Patient Knowledge, Attitudes and Perceptions Regarding Breast Cancer Biomarkers, Testing, and Quality of Life

Hope S. Rugo[1], Kelly Shanahan[2], Elizabeth Attias[3], Miriam Portman[3], Jenna Gencarelli[3]

[1]University of California San Francisco Comprehensive Cancer Center, San Francisco, CA; [2]METAvivor; [3]Sermonix Pharmaceuticals, Columbus, OH

Presented at the 2020
San Antonio Breast
Cancer Symposium®
December 8–11, 2020

BACKGROUND

Precision medicine and molecular testing for specific biomarkers to inform treatment selection has evolved to become the standard of care for advanced breast cancer. Although there is widespread education for health care practitioners, there is less education available for patients. Two surveys from the oncology literature (Pinheiro 2017 , Ciardiello 2016) reported that 90% of physicians worldwide utilize biomarkers and patients would like to understand how testing informs treatment.

The objective of our online survey was to gain a greater understanding of patient familiarity with biomarker testing, specifically ESR1 mutation testing, and patient willingness to request information and testing. Patients were also surveyed about other areas of education of greatest interest, including quality of life (QOL) issues.

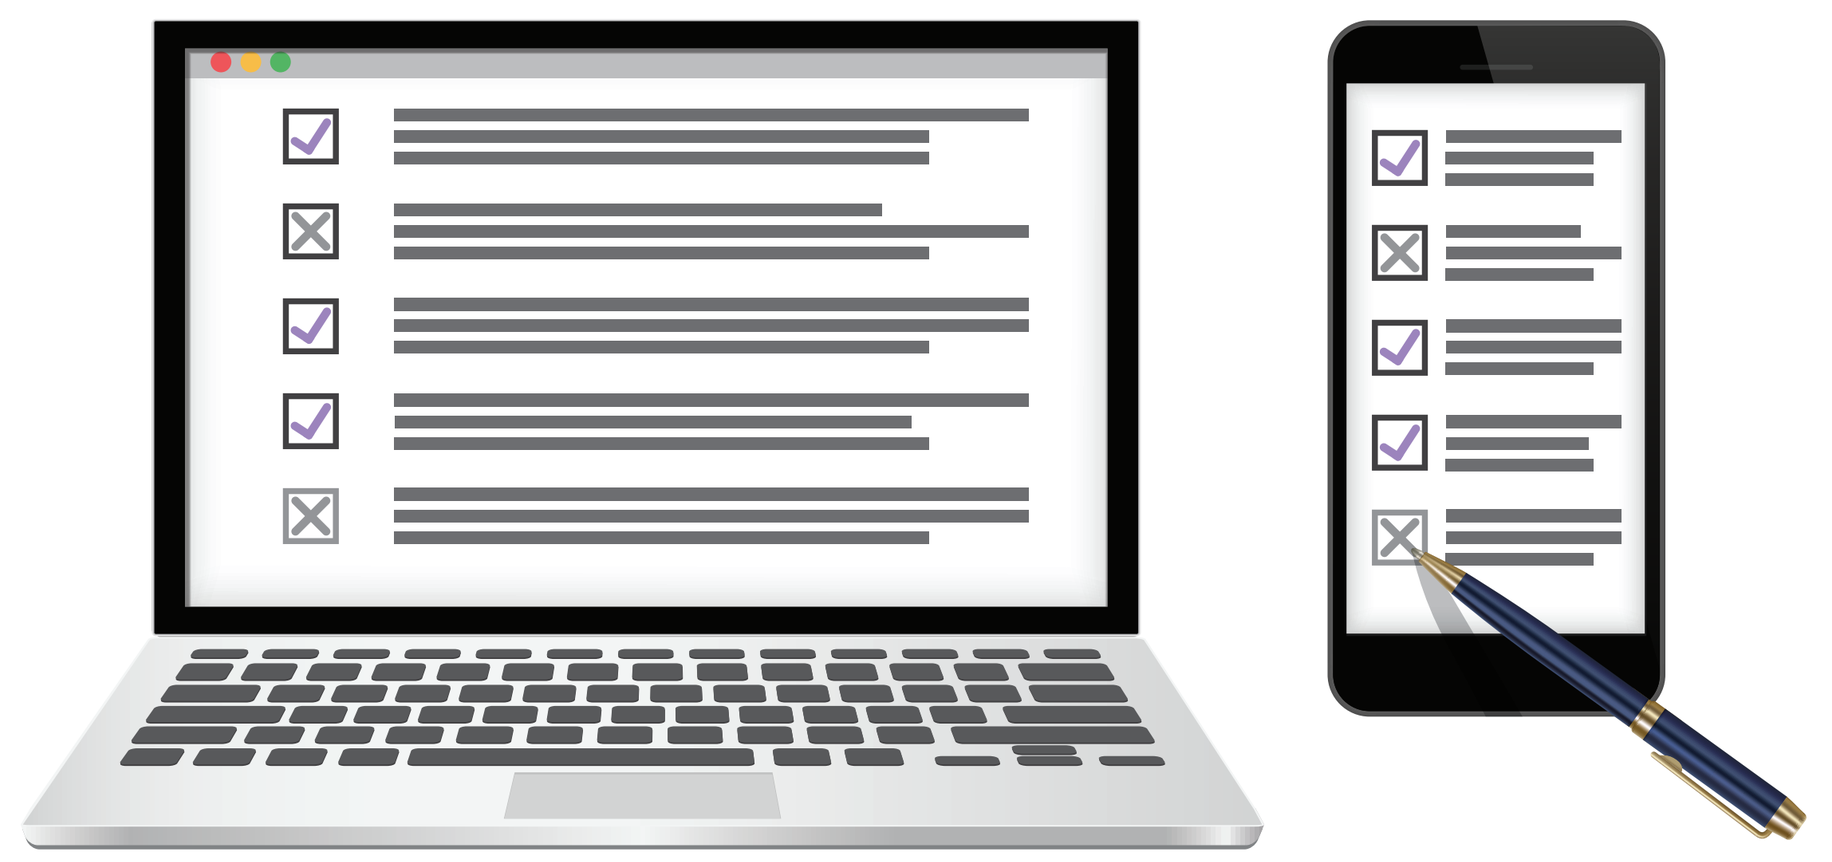

PATIENT ONLINE SURVEY

METHODS

A 10 question on-line survey was sent to patients through 2 patient advocacy organizations: CURE's CURExtra e-newsletter, and METAvivor's social media channels; METAvivor's survey included 2 additional questions to determine if respondents had metastatic disease and were currently on treatment. The survey asked about side effects impacting quality of life, satisfaction with information received about QOL issues, and knowledge about biomarkers (specifically ESR1), as well as willingness to request ESR1 testing.

RESULTS

A total of 343 completed responses were received: 177 from CURE and 166 from METAvivor, 82.5% from patients, 11% from patients who were patients and advocates, 2.3% from advocates, and 4.3% from 'other'.

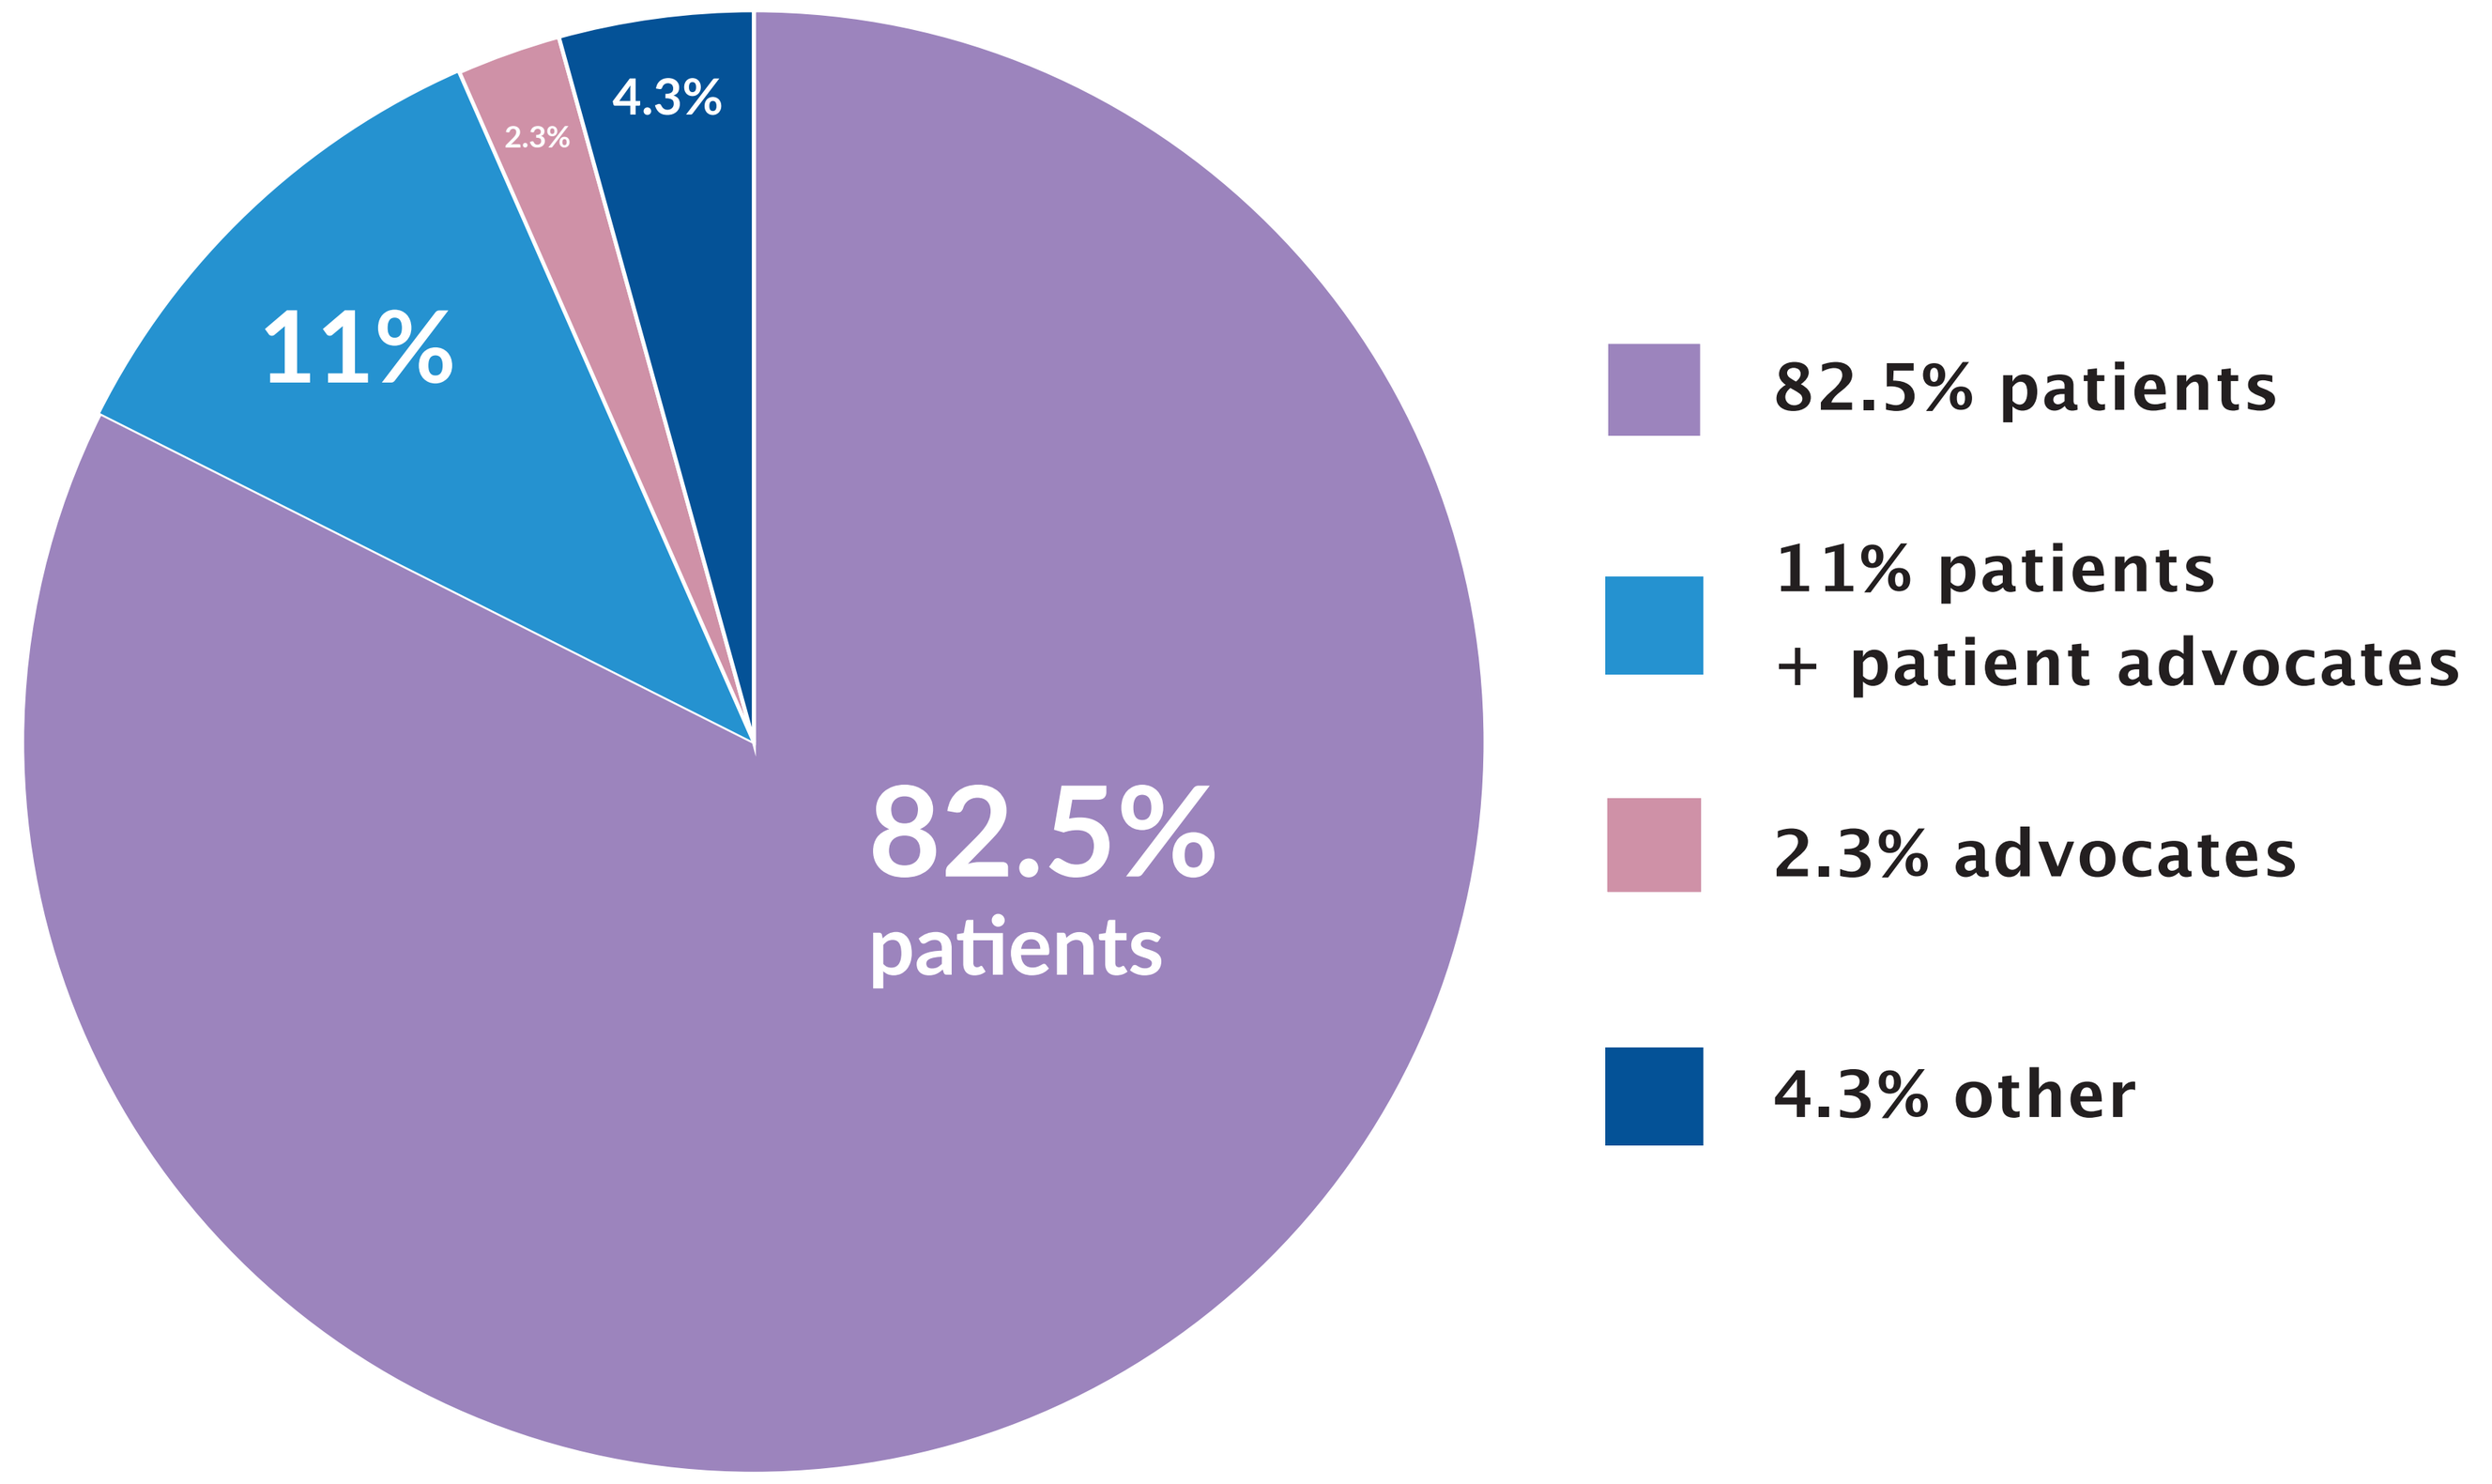
4.3%
2.3%
11%
82.5%
patients
82.5% patients
11% patients
+ patient advocates
2.3% advocates
4.3% other

22% of patients surveyed were familiar with or aware of biomarker testing; only 12% were familiar with ESR1 mutations and biomarker testing.

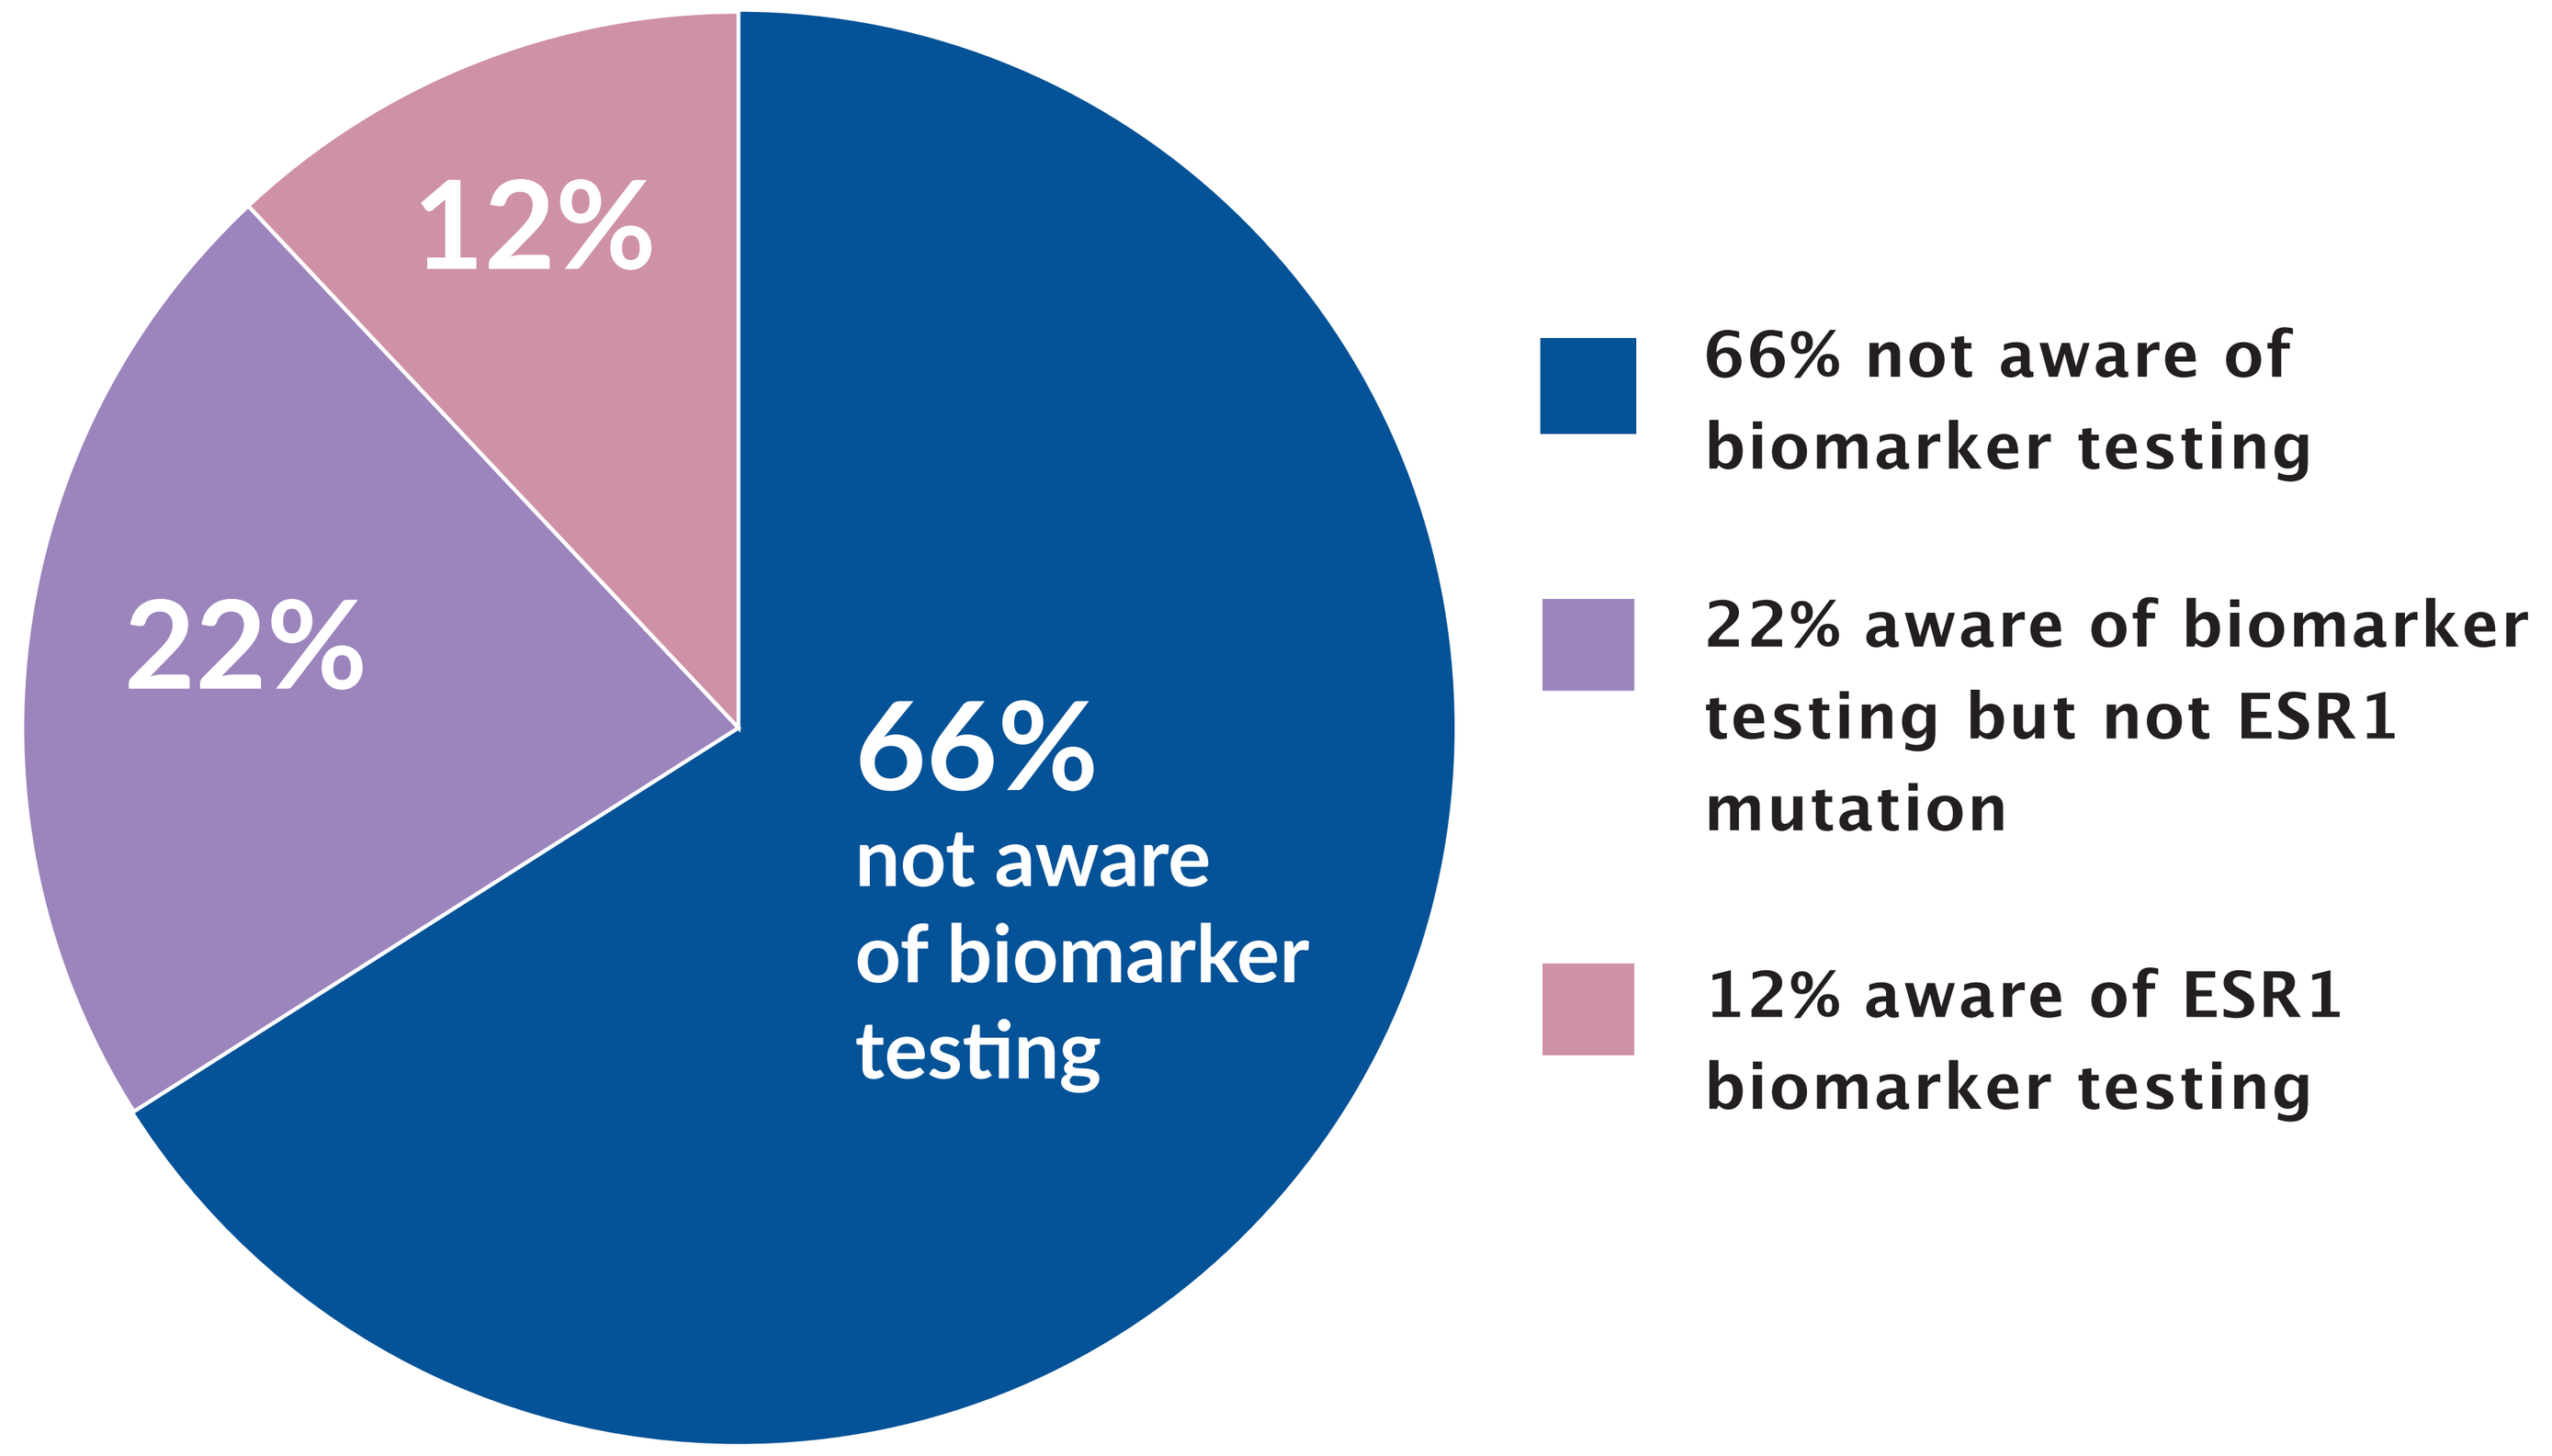
12%
22%
66%
not aware
of biomarker
testing
66% not aware of biomarker testing
22% aware of biomarker testing but not ESR1 mutation
12% aware of ESR1 biomarker testing

SURVEY RESPONDENTS IDENTIFIED 3 AREAS OF GREATEST INTEREST:

- **78%** New treatments in development
- **63%** Quality of life discussions
- **60%** Management of side effects

SURVEY RESPONDENTS IDENTIFIED KEY ISSUES THEY HAVE BEEN STRUGGLING WITH THAT NEGATIVELY IMPACT THEIR QOL:

- **62%** Joint pain
- **52%** Sexual intimacy & recurrent UTIs
- **51%** GI disturbances
- **48%** Bone pain

Disease subgroups included ER+/HER2– (53%), ER+/HER2+ (16%), TNBC (12%) and ER–/HER2+ (11%).

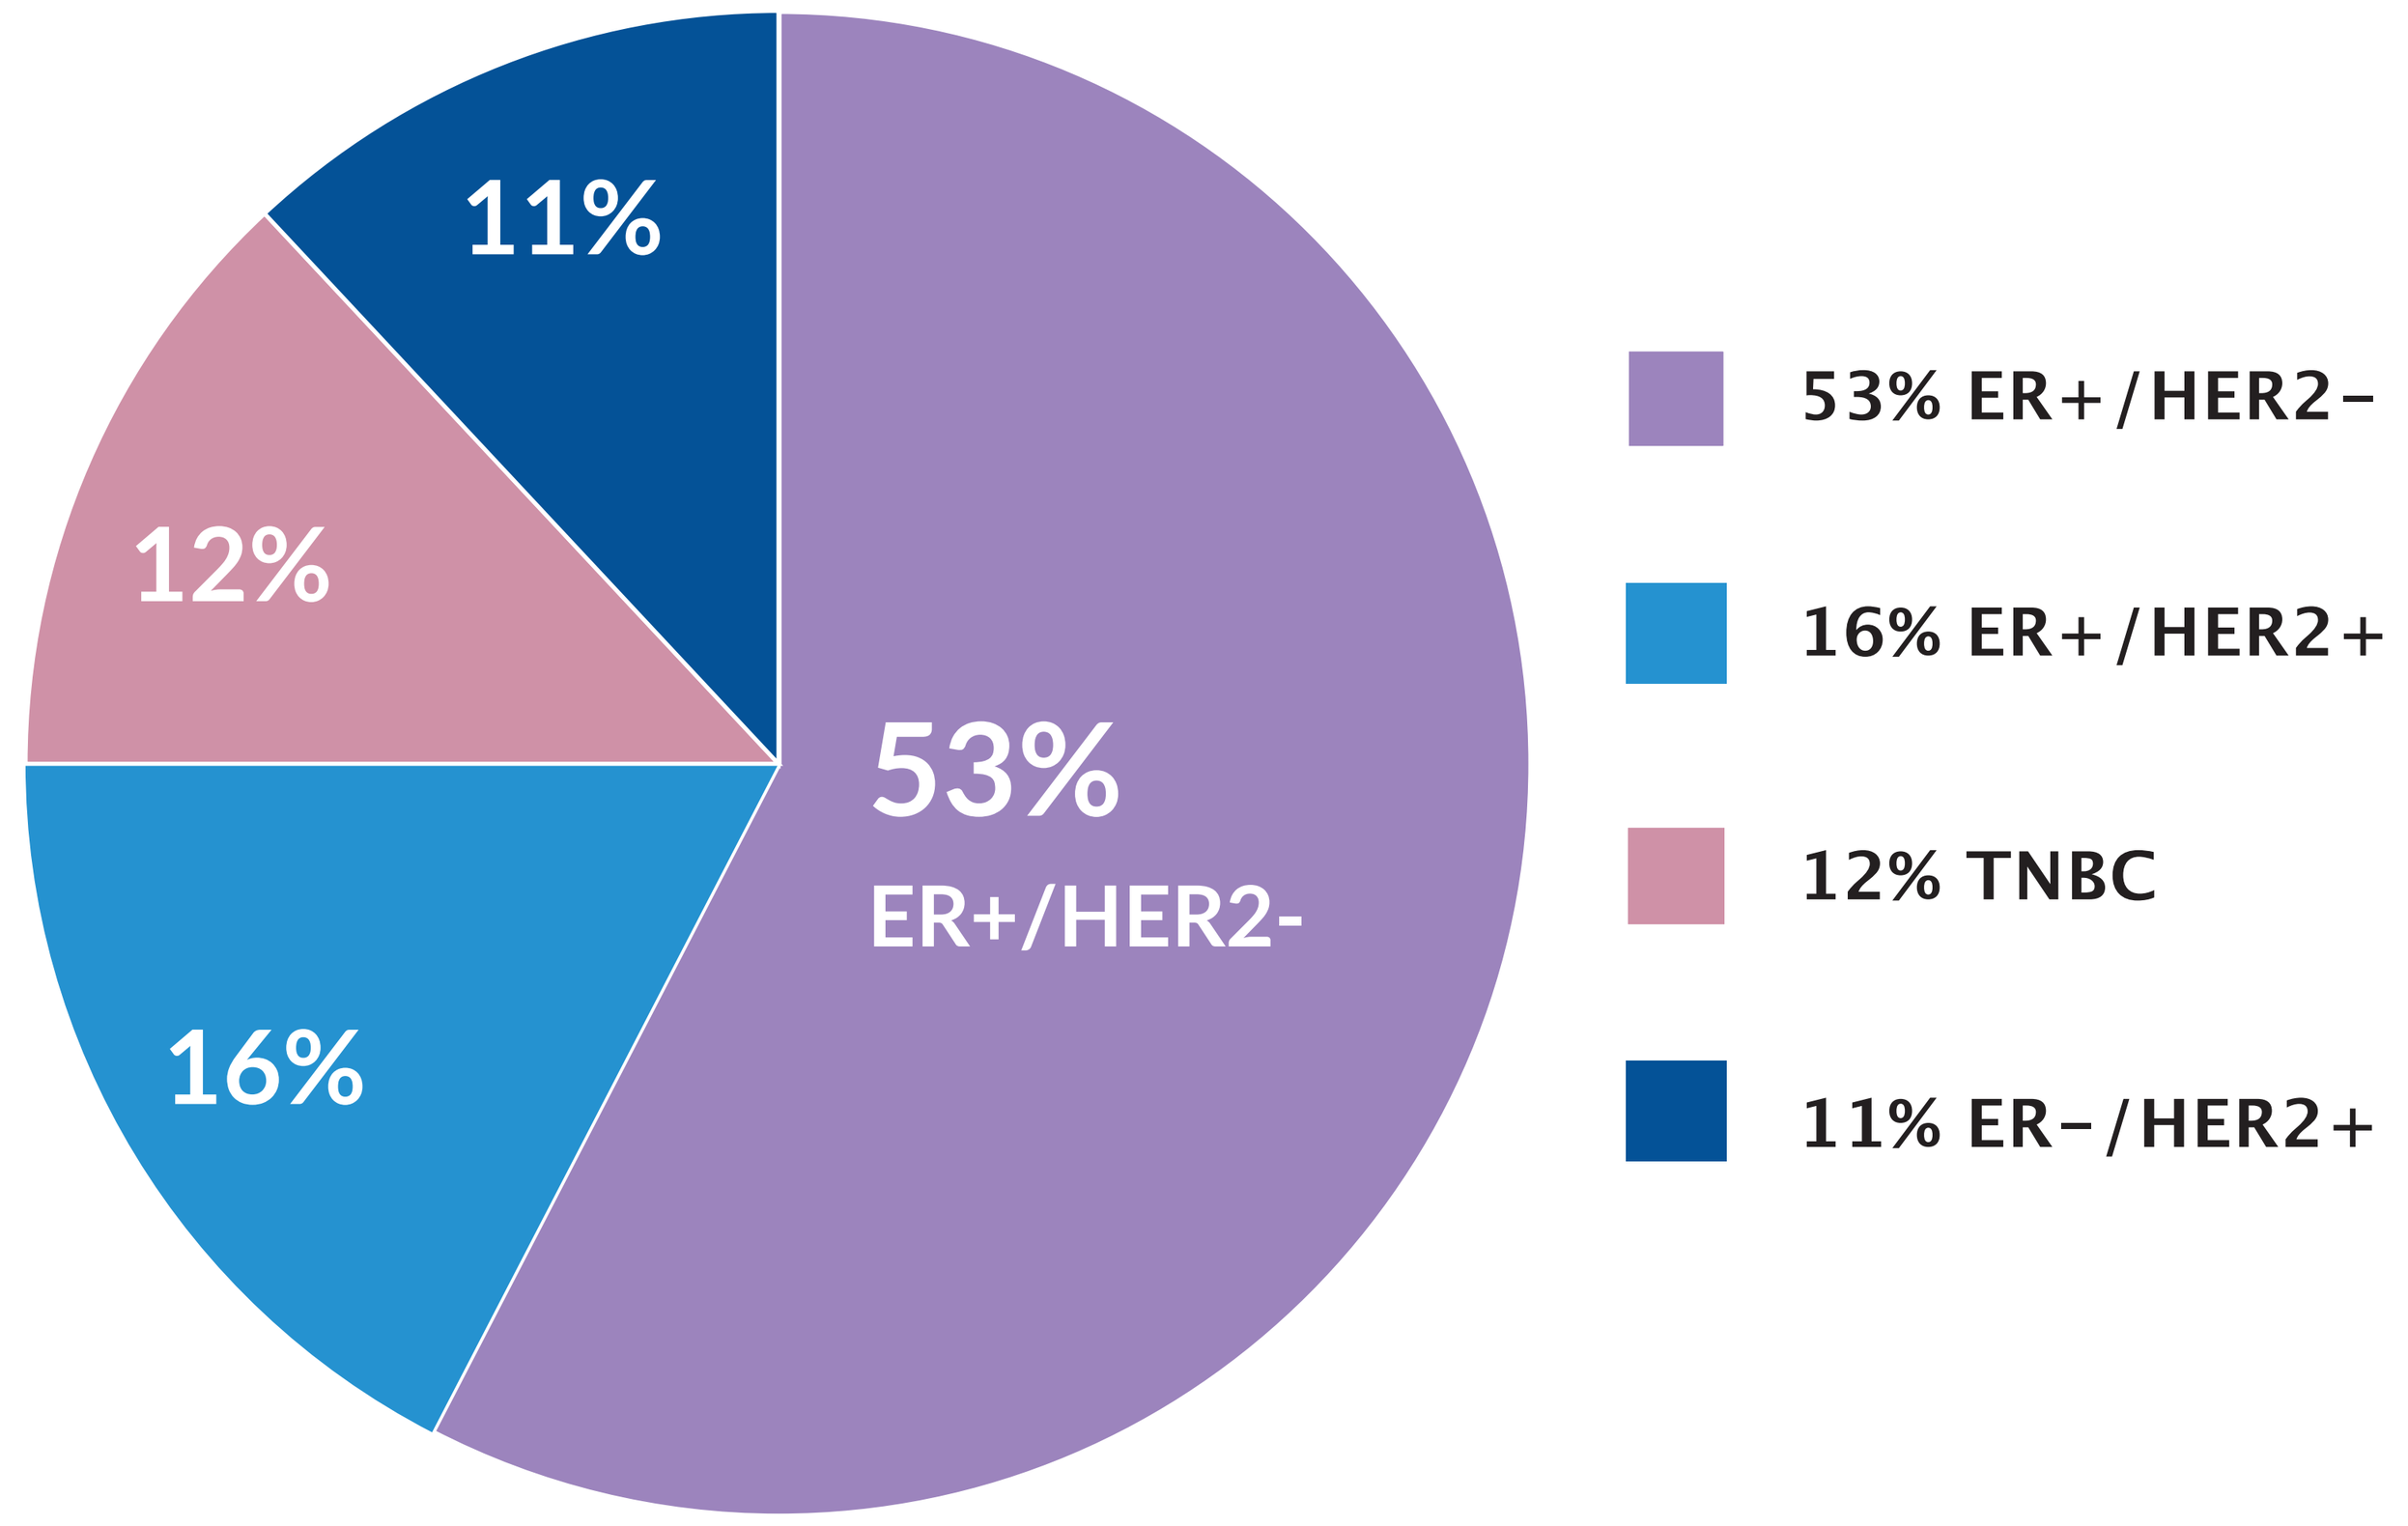
11%
12%
16%
53%
ER+/HER2-
53% ER+/HER2–
16% ER+/HER2+
12% TNBC
11% ER–/HER2+

The mean and median times from diagnosis were 8.25 and 6 years.

75% of respondents were very likely (52%) or likely (23%) to ask their provider about ESR1 mutations if this would aid in tailoring treatment*.

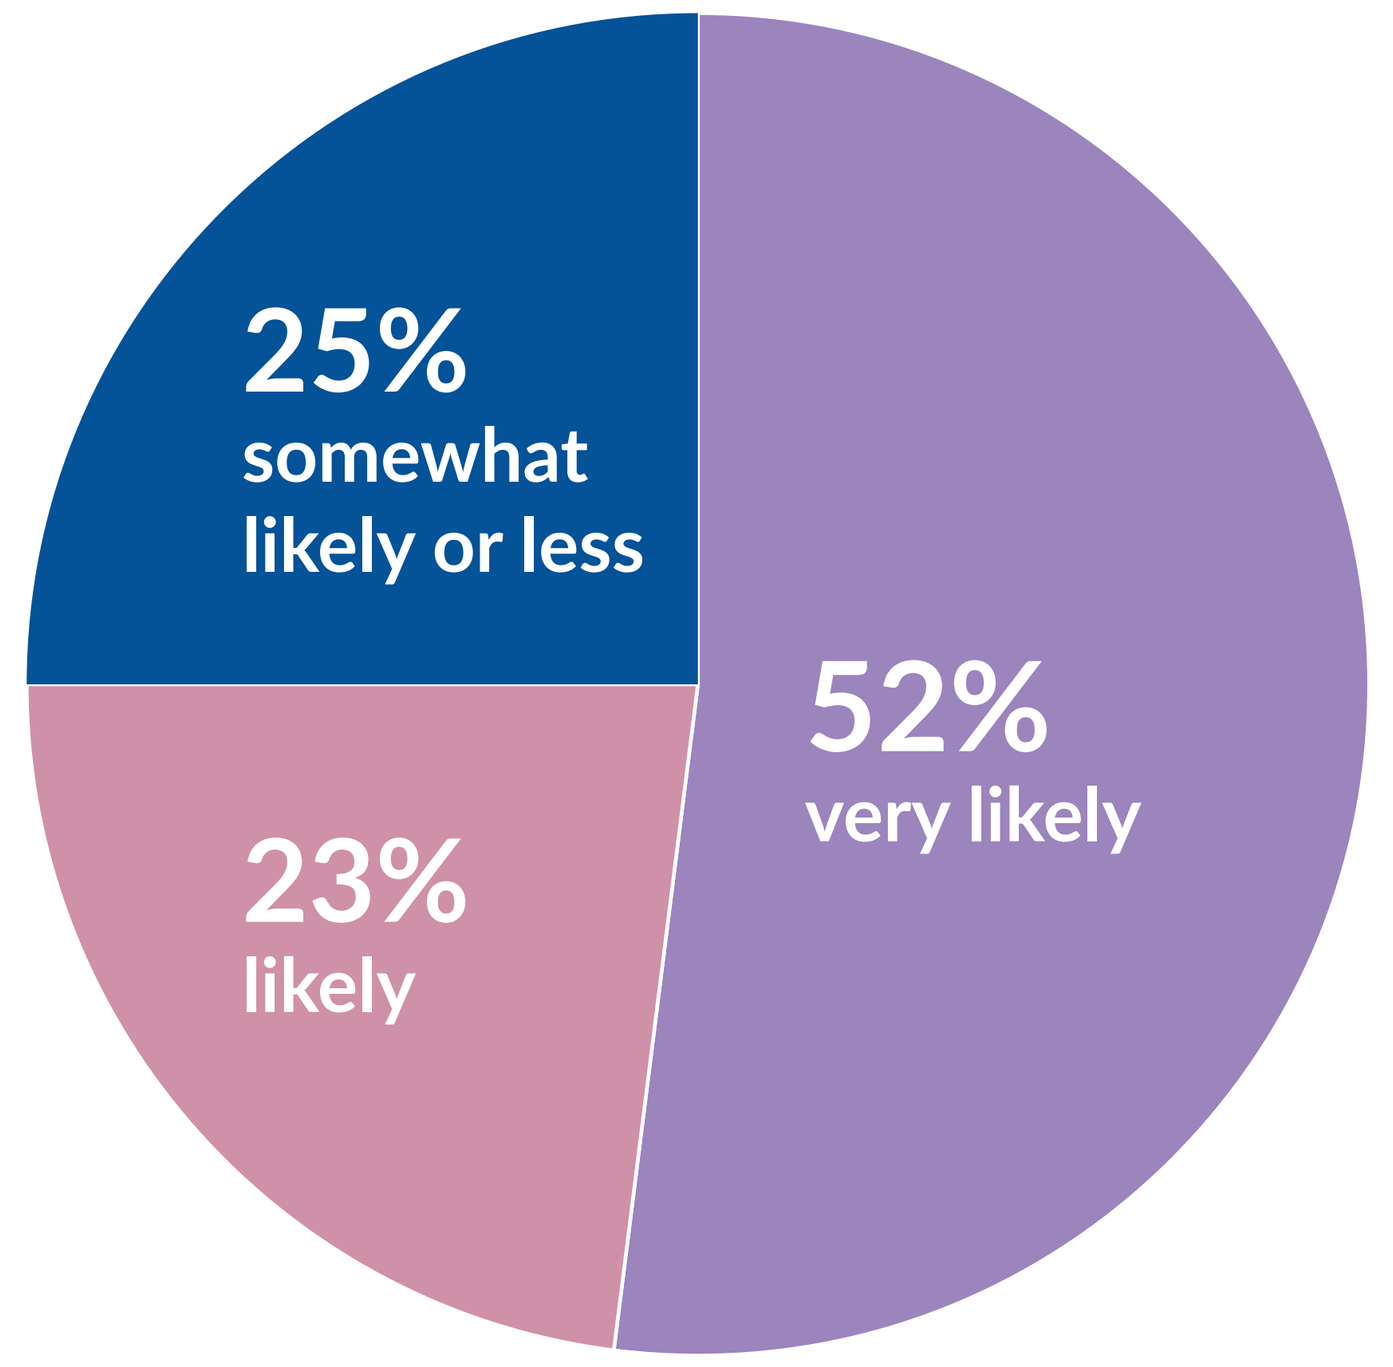
25%
somewhat
likely or less
52%
very likely
23%
likely

*METAvivor respondents more likely to ask provider than CURE respondents (78 vs 70%).

71% were very likely (49%) or likely (22%) to request ESR1 mutation testing.

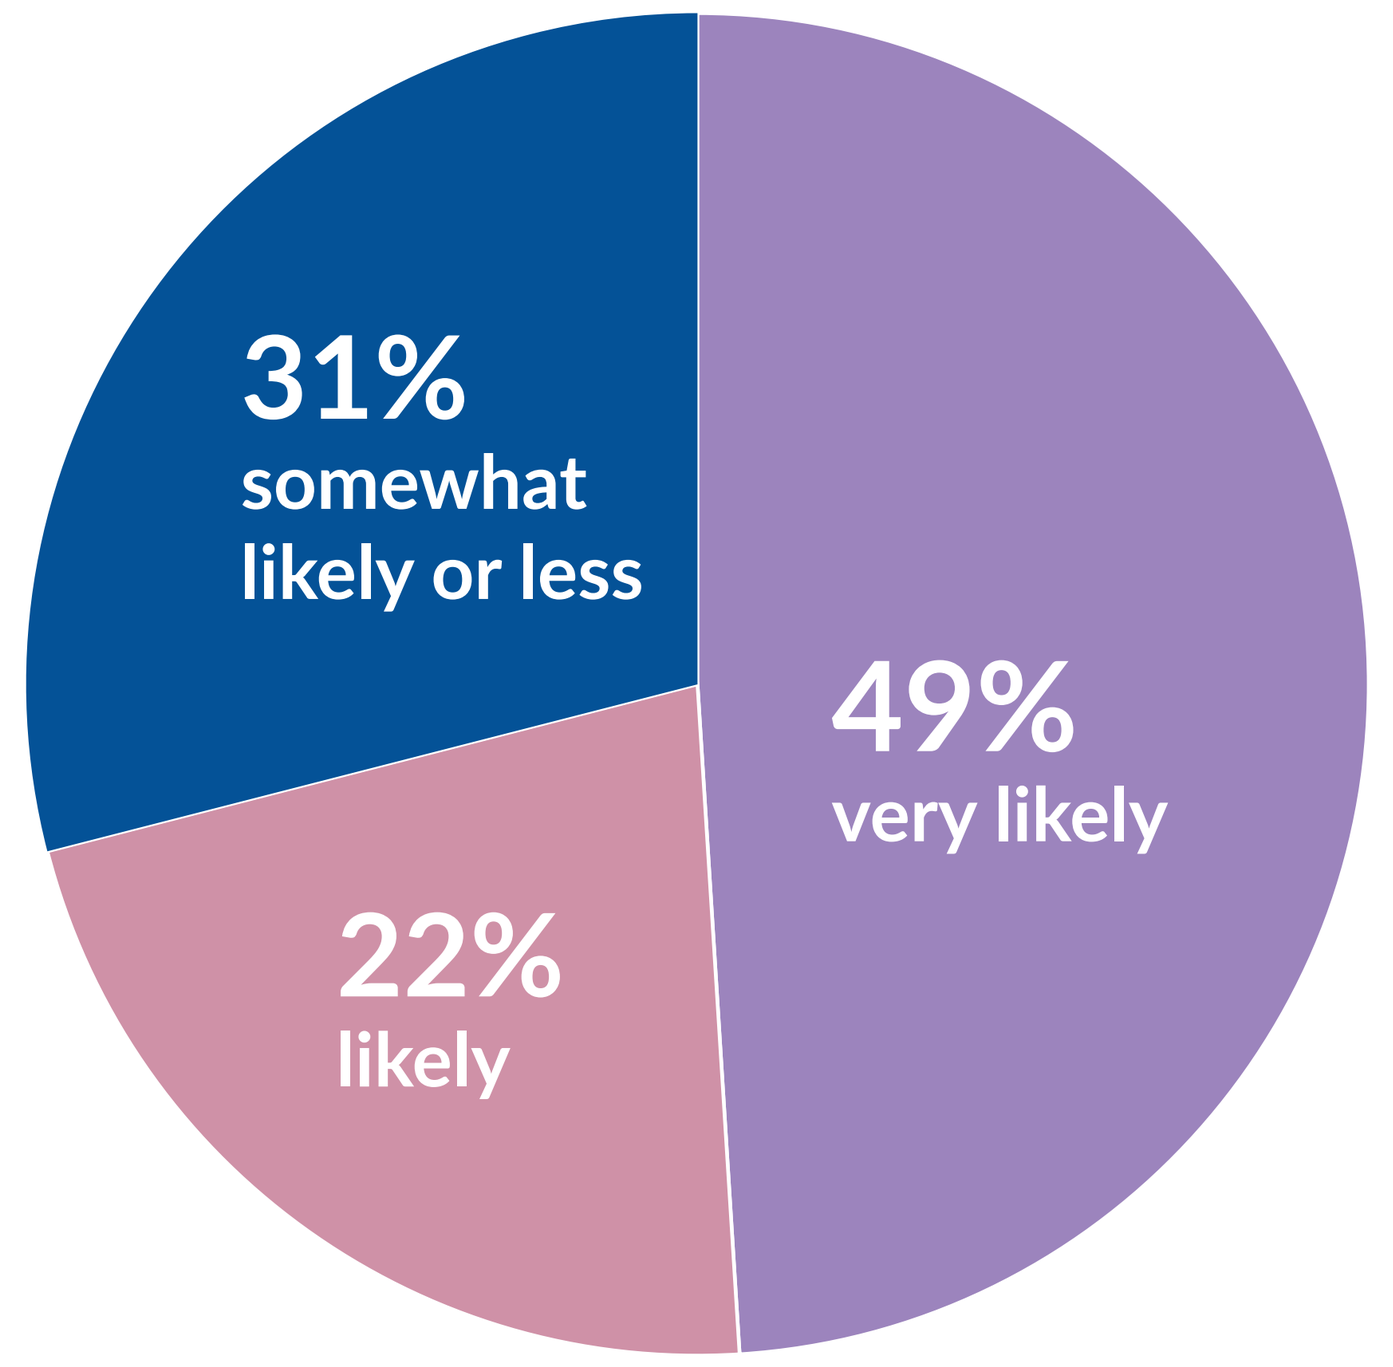
31%
somewhat
likely or less
49%
very likely
22%
likely

55% were very satisfied or satisfied with the QOL information that they were receiving from their provider.

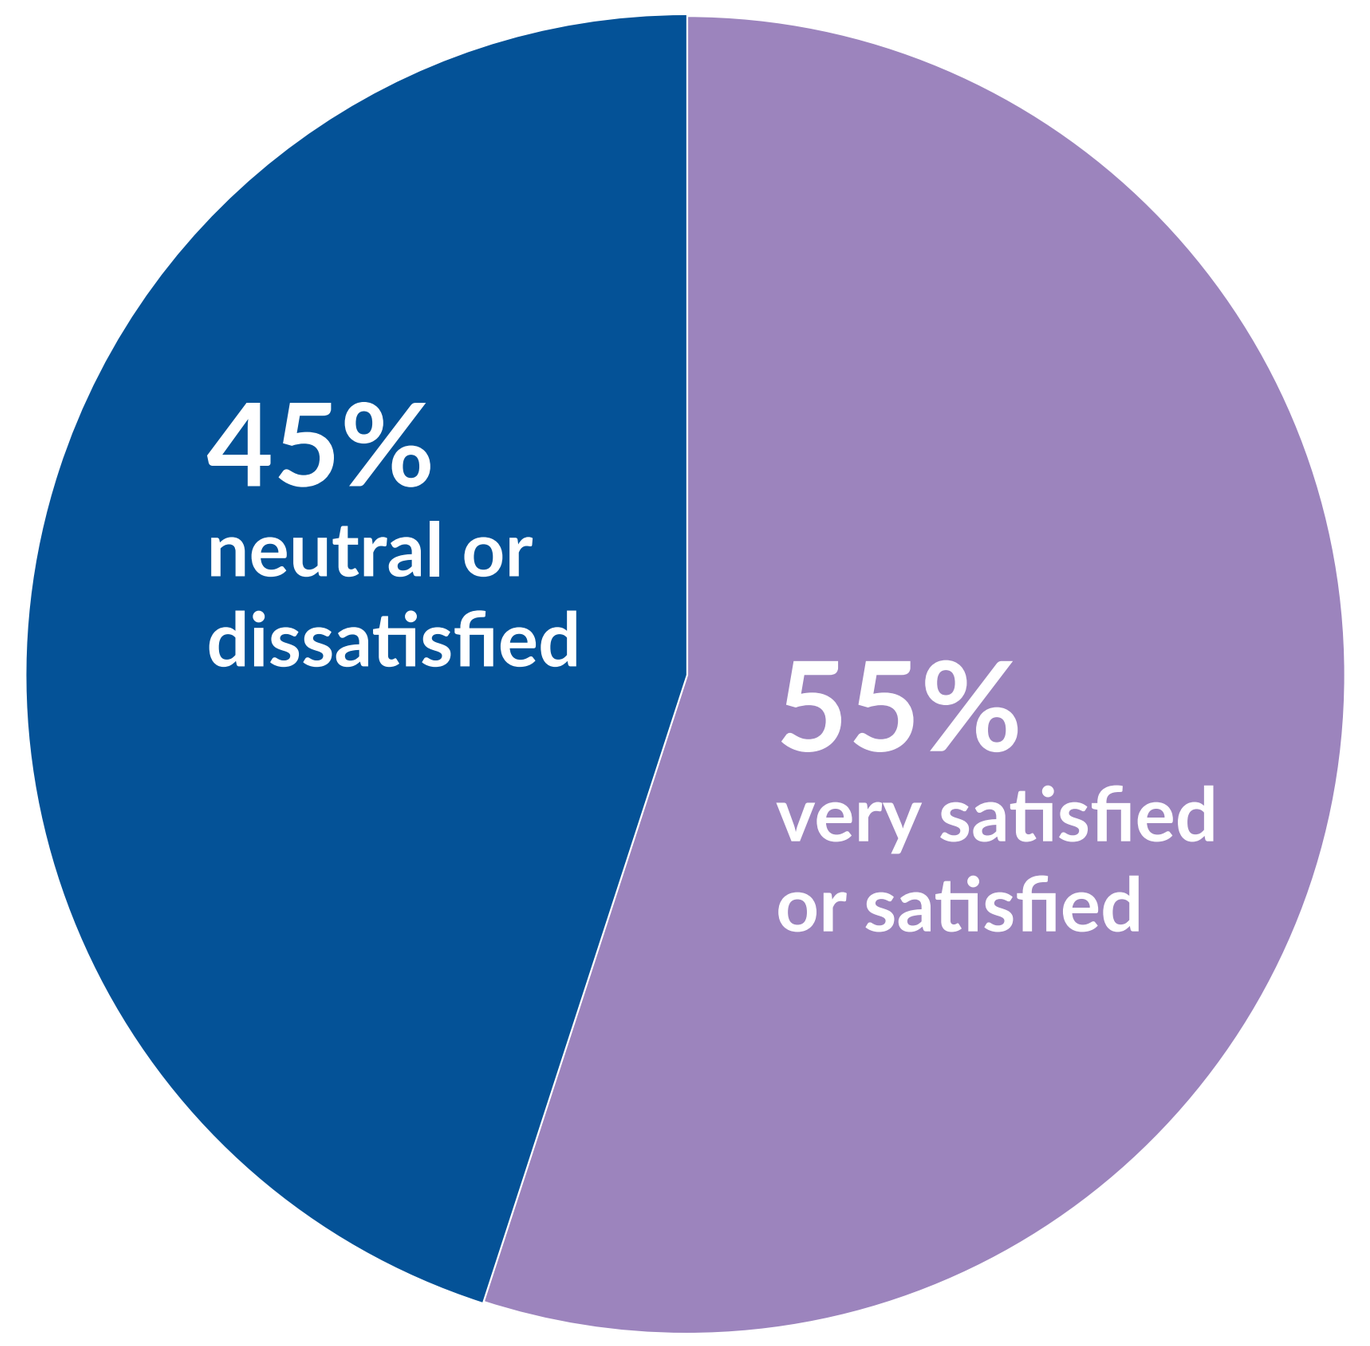
45%
neutral or
dissatisfied
55%
very satisfied
or satisfied

54% looked to their providers as their primary source of information, with 27% citing the internet and medical journals as additional sources of information.

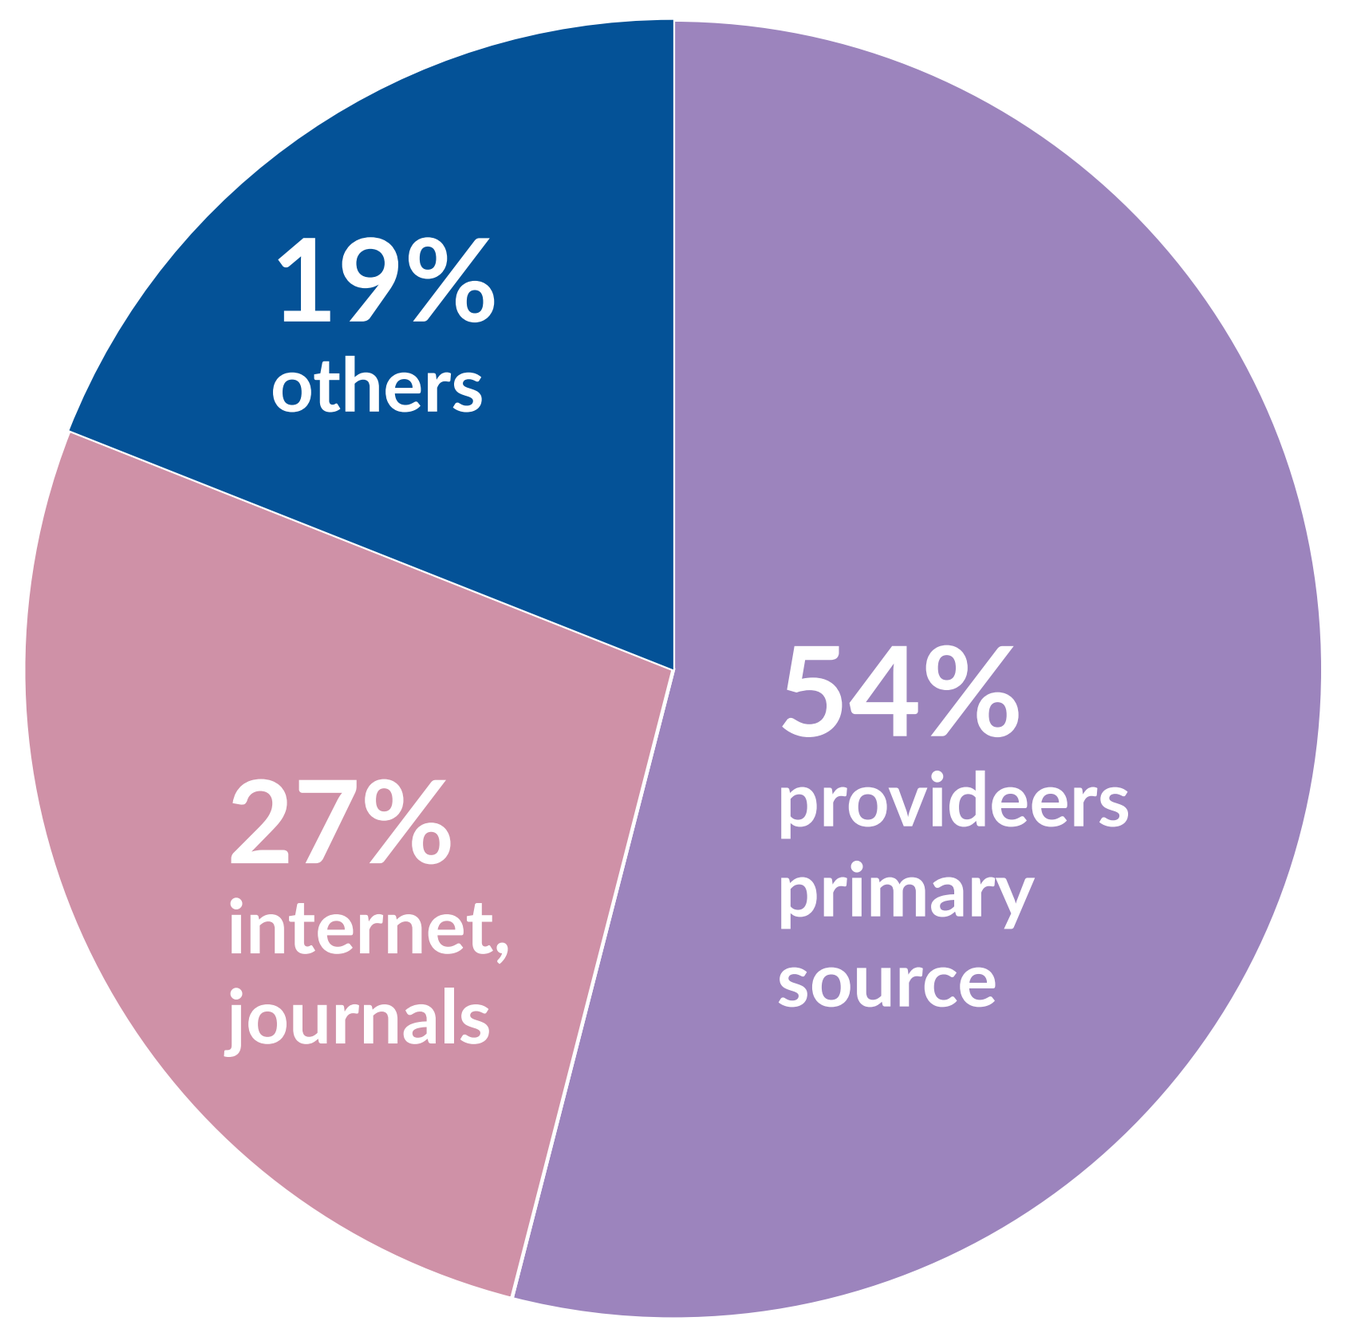
19%
others
27%
internet,
journals
54%
provideers
primary
source

CONCLUSIONS

- The results of this survey indicate knowledge gaps in patients with breast cancer
- Specific areas with knowledge gaps include genomic testing to tailor therapy and the role of ESR1
- QOL issues remain an area of unmet need
- These data underscore the desire of breast cancer patients for information related to new treatments in development, discussions with their provider specific to QOL, and how to manage therapy side effects
- Providing patient education tools for providers and patient advocacy organizations including information about tumor genomics, treatment options, and side effect management is of utmost importance

This presentation is the intellectual property of the author/presenter. Contact Hope S. Rugo, MD, at hope.rugo@ucsf.edu for permission to reprint and/or distribute.

REFERENCES

1. Pinheiro APM, Pocock RH, Switchenko JM, et al. Discussing molecular testing in oncology care: Comparing patient and physician information preferences. Cancer. 2017;123(9):1610–1616. doi:10.1002/cncr.30494
2. Ciardiello F, Adams R, Tabernero J, et al. Awareness, Understanding, and Adoption of Precision Medicine to Deliver Personalized Treatment for Patients With Cancer: A Multinational Survey Comparison of Physicians and Patients. Oncologist. 2016;21(3):292–300. doi:10.1634/theoncologist.2015-0279

SUPPORT: Sermonix Pharmaceuticals