Lasofoxifene as a Potential Treatment for Aromatase Inhibitor Resistant ER+ Breast Cancer

Muriel Lainé [1], Marianne Greene [1], Tiffany Leng [1], Justyna Kurleto [1], Sophia Li [1], Barry Komm [2] and Geoffrey L. Greene [1]

[1] Ben May department, University of Chicago, Chicago Il

[2] Sermonix Pharmaceuticals

Introduction

In postmenopausal women breast cancer is typically treated with endocrine therapy or estrogen depravation, which is achieved with aromatase inhibitors (AI) such as letrozole. The AI approach have been shown to be superior to tamoxifen treatment but in 20% of cases, patients are resistant to AI treatment or undergo relapse. Women that are resistant to AI therapy are treated with fulvestrant or a combination of fulvestrant and a CDK4/6 inhibitor as a second line of therapy. We have previously demonstrated in a xenograph model of ET resistant mutant ER+ metastatic breast cancer (MBC) that lasofoxifene alone and in combination with palbociclib was superior to fulvestrant or a combination of fulvestrant+palbociclib. In the current study, we compared the efficacy of lasofoxifene +/- palbociclib to fulvestrant +/- palbociclib combinations in a letrozole-induced, AI-resistant breast tumor model (MCF-7 LTLT cells) that does not express ERα activating mutations.

Results

Lasofoxifene alone and in combination with palbociclib significantly slows primary tumor progression

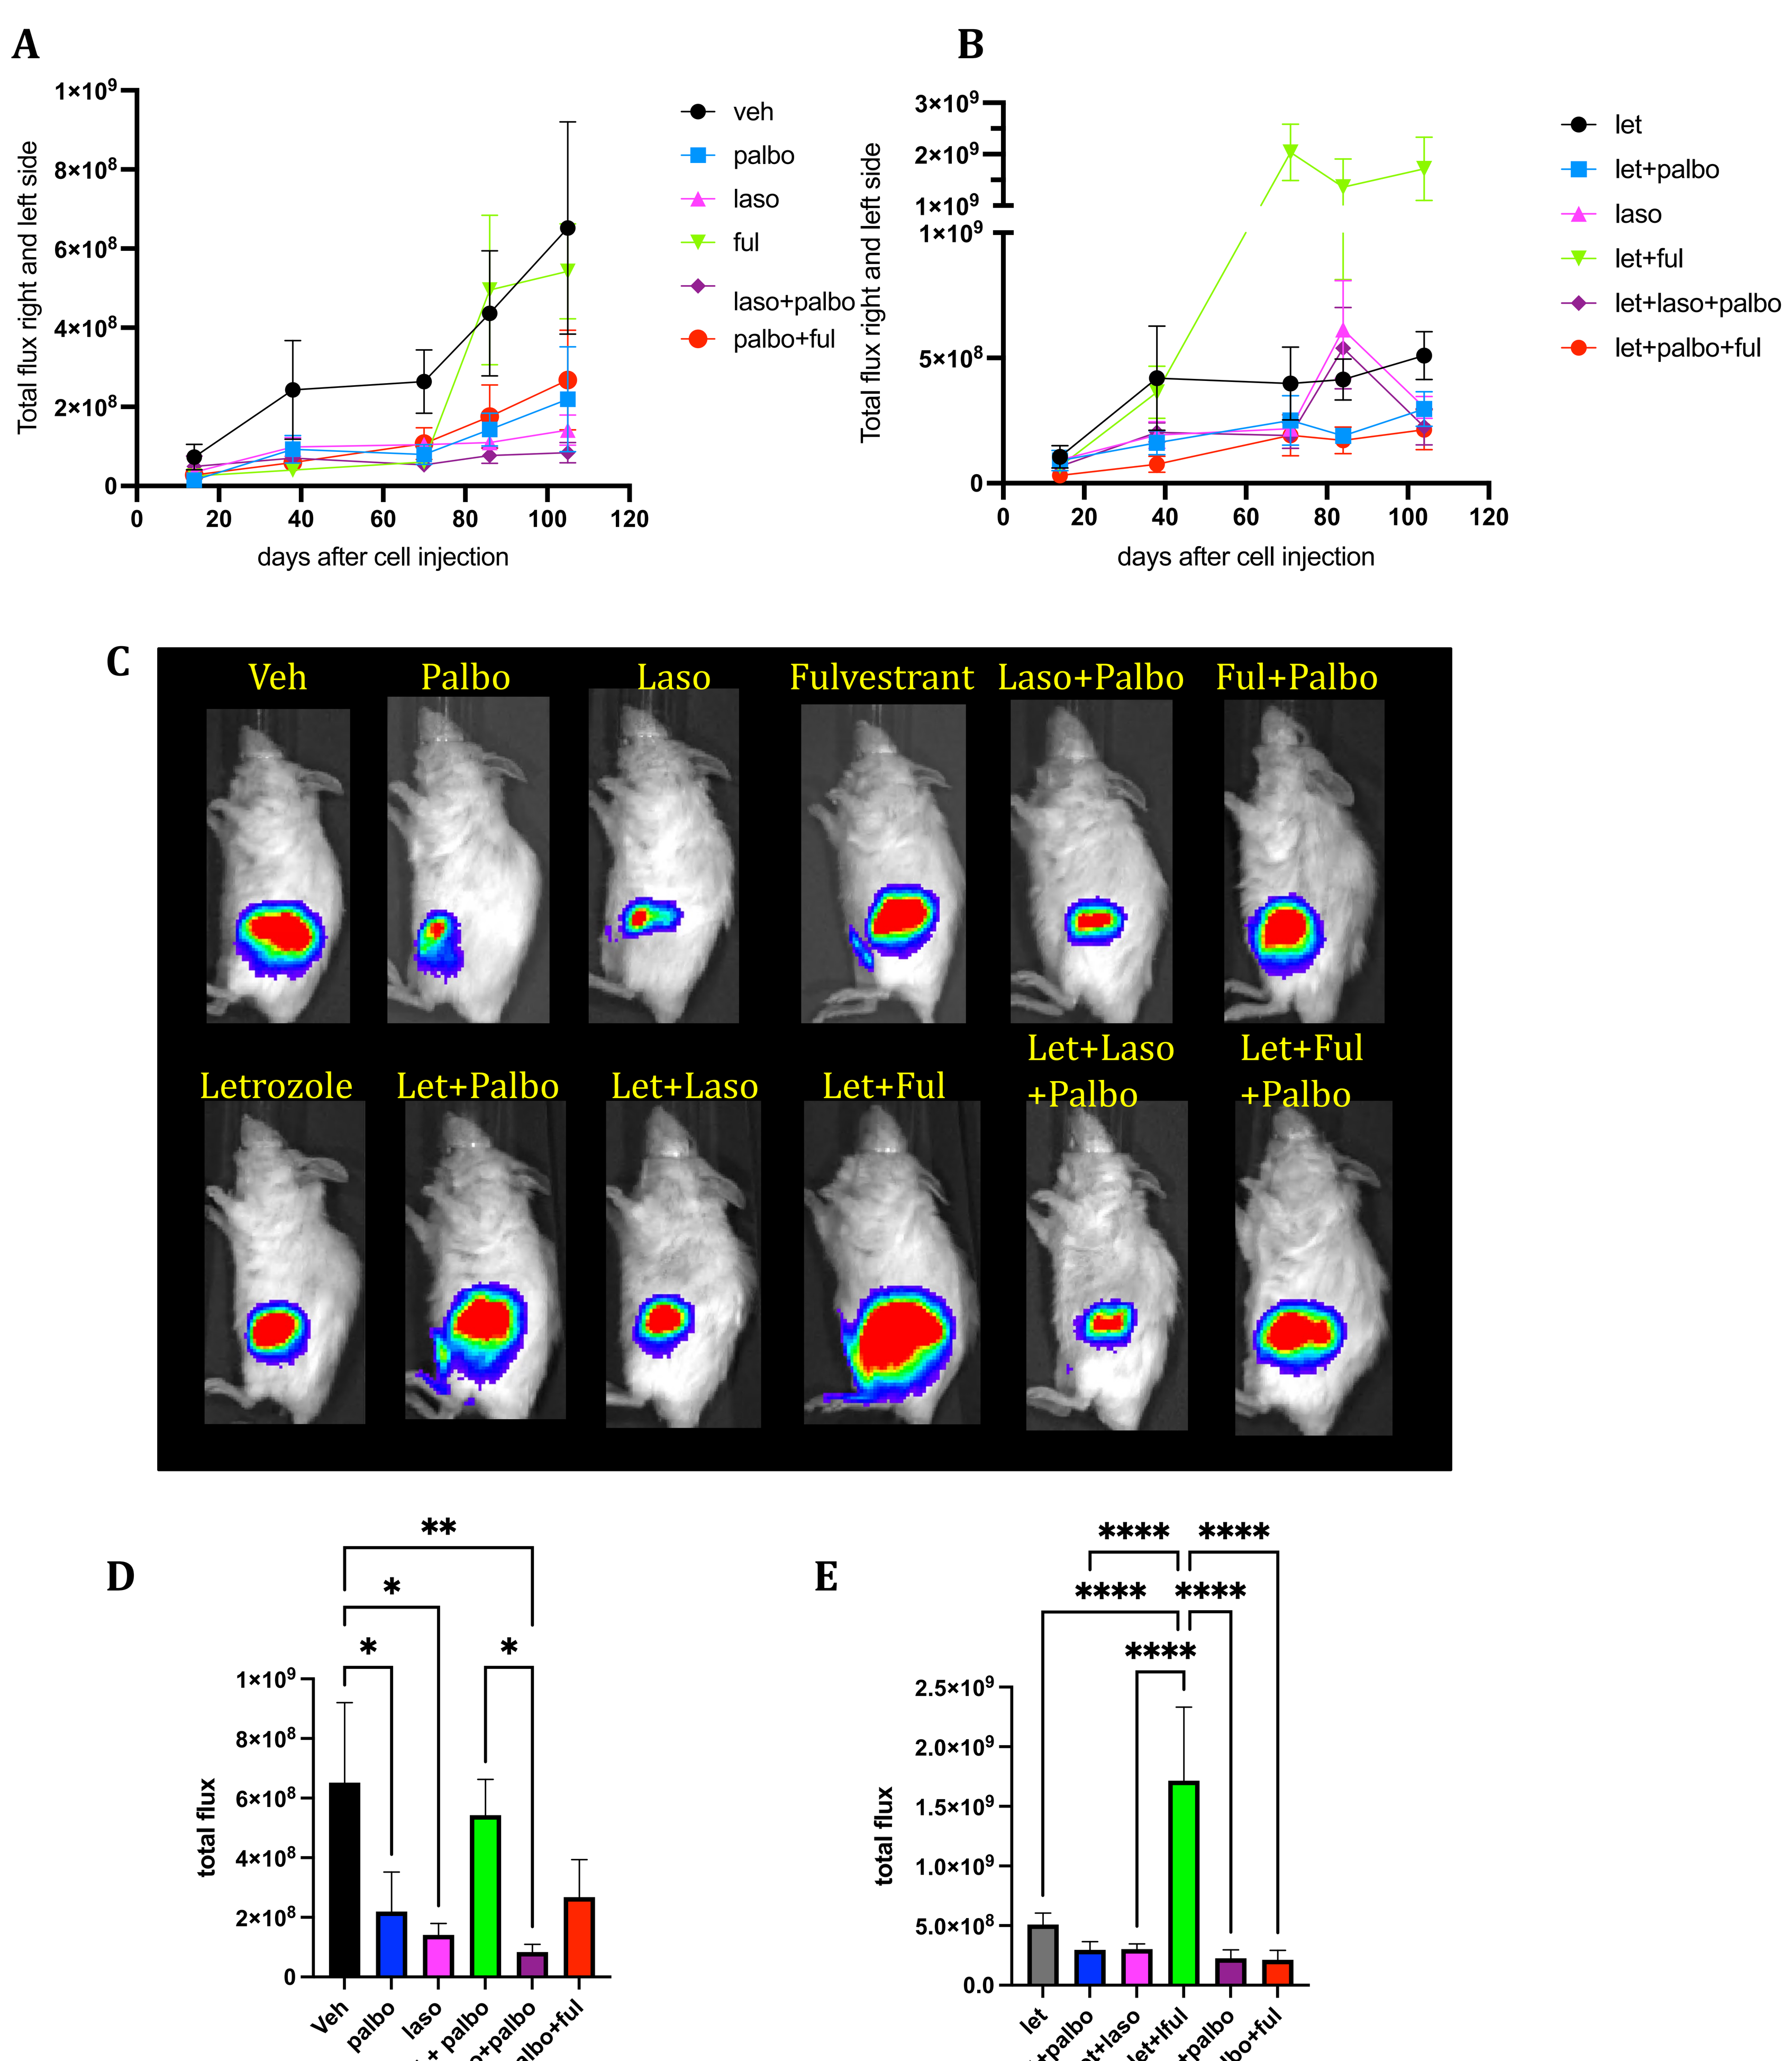
A
B
Total flux right and left side
days after cell injection
veh
palbo
laso
ful
laso+palbo
palbo+ful
let
let+palbo
let+ful
let+laso+palbo
let+palbo+ful
C
Veh
Palbo
Laso
Fulvestrant
Laso+Palbo
Ful+Palbo
Letrozole
Let+Palbo
Let+Laso
Let+Ful
Let+Laso +Palbo
Let+Ful +Palbo
D
E
total flux
ful + palbo
let+laso

Fig 1: A & B, Quantification of total photon flux measure over time in Xenogen IVIS imager. **C**, representative images at 11 weeks after start of treatment. **D & E**, Histograms of total photon flux for each group at the study end point. P values are calculated by one way Anova *p<0.05, **p<0.001, ***p<0.001, ****p<0.0001. N=3 to 6

Lasofoxifene + palbociclib combination significantly reduces tumor burden compared to palbociclib + fulvestrant

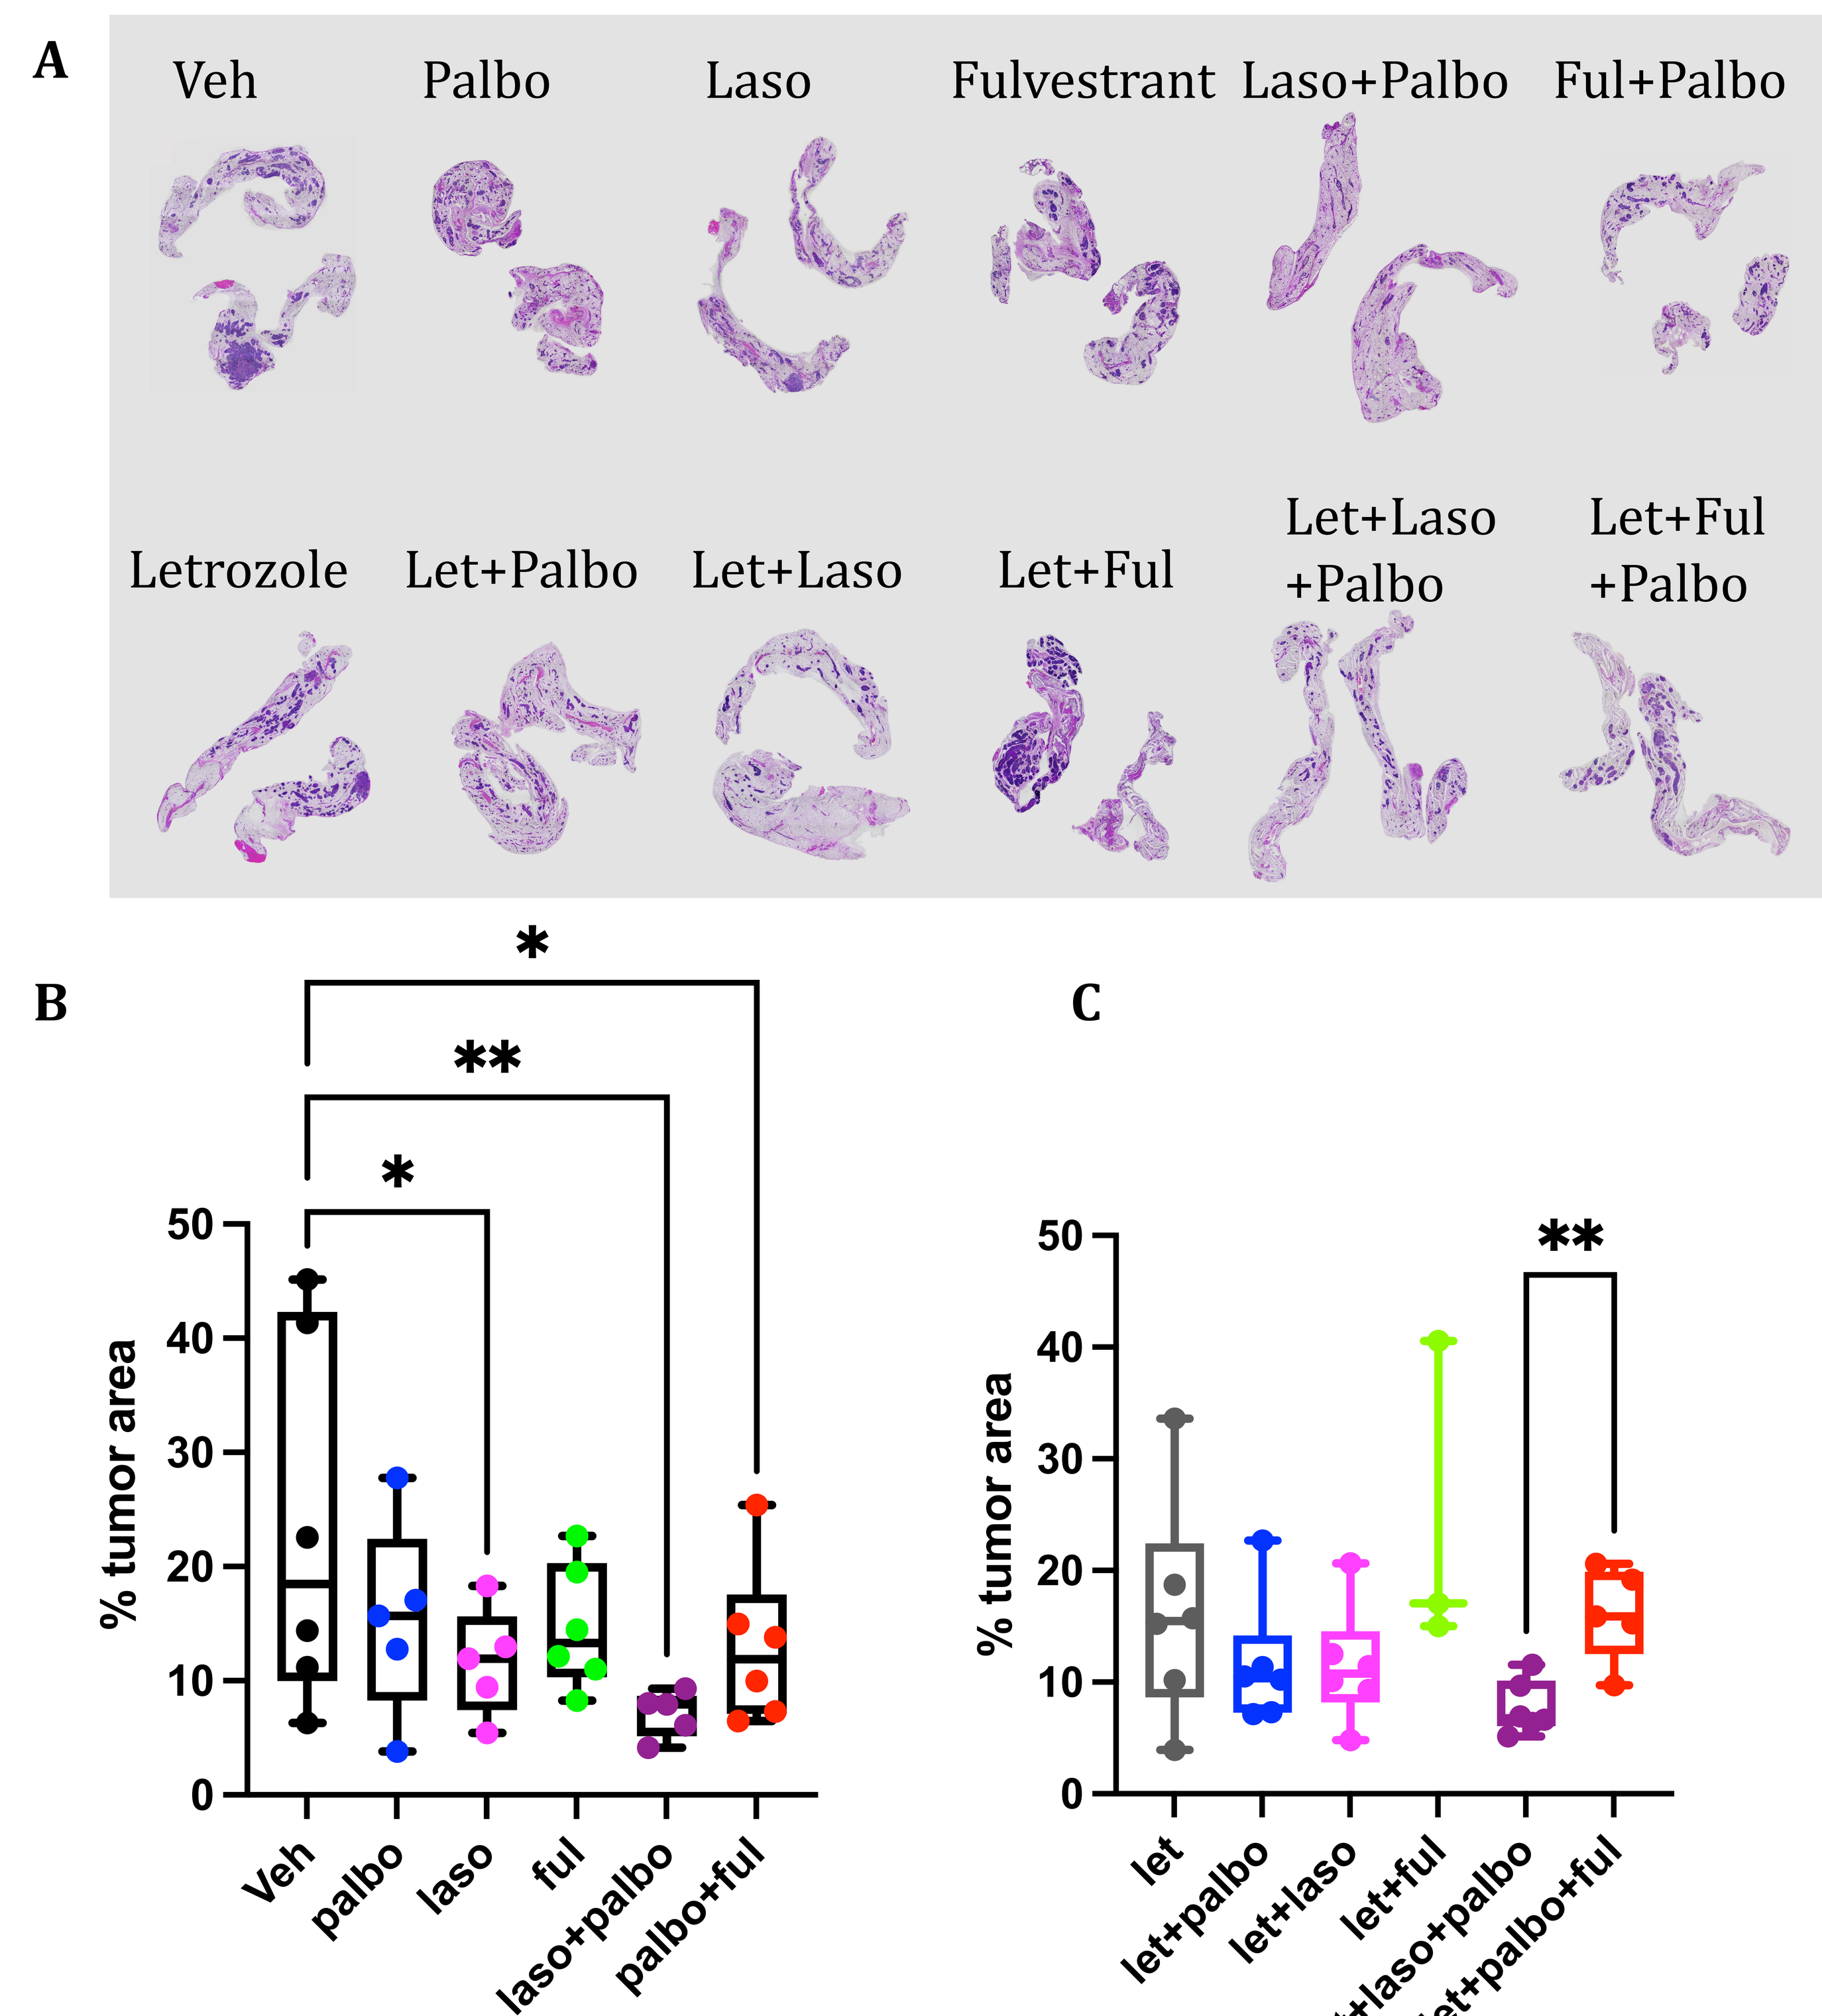
A
Veh
Palbo
Laso
Fulvestrant
Laso+Palbo
Ful+Palbo
Letrozole
Let+Palbo
Let+Laso
Let+Ful
Let+Laso +Palbo
Let+Ful +Palbo
B
C
% tumor area

Fig 2: A, H&E staining of representative mammary glands for each treatment group. **B & C**, Box plot quantification as percent tumor area based on H&E staining analyzed with NSI element software. P values are calculated by one way Anova *p<0.05, **p<0.001. N=3-6 mice

Lasofoxifene + palbociclib combination reduces tumor cell proliferation

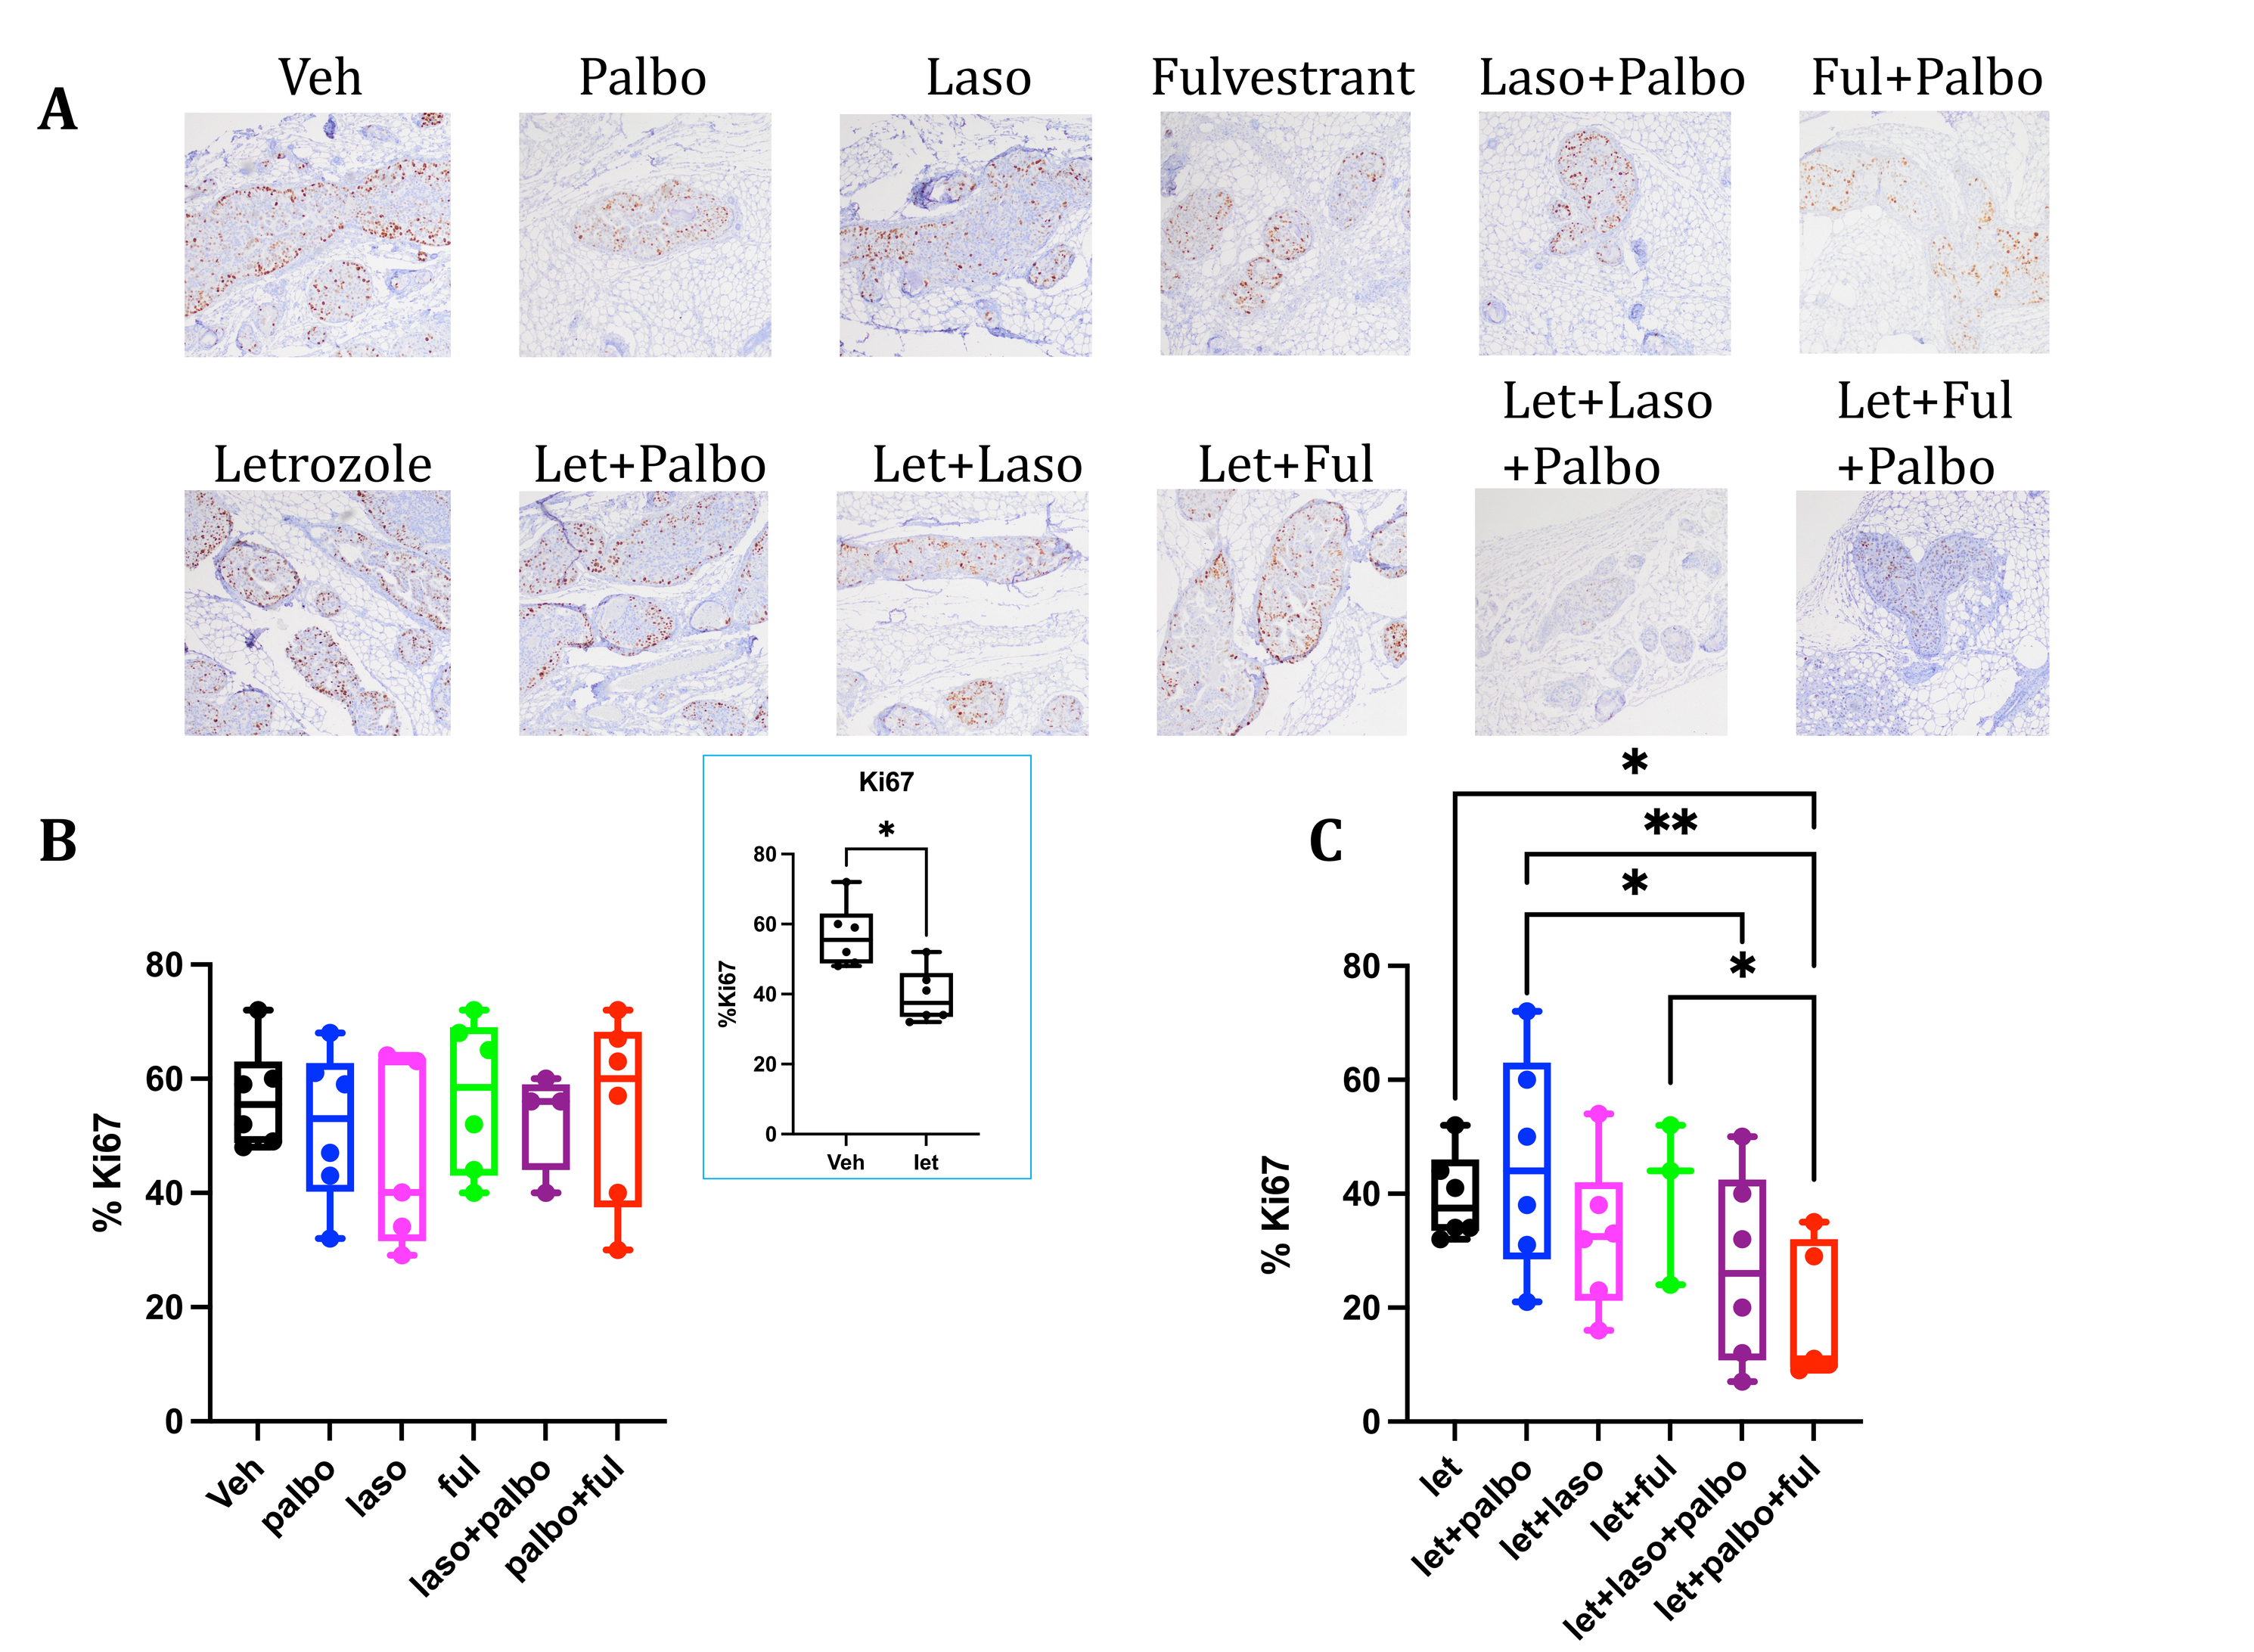
A
Veh
Palbo
Laso
Fulvestrant
Laso+Palbo
Ful+Palbo
Letrozole
Let+Palbo
Let+Laso
Let+Ful
Let+Laso +Palbo
Let+Ful +Palbo
B
C
Ki67
% Ki67

Fig 3: A, IHC staining of mammary glands with Ki67 antibody showing overall lower percentage of proliferation in the letrozole+laso+palbo group. **B & C**, Box plot representing the quantification of the Ki67 percent. Insert box plot comparing veh to letrozole treatment group. P values are calculated by one way Anova *p<0.05, **p<0.001, N=3 to 6 mice

Lasofoxifene + palbociclib combination reduces bone metastasis

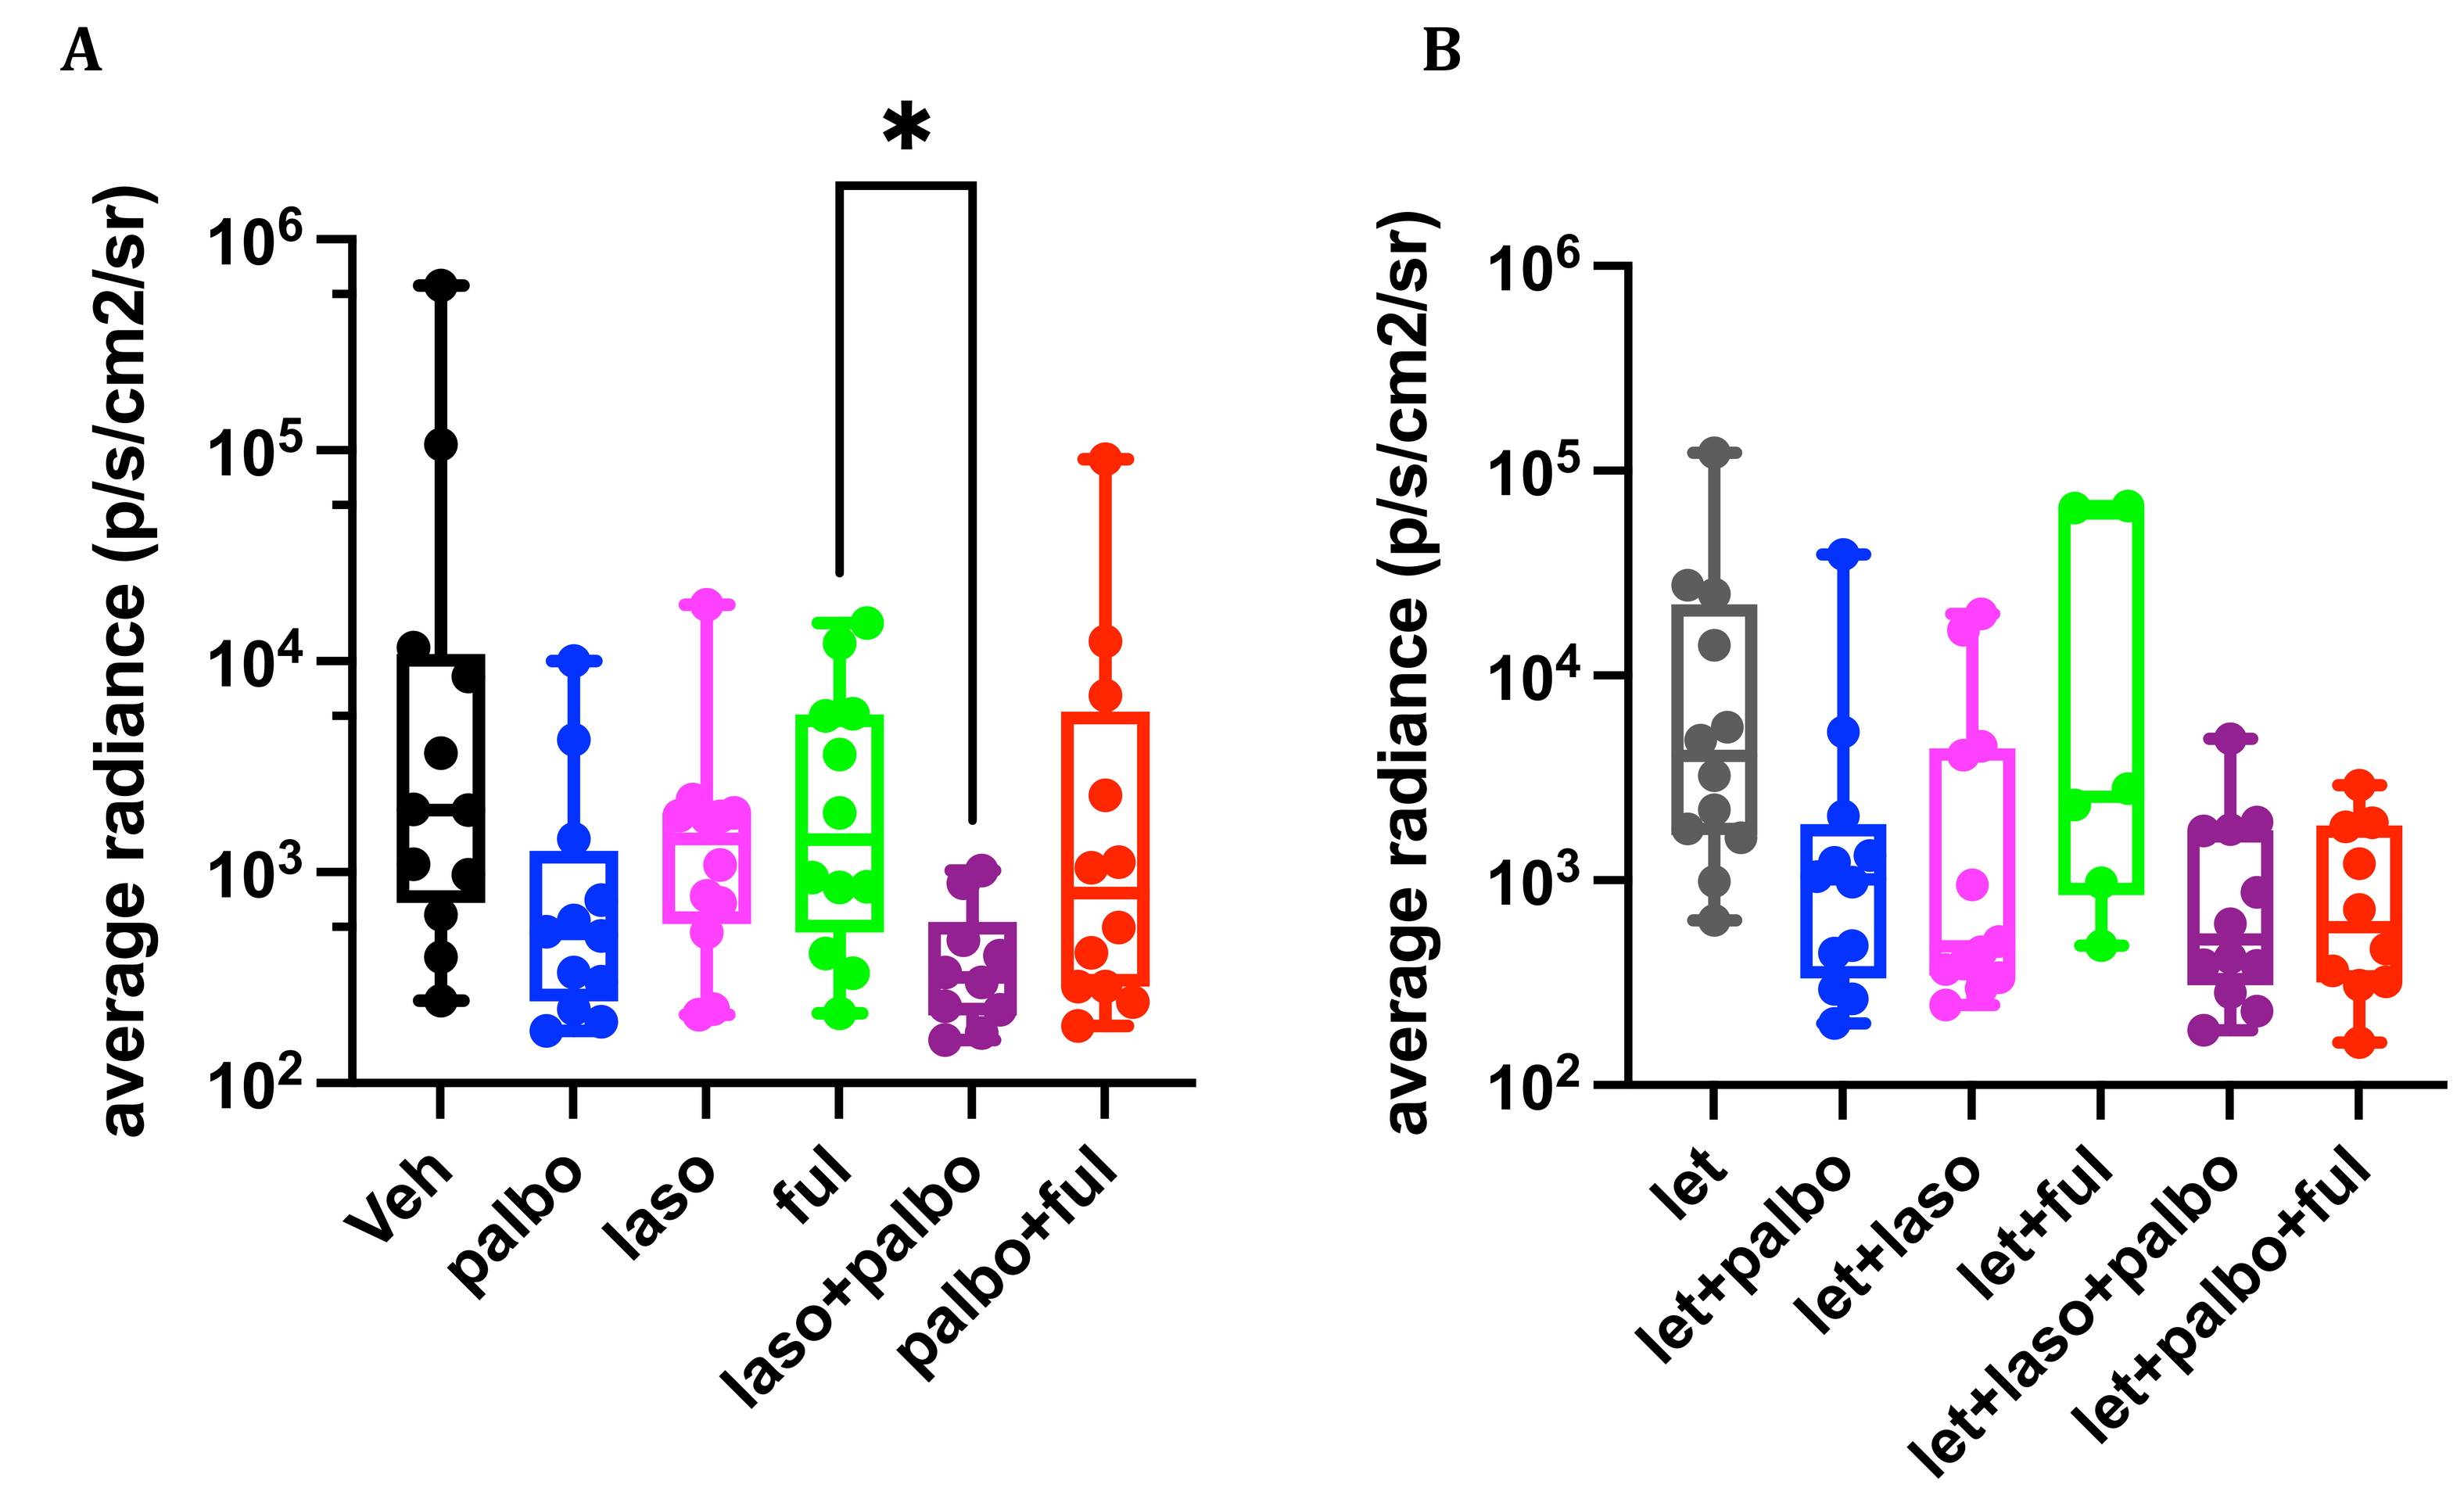
A
B
average radiance (p/s/cm2/sr)
Veh
palbo
laso
ful
laso+palbo
palbo+ful
let
let+palbo
let+laso
let+ful
let+laso+palbo
let+palbo+ful

Fig 4: A&B, Box plot representing average radiance of bones measured ex-vivo in a Xenogen IVIS imager. P values are calculated by one way Anova *p<0.05, N=6-12 bones

Experimental design

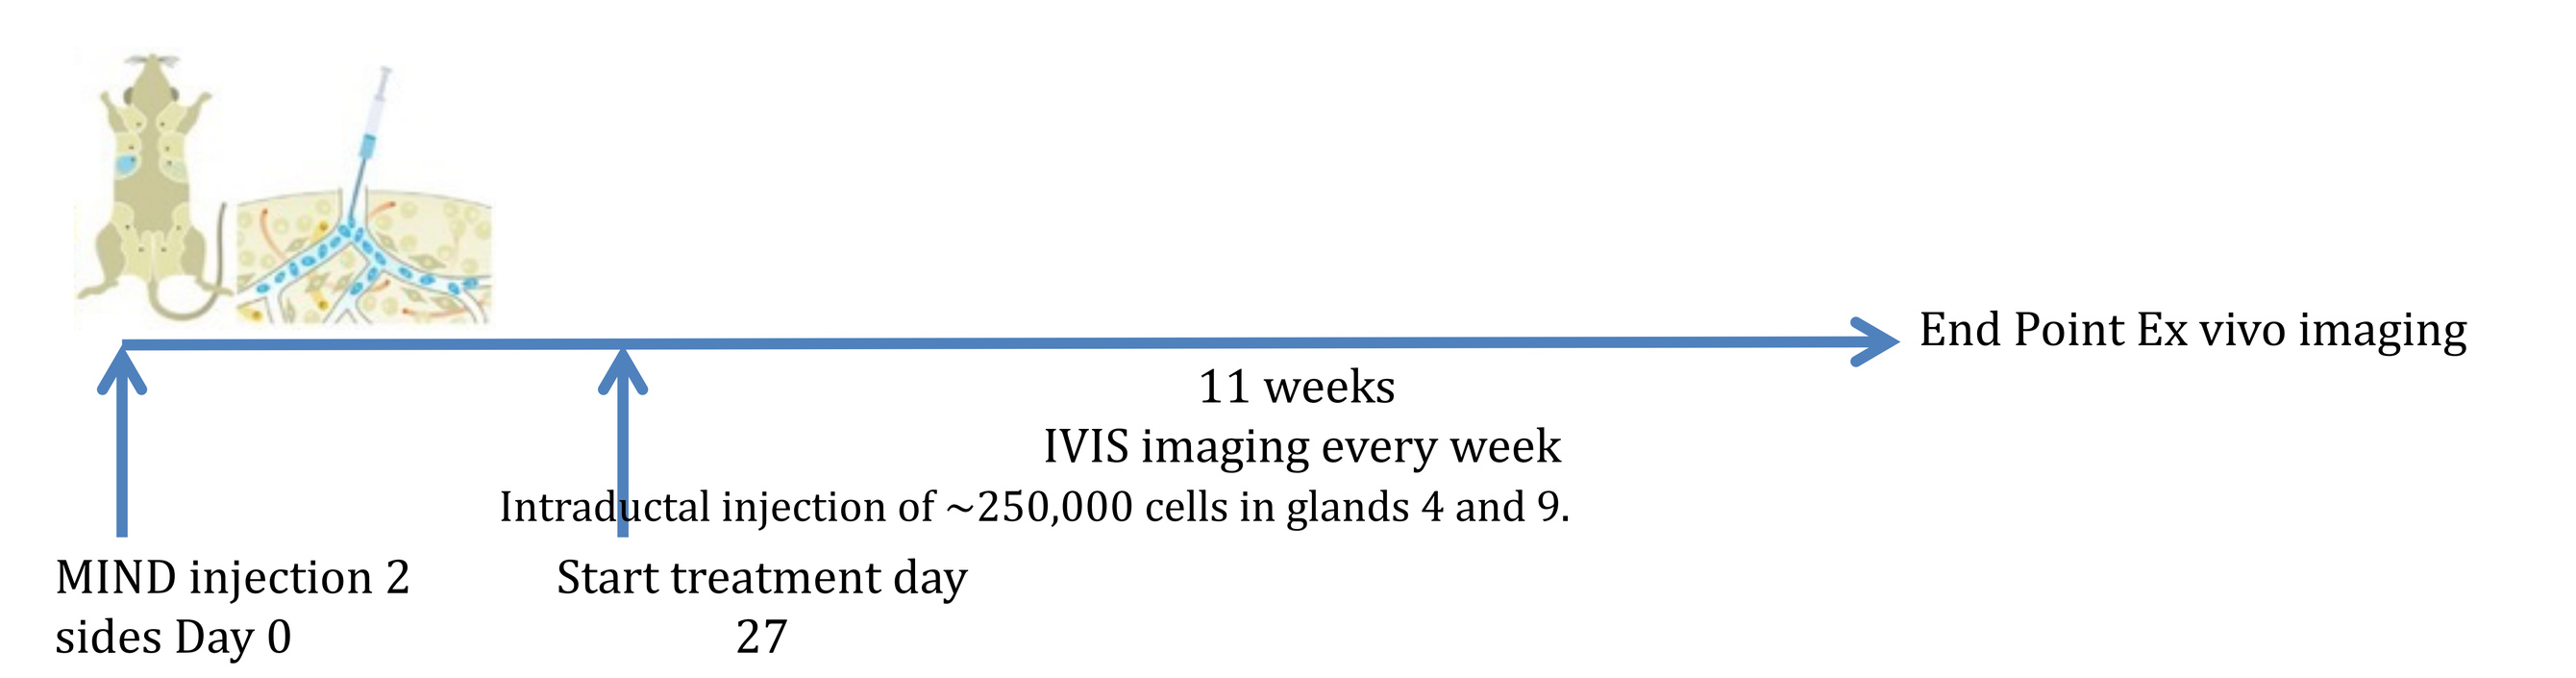
End Point Ex vivo imaging
11 weeks
IVIS imaging every week
Intraductal injection of ~250,000 cells in glands 4 and 9.
MIND injection 2 sides Day 0
Start treatment day 27

Treatments:
- Veh 5 days/week sc
- Laso 10 mg/kg, 5 times/week sc
- Palbo 70 mg/kg
- Fulvestrant 5mg/mouse (~250 mg/kg) 1/week sc
- Laso + Palbo
- Palbo + fulvestrant

Treatments:
- Letrozole 1 μg 5 days/week sc
- Let + Laso 10 mg/kg, 5 times/week sc
- Let + Palbo 70 mg/kg
- Let + Fulvestrant 5mg/mouse (~250 mg/kg) 1/week sc
- Let + Laso + Palbo
- Let + Palbo + fulvestrant

Conclusions

- Lasofoxifene alone or in combination with palbociclib inhibits primary tumor growth in a model of AI resistant breast cancer.
- The combination of lasofoxifene with palbociclib reduces tumor cell proliferation.
- Lasofoxifene + palbociclib decreases metastasis to the bone.
- Lasofoxifene +/- Palbociclib is a promising therapy for AI resistant breast cancer.

Acknowledgments

MCF7 LTLT were kindly provided by Dr Tekmal San Antonio TX.
Histology was performed by the Human Tissue Resource Center at university of Chicago
Imaging was performed at the small imaging facility at the University of Chicago
Mouse housing and treatment was performed at the Animal Resource Center at the University of Chicago