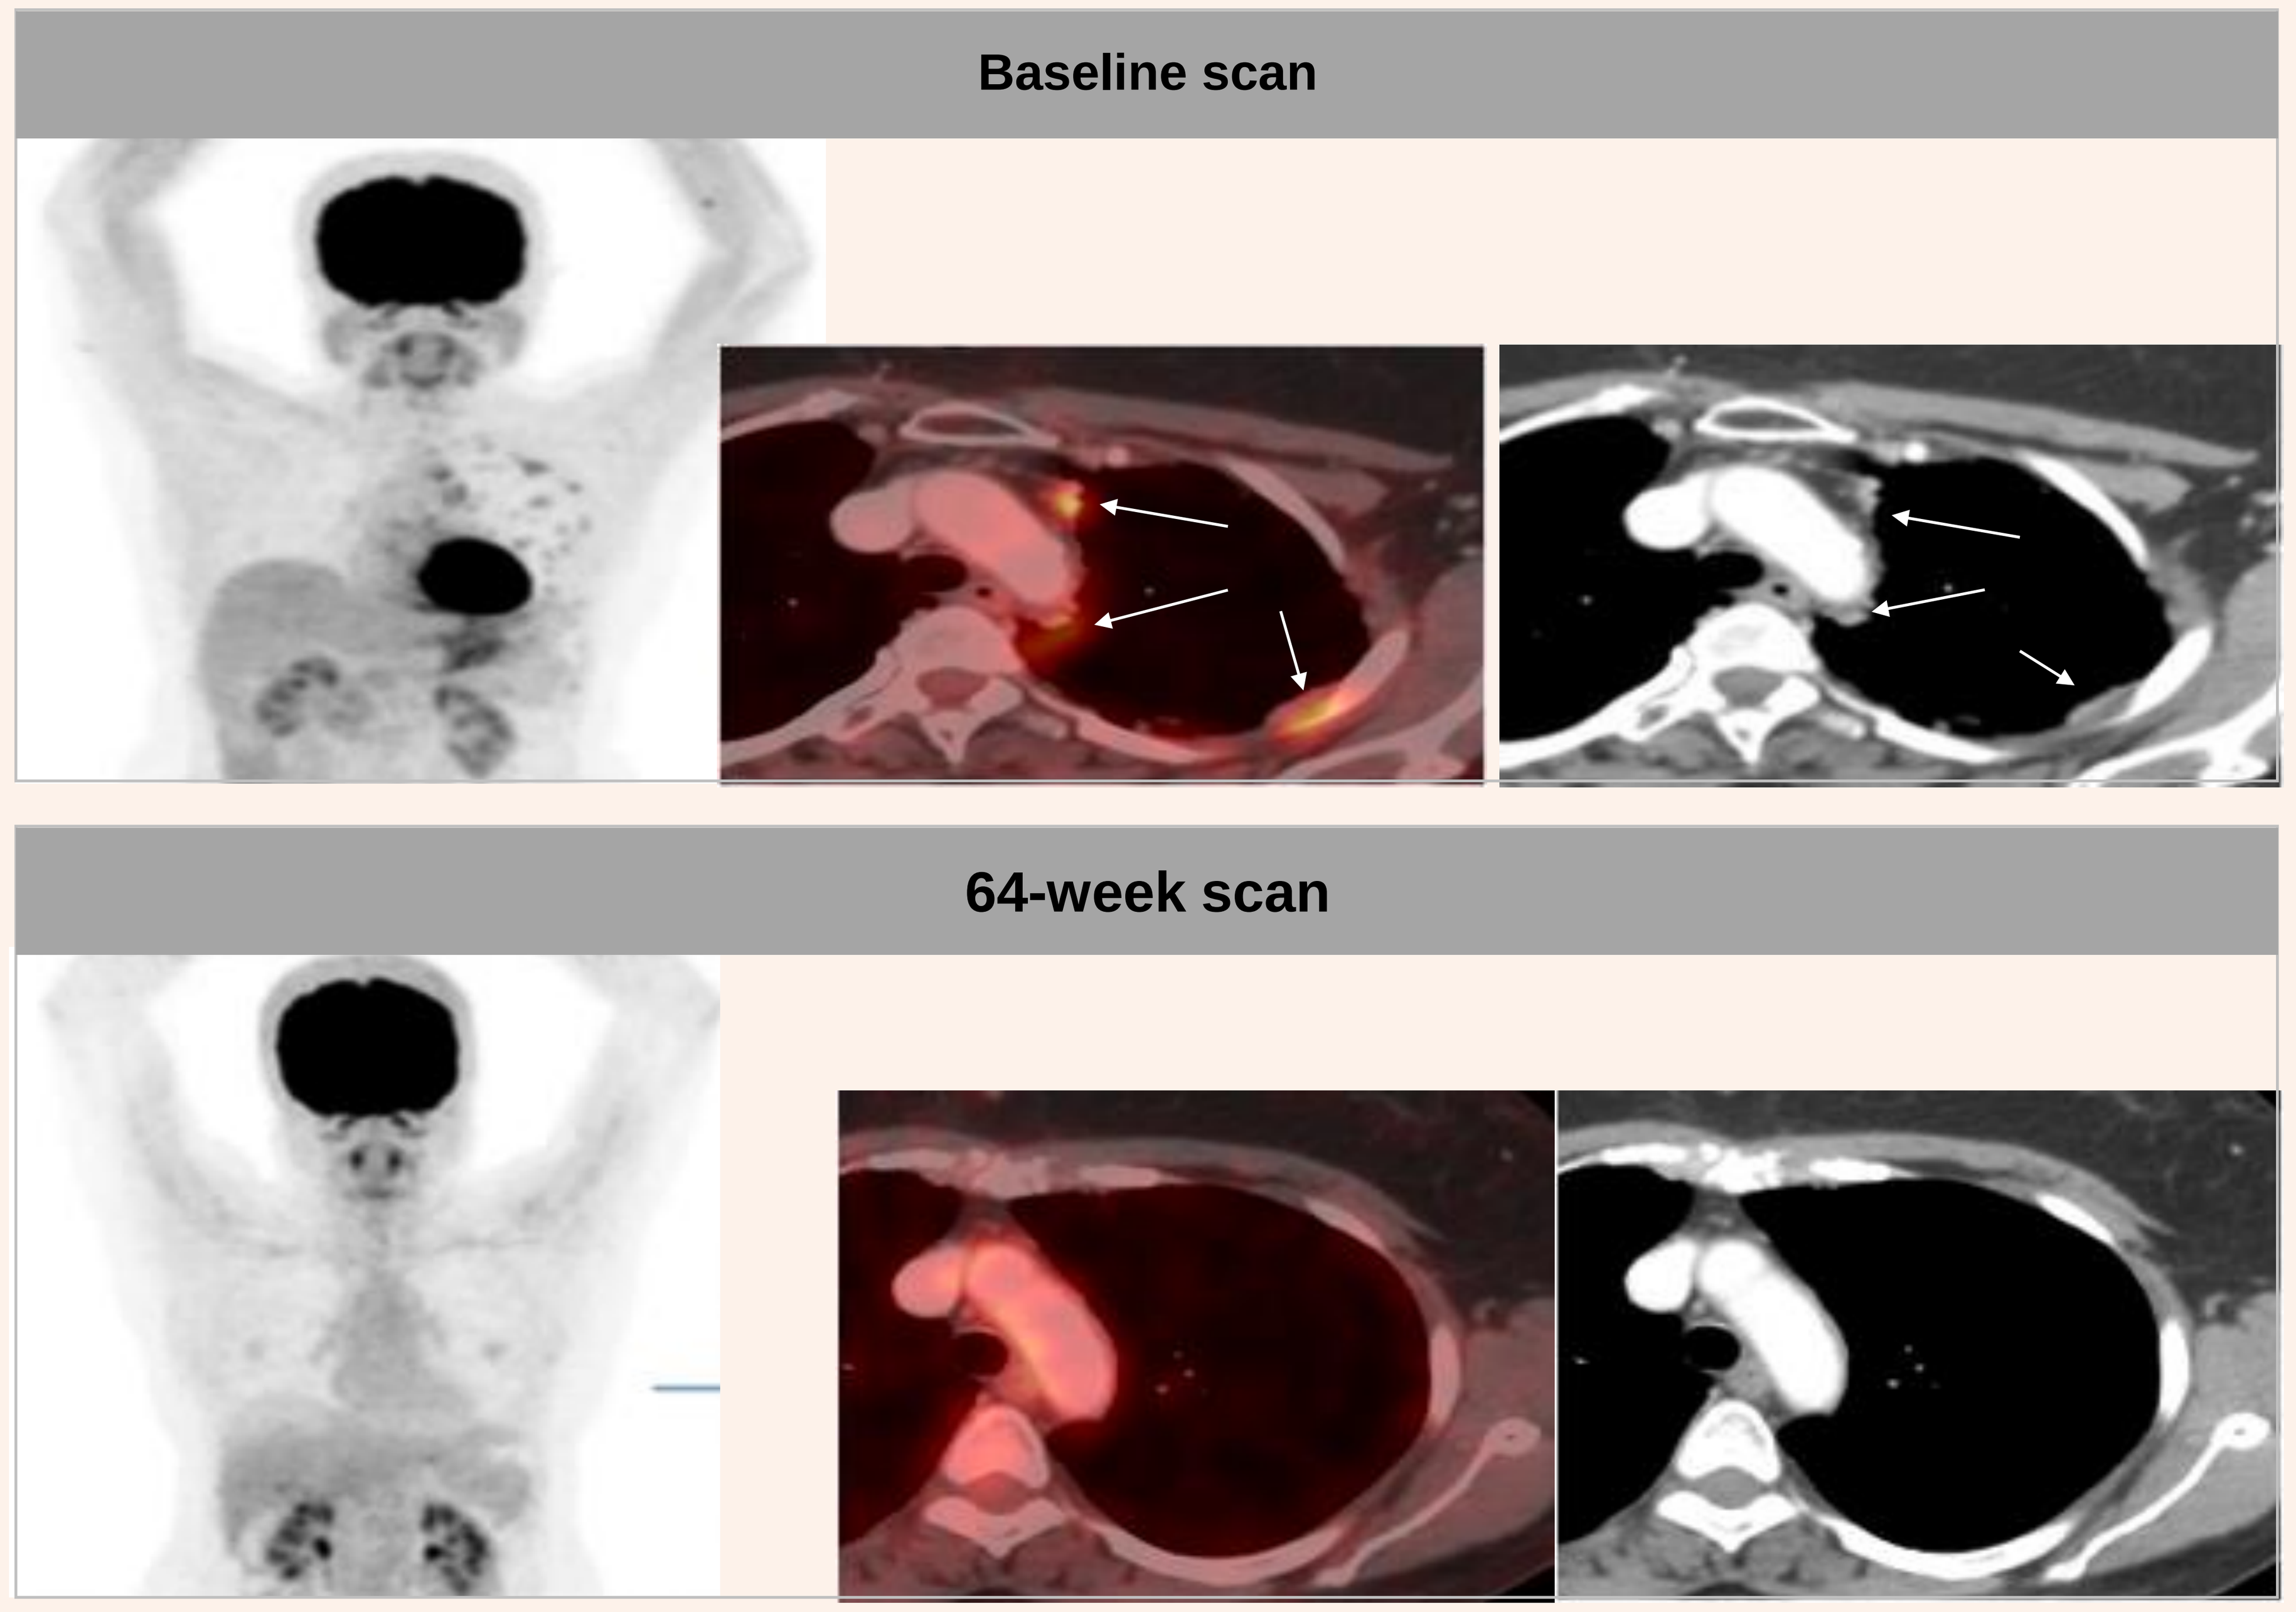
Baseline scan
64-week scan

Source: Data on file. Sermonix Pharmaceuticals, Inc.

Figure 3. Mutant Allele Fraction (MAF) at Baseline and 8-weeks for patients on Lasofoxifene in Elaine 1: Clearance of ctDNA in CR of these ESR1mut variants is consistent with target engagement.

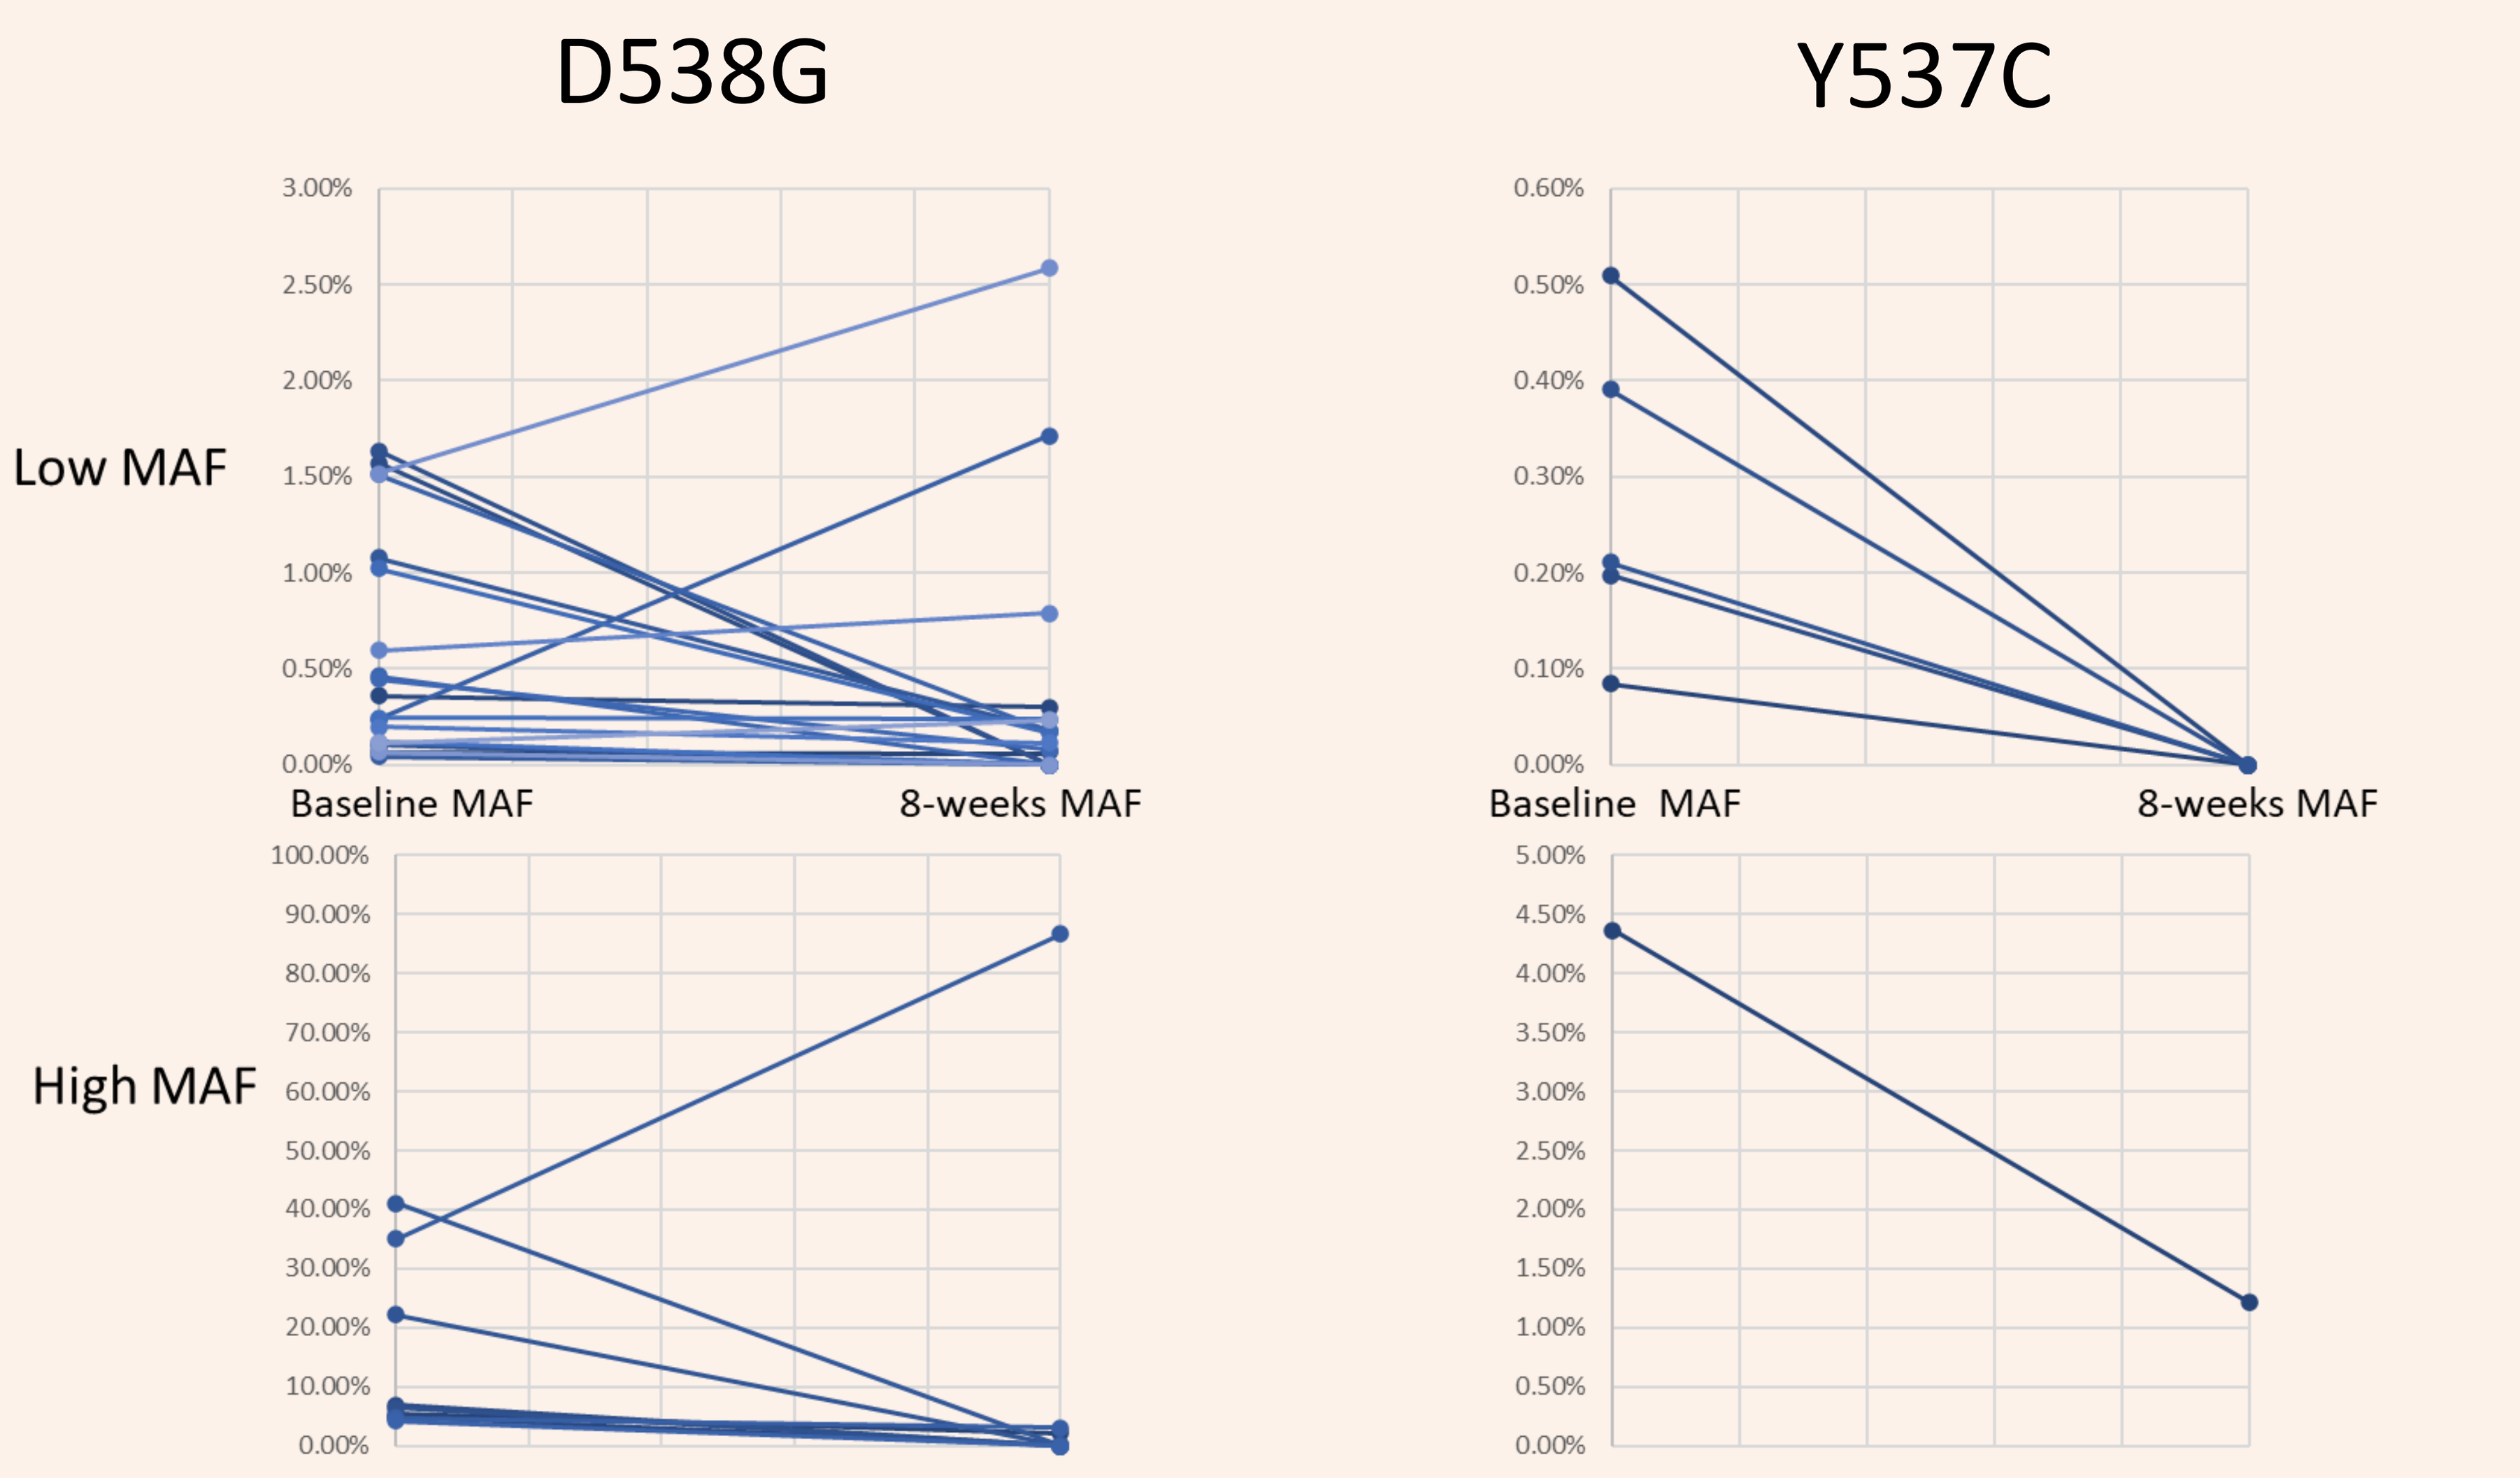
D538G
Y537C
Low MAF
High MAF
Baseline MAF
8-weeks MAF
Baseline MAF
8-weeks MAF
3.00%
2.50%
2.00%
1.50%
1.00%
0.50%
0.00%
0.60%
0.50%
0.40%
0.30%
0.20%
0.10%
0.00%
100.00%
90.00%
80.00%
70.00%
60.00%
50.00%
40.00%
30.00%
20.00%
10.00%
0.00%
5.00%
4.50%
4.00%
3.50%
3.00%
2.50%
2.00%
1.50%
1.00%
0.50%
0.00%

Source: Data on file. Sermonix Pharmaceuticals, Inc. Note: Scales adjusted to accommodate for wide range of MAF seen.

Key Conclusions

- Achieving a CR with endocrine treatment of MBC in the post CDK4/6i setting is uncommon, particularly with single-agent endocrine therapy.
- Patients acquiring ESR1 mutations develop relative endocrine resistance due to the constitutively active estrogen receptor.
- This 63-year-old postmenopausal woman had previously progressed on an AI in combination with a CDK 4/6 inhibitor. Upon progression, she was found to have ESR1 mutation. Given her visceral disease and the presence of an ESR1 mutation, her prognosis was not favorable.
- This case is the first report of any single-agent hormonally-based regimen to achieve a durable complete clinical remission in a metastatic ER+/HER2− breast cancer patient with an ESR1 mutation.
- Mutations in the ESR1 gene have emerged as an important driver of resistance to endocrine therapies, which form the backbone of treatment for patients with ER+/HER2 breast cancer.
- The notable clinical response and clearance of ESR1mut ctDNA in this case report suggests that lasofoxifene may potentially play a significant role as a targeted therapy in addressing the unmet medical need for patients with acquired ESR1 mutations.

Disclosures

- Sermonix Pharmaceuticals sponsored the study and provided support for the medical writing assistance of Joe Petroziello (MJH Life Sciences).

References

1. Fan P, et al. Cancer Drug Resist. 2019;2:198-209. **2.** De Santo I, et al. Cancers (Basel). 2019;11:1894. **3.** Brett JO, et al. Breast Cancer Res. 2021;23:85. **4.** Herzog SK, et al. Br J Cancer. 2022;126:174-186. **5.** Andreano KJ, et al. Mol Cancer Ther. 2020;19:1395-1405. **6.** Lainé M, et al. Breast Cancer Research. 2021;23:54.

Annual Metastatic Breast Cancer Research Conference (MBCRC)
September 7-9, 2022, in Park City, Utah