Lasofoxifene alone or in combination with palbociclib is an effective treatment for therapy-resistant ER-positive metastatic breast cancer

Lainé M[1], Kurleto J[1], Greene M[1], Chang YF[1], Phung L[1], Tan T[1], Hiipakka R[1], Komm B[2] and Greene GL[1]
[1] Ben May department for cancer research, University of Chicago, Chicago Il
[2] CSO, Sermonix Pharmaceuticals

Introduction

Patients with estrogen receptor positive (ERα+) Metastatic Breast Cancer (MBC) are typically treated with fulvestrant, a selective estrogen receptor degrader, or a combination of fulvestrant and palbociclib a CDK4/6 inhibitor. Approximately 25-40% of patients with ERα+ MBC have mutations in the ligand-binding domain of ERα. One of the most common mutations is Y537S, which confers ERα constitutive activity and renders tumors resistant to endocrine therapies. Lasofoxifene, a selective estrogen receptor modulator (SERM), was developed for the treatment of vaginal atrophy and osteoporosis. A previous study, performed in a mutant ERα+ MCF-7 xenograph MBC mouse model, showed that lasofoxifene was more effective than fulvestrant at inhibiting tumor growth and metastasis to the liver and lung. In the current dose/response study, we compared the efficacy of lasofoxifene + palbociclib combinations to fulvestrant + palbociclib combinations. Engineered and luciferase-GFP tagged MCF-7 Y537S cells were injected into the mammary ducts of NSG mice (MIND model) and tumor progression was monitored by live luminescence imaging of primary tumors, as well as ex vivo imaging and histochemical analysis of metastatic sites at study endpoint. All dose combinations of lasofoxifene + palbociclib were more effective than fulvestrant + palbociclib at inhibiting primary tumor growth as well as bone, lung, liver and brain metastasis. These data show that lasofoxifene has potential as an effective therapy for endocrine resistant, mutant ERα+ metastatic breast cancer.

Experimental design

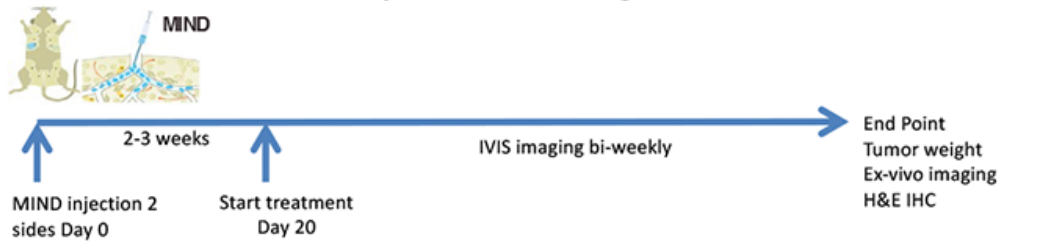
MIND
2-3 weeks
IVIS imaging bi-weekly
End Point
Tumor weight
Ex-vivo imaging
H&E IHC
MIND injection 2 sides Day 0
Start treatment Day 20

Treatments 12 groups:
- Veh 5 days/wk sc
- Laso 5 mg/kg and 10 mg/kg, 5 days/wk sc
- Fulvestrant 5mg/mouse, 1 day/wk sc
- Palbociclib 35 mg/kg and 70 mg/kg, gavage, 5 days/wk
- Fulvestrant + Palbo 35 mg/kg, 1 day/wk (sc), 5 days/wk gavage
- Fulvestrant + Palbo 70 mg/kg, 1 day/wk (sc), 5 days/wk gavage
- Laso 5mg/kg + Palbo 35 mg/kg, 5 days/wk, sc + gavage
- Laso 5mg/kg + Palbo 70 mg/kg, 5 days/wk, sc + gavage
- Laso 10mg/kg + Palbo 35 mg/kg, 5 days/wk, sc + gavage
- Laso 10mg/kg + Palbo 70 mg/kg, 5 days/wk, sc + gavage

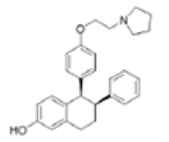

Lasofoxifene

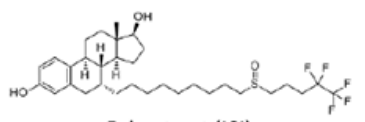

Fulvestrant (ICI)

Pathology, slide preparation and H&E staining was performed at the HTRC at University of Chicago. In vivo and ex-vivo imaging was performed at the OICF at University of Chicago.

Fig 1: CRISPR engineered MCF7 ERα-Y537S, cells were labeled with Luciferase-GFP plasmid via Lentiviral infection. Animal treatments were started 20 days after cell injection. Mice were imaged bi-weekly until sacrifice at day 96. Mice (NSG, 6 mice/group) were injected with 250,000 MCF7 ERα Y537S on both side gland 4 and 9. MCF7 CRISPR/Cas9 mutants were a gracious gift of Ben Park from the Sidney Kimmel Cancer Center at Johns Hopkins.

Results

Lasofoxifene + palbociclib is significantly more effective than palbociclib alone or a combination of fulvestrant and palbociclib at reducing primary tumors growth and final tumor weight

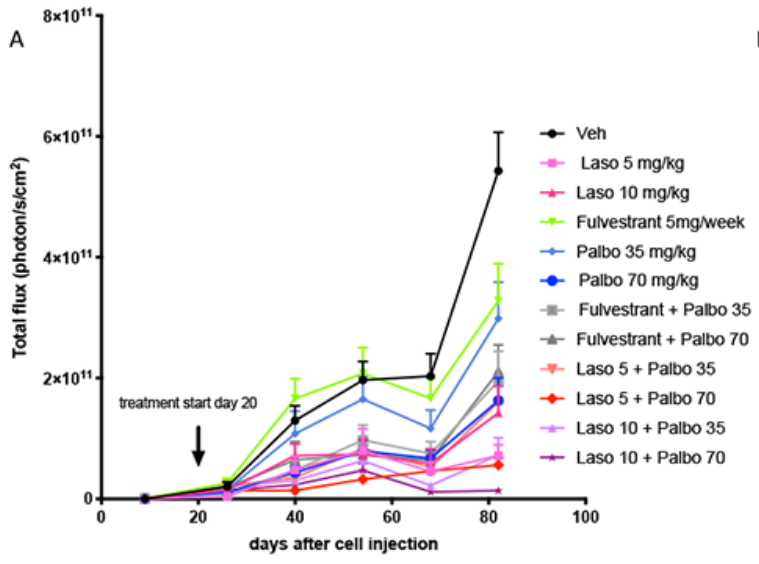
A
Total flux (photon/s/cm²)
treatment start day 20
days after cell injection
Veh
Laso 5 mg/kg
Laso 10 mg/kg
Fulvestrant 5mg/week
Palbo 35 mg/kg
Palbo 70 mg/kg
Fulvestrant + Palbo 35
Fulvestrant + Palbo 70
Laso 5 + Palbo 35
Laso 5 + Palbo 70
Laso 10 + Palbo 35
Laso 10 + Palbo 70

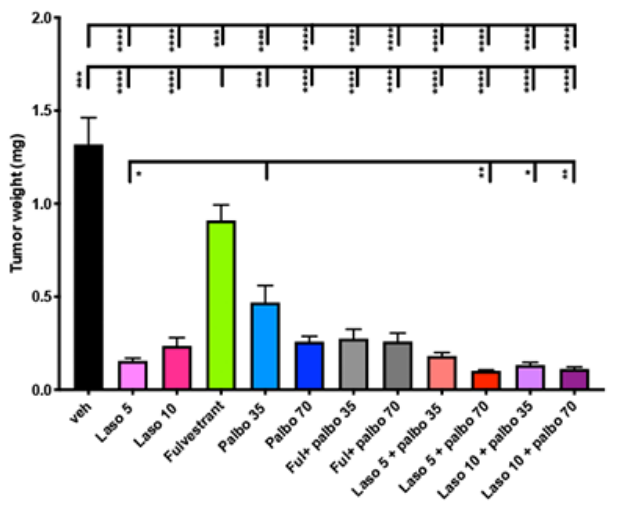
B
Tumor weight (mg)
veh
Laso 5
Laso 10
Fulvestrant
Palbo 35
Palbo 70
Ful+ palbo 35
Ful+ palbo 70
Laso 5 + palbo 35
Laso 5 + palbo 70
Laso 10 + palbo 35
Laso 10 + palbo 70

Fig 2: A) Tumor growth followed by quantitation of Bioluminscence signal measured by in vivo IVIS imaging for each treatment group. **B)** Tumor weight at sacrifice. Statistics: one-way anova *p<0.05, **p<0.01, ***P<0.001, ****P<0.00001. n=5-6

Lasofoxifene + palbociclib is more effective than fulvestrant + pabociclib and significantly more effective than fulvestrant alone at inhibiting metastasis to the lung

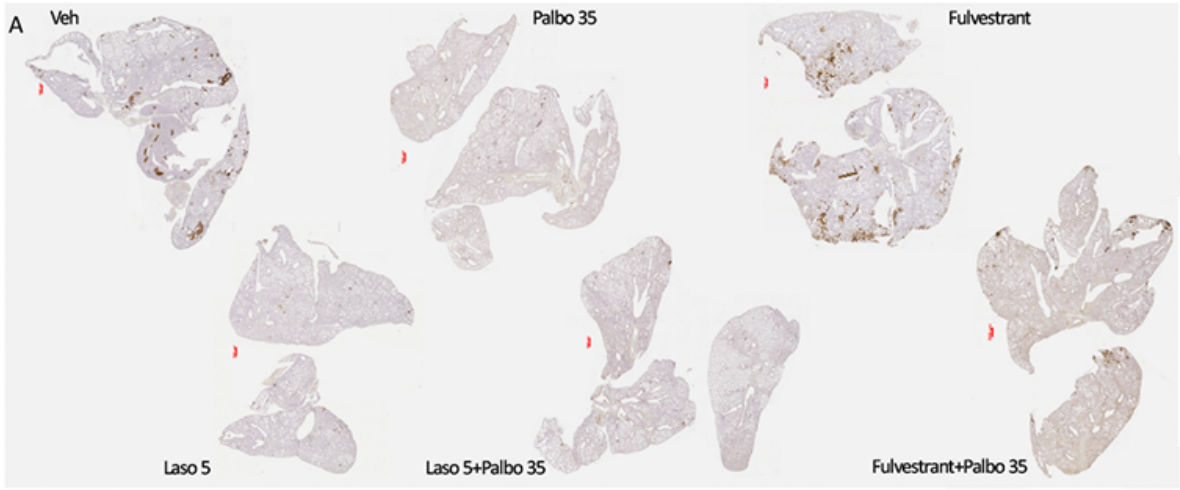
A
Veh
Palbo 35
Fulvestrant
Laso 5
Laso 5+Palbo 35
Fulvestrant+Palbo 35

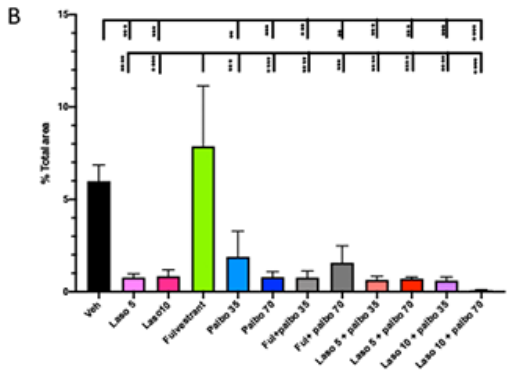
B
% Total area

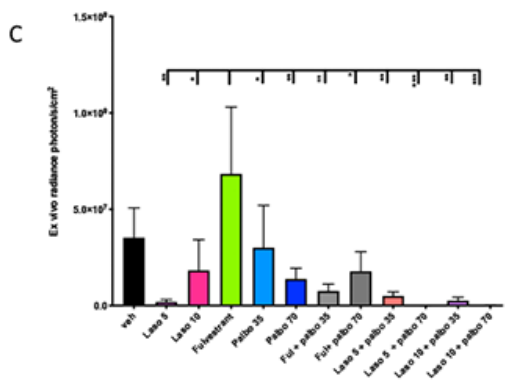
C
Ex vivo radiance photon/s/cm²

Fig 3: A) Representative IHC staining with anti-luciferase antibody showing lung metastases (brown color). **B)** Quantitation of IHC staining performed with Nikon NIS element analysis, showing metastasis as percent of total lung area. **C)** Quantitation of ex-vivo radiance of luciferase activity of excised lungs. P values determined by one-way anova, * p<0.05, **p<0.01, ***p<0.001,**** p<0.0001. N=5-6 mice.

Lasofoxifene + palbociclib is more effective than fulvestrant + palbociclib and significantly more effective than fulvestrant alone at inhibiting metastasis to the liver

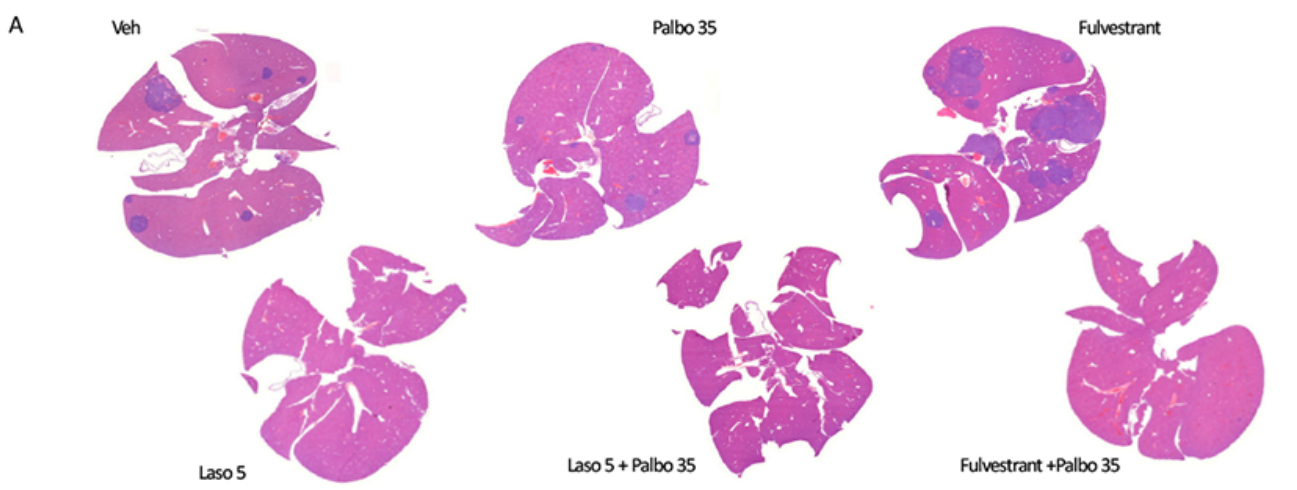
A
Veh
Palbo 35
Fulvestrant
Laso 5
Laso 5 + Palbo 35
Fulvestrant +Palbo 35

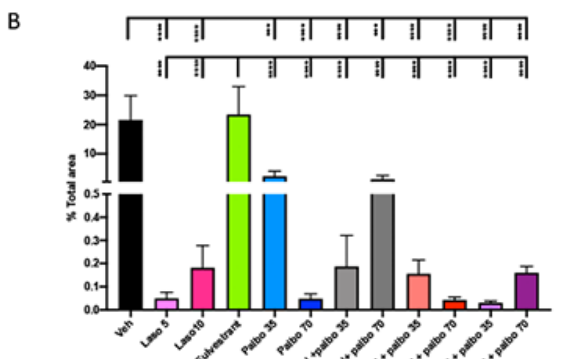
B
% Total area

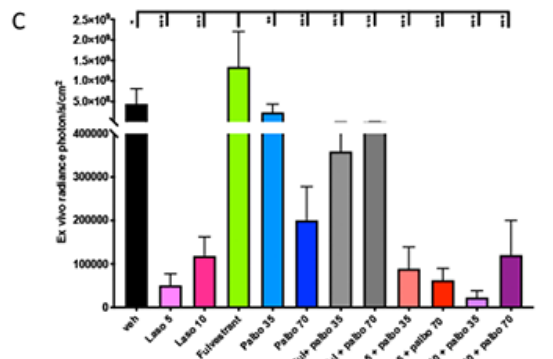
C
Ex vivo radiance photon/s/cm²

Fig 4: **A)** Representative H&E staining of livers. **B)** quantitation of the H&E performed with Image J (FJI) showing the area of tumor cells normalized to the total liver area. **C)** Quantitation of ex-vivo radiance of luciferase activity of excised livers. P values determined by one-way anova, * p<0.05, **p<0.01, ***p<0.001,**** p<0.0001. N=5-6 mice.

Lasofoxifene + palbociclib is more effective than fulvestrant + palbociclib and significantly more effective than fulvestrant alone at inhibiting metastasis to bone and brain

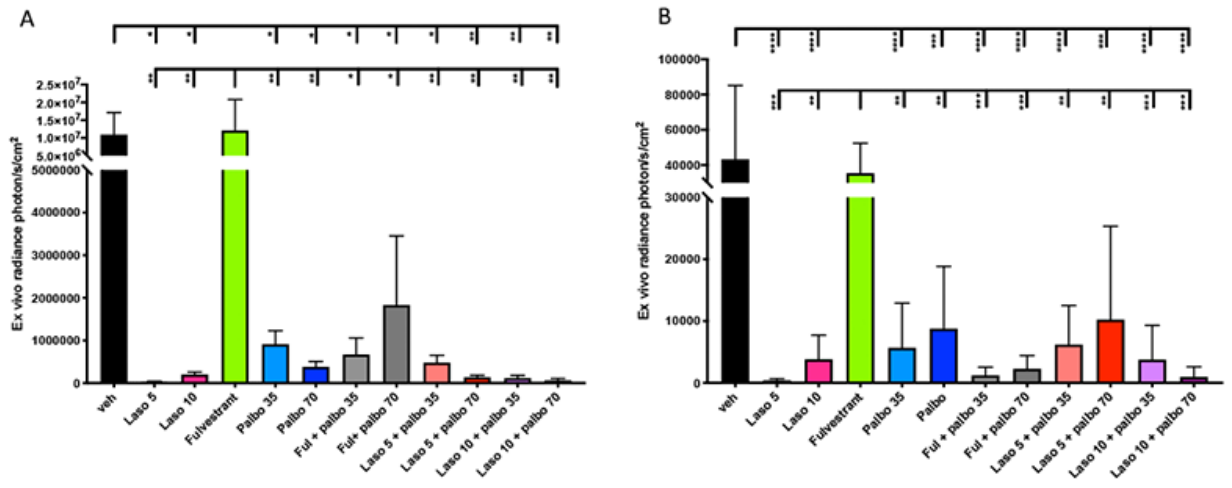
A
Ex vivo radiance photon/s/cm²
B
Ex vivo radiance photon/s/cm²

Fig 5: A) Ex-vivo radiance of luciferase activity quantitation of excised long bones (N= 10-12). **B)** Ex-vivo radiance of luciferase activity quantitation of excised brains. P values determined by one-way anova, * p<0.05, **p<0.01, ***p<0.001,**** p<0.0001, N= 5-6 mice.

CONCLUSIONS

- **Lasofoxifene alone or in combination with palbociclib is an effective inhibitor of tumor growth for the MCF7 Y537S ERα+ MBC xenograph model.**
- **Lasofoxifene + palbociclib is significantly more effective than fulvestrant + palbociclib at inhibiting tumor growth and metastasis to the lung, liver, brain and long bones. Excellent correlation between ex vivo imaging and IHC or H&E assessments of metastatic burden.**
- **Importantly, lasofoxifene + palbociclib produces a synergistic inhibition of tumor growth and metastasis. In addition, lasofoxifene appears to drive synergism in combination with palbociclib, whereas palbociclib appears to drive syngergism in combination with fulvestrant.**
- **Lasofoxifene shows promise as an effective therapy for women with ER+ metastatic breast cancers that express constitutively active ERα mutations, such as D538G and Y537S. A phase 2 clinical trial is currently under way to evaluate Lasofoxifene versus Fulvestrant in Advanced or Metastatic ER+/HER2− Breast Cancer with an ESR1 Mutation.**