112 (Abstract No.)

An Open-label, Randomized, Multi-center Phase 2 Study Evaluating the Activity of Lasofoxifene Relative to Fulvestrant for the Treatment of Postmenopausal Women with Locally Advanced or Metastatic ER+/HER2- Breast Cancer with an ESR1 Mutation

Paul V. Plourde MD[1], Lee S Schwartzberg MD[2], Geoffrey L Greene PhD[3], David J Portman MD[1], Barry S Komm PhD[1], Simon N Jenkins PhD[1], Ping-Yu Liu PhD[4], Miriam D Portman MD[1] and Matthew P Goetz MD[5]

[1] Sermonix Pharmaceuticals, Columbus OH, [2] West Cancer Center, Memphis TN, [3] U. of Chicago, Chicago IL, [4] Fred Hutchinson Cancer Research Center Seattle, WA, [5] Mayo Clinic, Rochester, MN

Presented at the 2018 San Antonio Breast Cancer Symposium®
December 4-8, 2018

INTRODUCTION

One cause of endocrine resistance is the presence of activating ESR1 mutations within breast cancer cells, which from preclinical studies demonstrate constitutive, ligand-independent estrogen receptor (ER) activation[1].

Because of the relative endocrine resistance of these mutations to approved hormonal agents, there is a significant unmet medical need for the investigation of new targeted endocrine therapies that have potent activity in the presence of ESR1 mutations.

Sermonix is initiating a Phase 2 clinical trial in postmenopausal women with ER+HER2- breast cancer who progressed on an aromatase inhibitor (AI) in combination with a CDK 4/6 inhibitor, and who have an ESR1 mutation. The clinical trial will evaluate investigational lasofoxifene, a potent selective estrogen receptor modulator (SERM).

FIGURE 1.

A) Many mutations have been described in the ligand binding domain of the estrogen receptor. B) Breakdown of types of mutations vs WT found in ER+/HER2- mBC patients with prior endocrine therapy, and C) chart showing prevalence of ESR1 mutations, both adapted from Kuang[2]

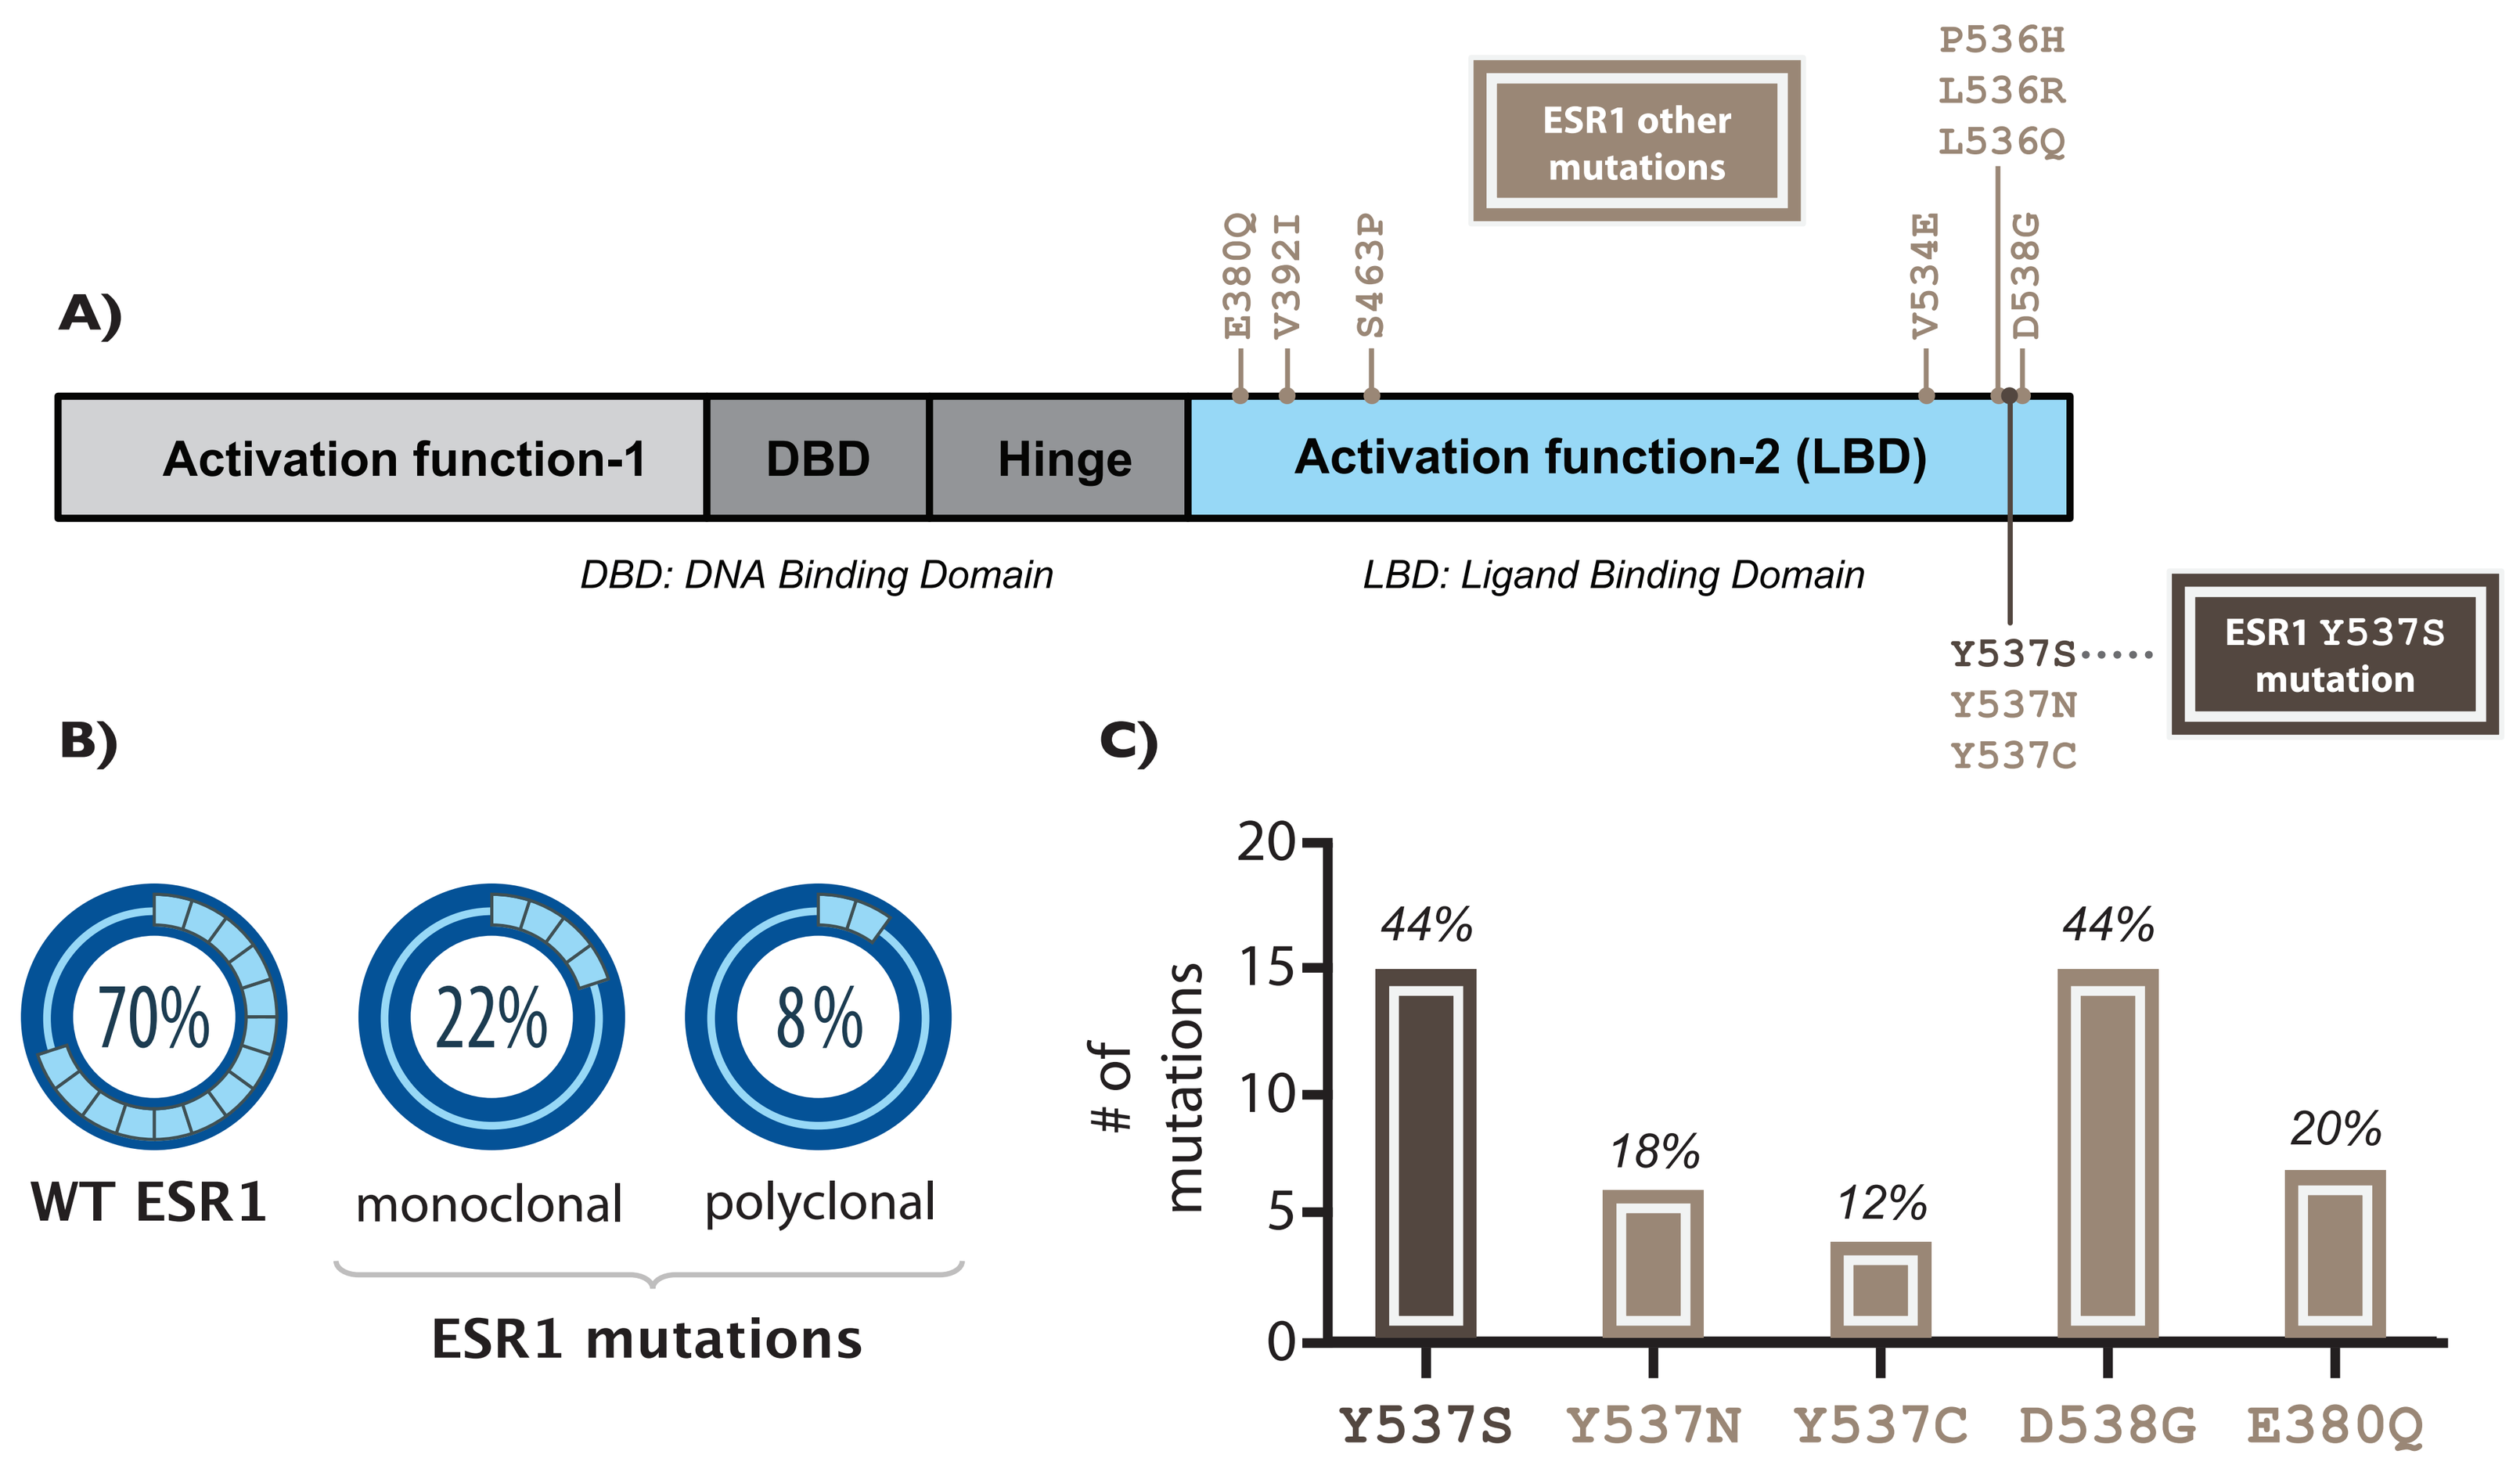
A)
ESR1 other mutations
P536H
L536R
L536Q
E380Q
V392I
S463P
V534E
D538G
Activation function-1
DBD
Hinge
Activation function-2 (LBD)
DBD: DNA Binding Domain
LBD: Ligand Binding Domain
Y537S
Y537N
Y537C
ESR1 Y537S mutation
B)
70%
22%
8%
WT ESR1
monoclonal
polyclonal
ESR1 mutations
C)
of mutations
20
15
10
5
0
44%
18%
12%
44%
20%
Y537S
Y537N
Y537C
D538G
E380Q

LASOFOXIFENE

- Lasofoxifene has been shown to inhibit tumor proliferation within in vitro and in vivo breast cancer animal models[3,4]
- In vivo mice studies have also demonstrated dose dependent efficacy in ESR1 mutated breast cancer cells as well as a reduction in metastasis when compared to fulvestrant[5]
- In humans, lasofoxifene has excellent oral bioavailability and volume of distribution, and is 99% bound in plasma
- The drug has been well tolerated with oral doses as high as 10 mg given once a day (mg/d)
- In a large five-year osteoporosis trial, an 83% reduction of ER+ breast cancers with lasofoxifene versus placebo was observed[6]

FIGURE 2.

Schematic of estrogen receptor (ER) conformational states: WT unbound, WT bound, ER mutated, & ER mutated bound to lasofoxifene. Lasofoxifene binding causes an "OFF" state in ER mutated proteins.

See abstract #177/program #PD7-09 for further preclinical lasofoxifene data at this conference. Figure recreated based on illustration from Katzenellenbogen[7], with permission.

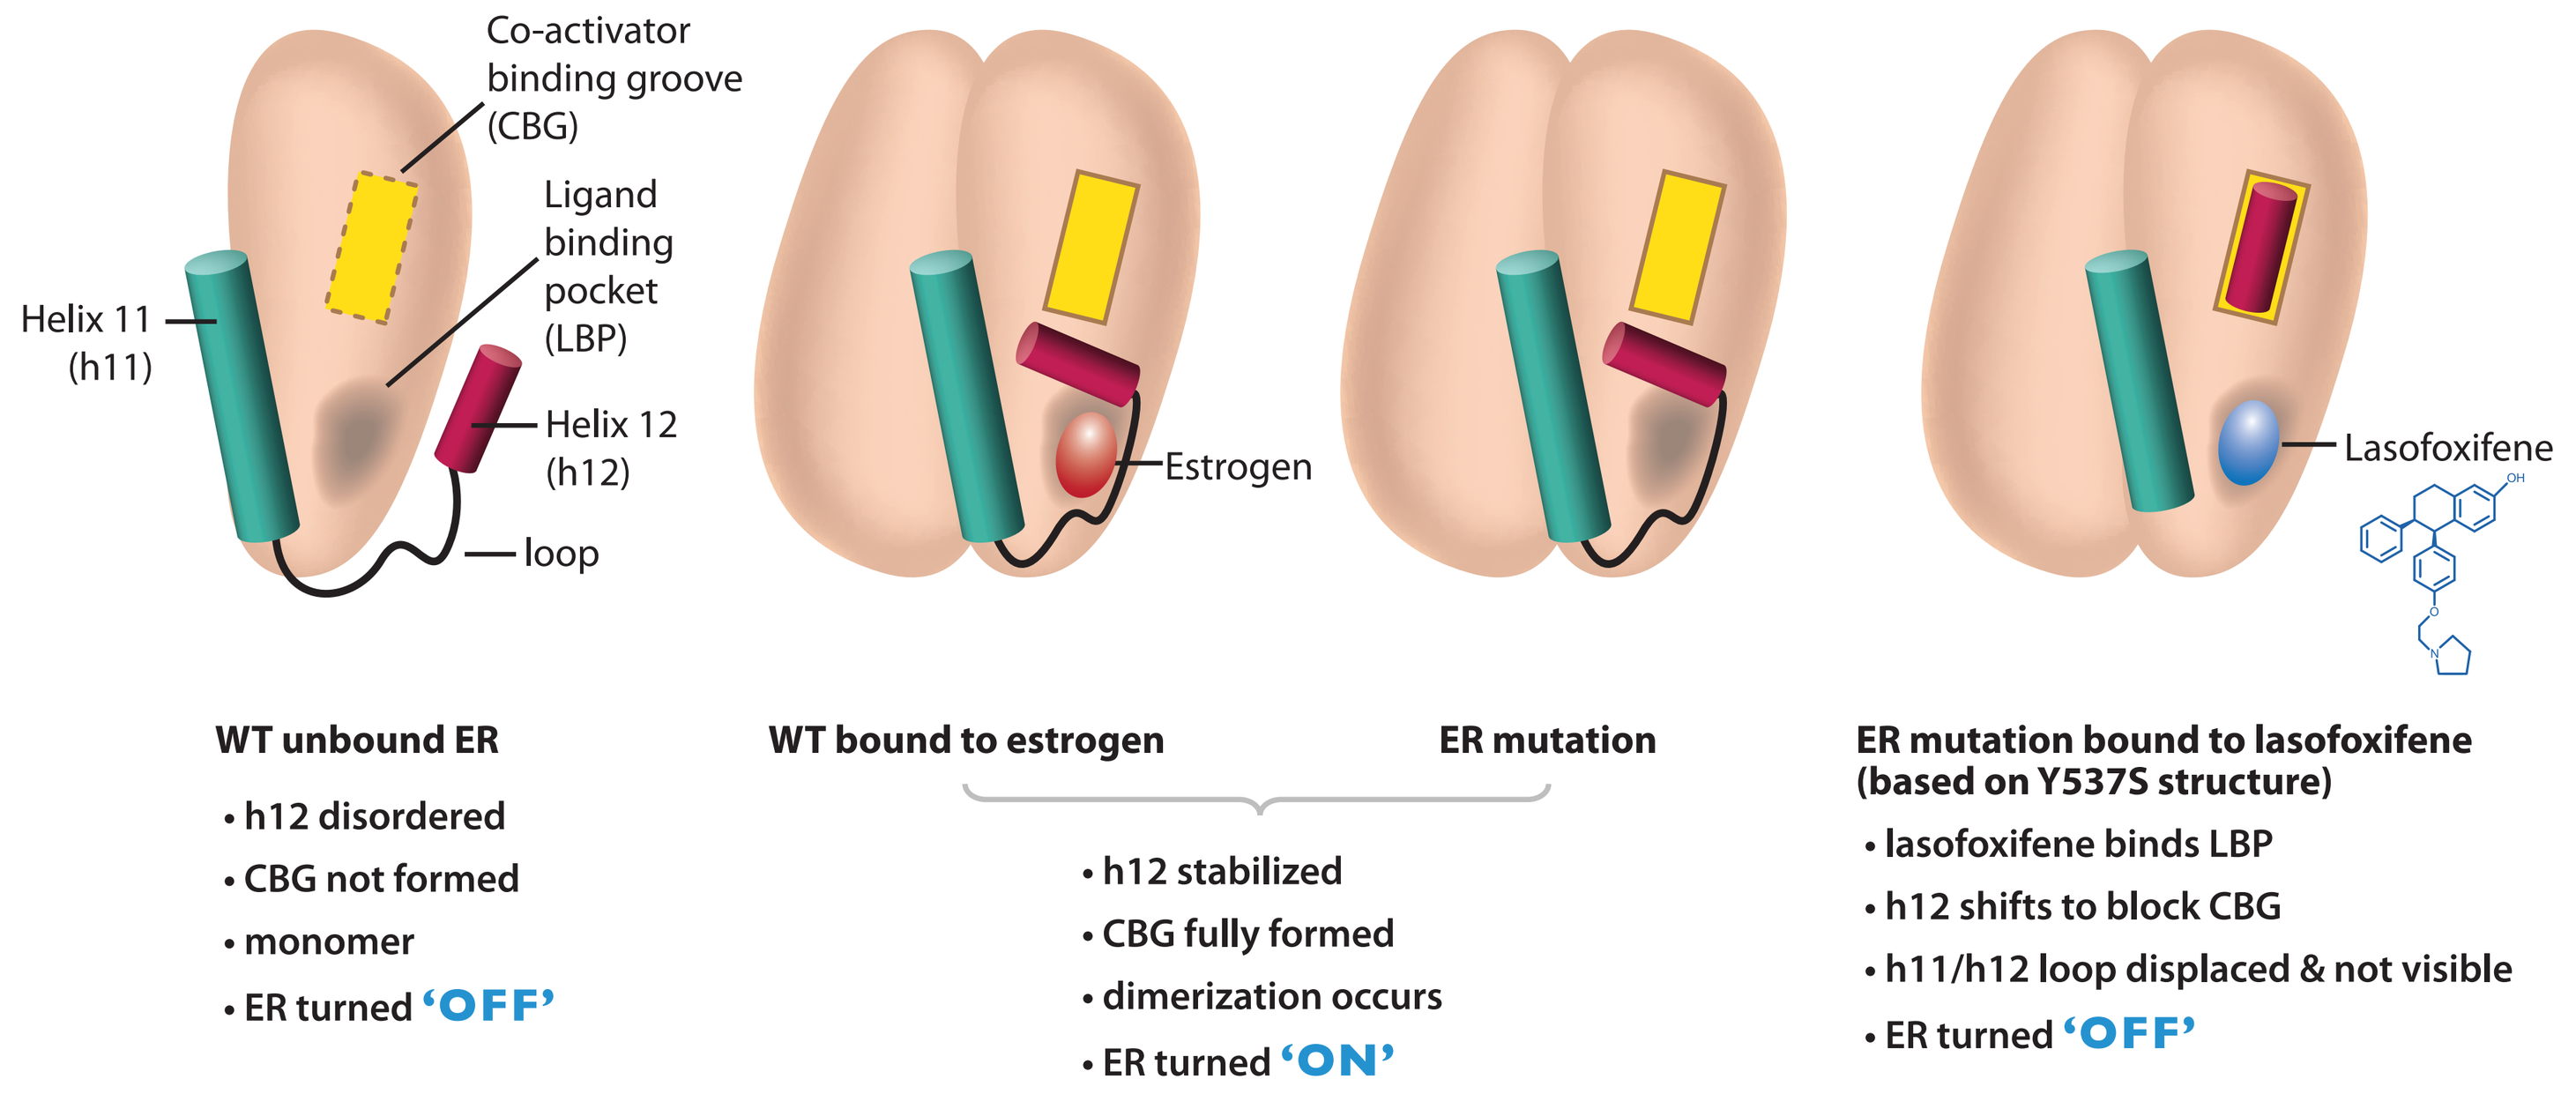
Co-activator binding groove (CBG)
Ligand binding pocket (LBP)
Helix 11 (h11)
Helix 12 (h12)
loop
Estrogen
Lasofoxifene
WT unbound ER
• h12 disordered
• CBG not formed
• monomer
• ER turned 'OFF'
WT bound to estrogen
ER mutation
• h12 stabilized
• CBG fully formed
• dimerization occurs
• ER turned 'ON'
ER mutation bound to lasofoxifene (based on Y537S structure)
• lasofoxifene binds LBP
• h12 shifts to block CBG
• h11/h12 loop displaced & not visible
• ER turned 'OFF'

PHASE 2 STUDY OVERVIEW

STUDY DESIGN

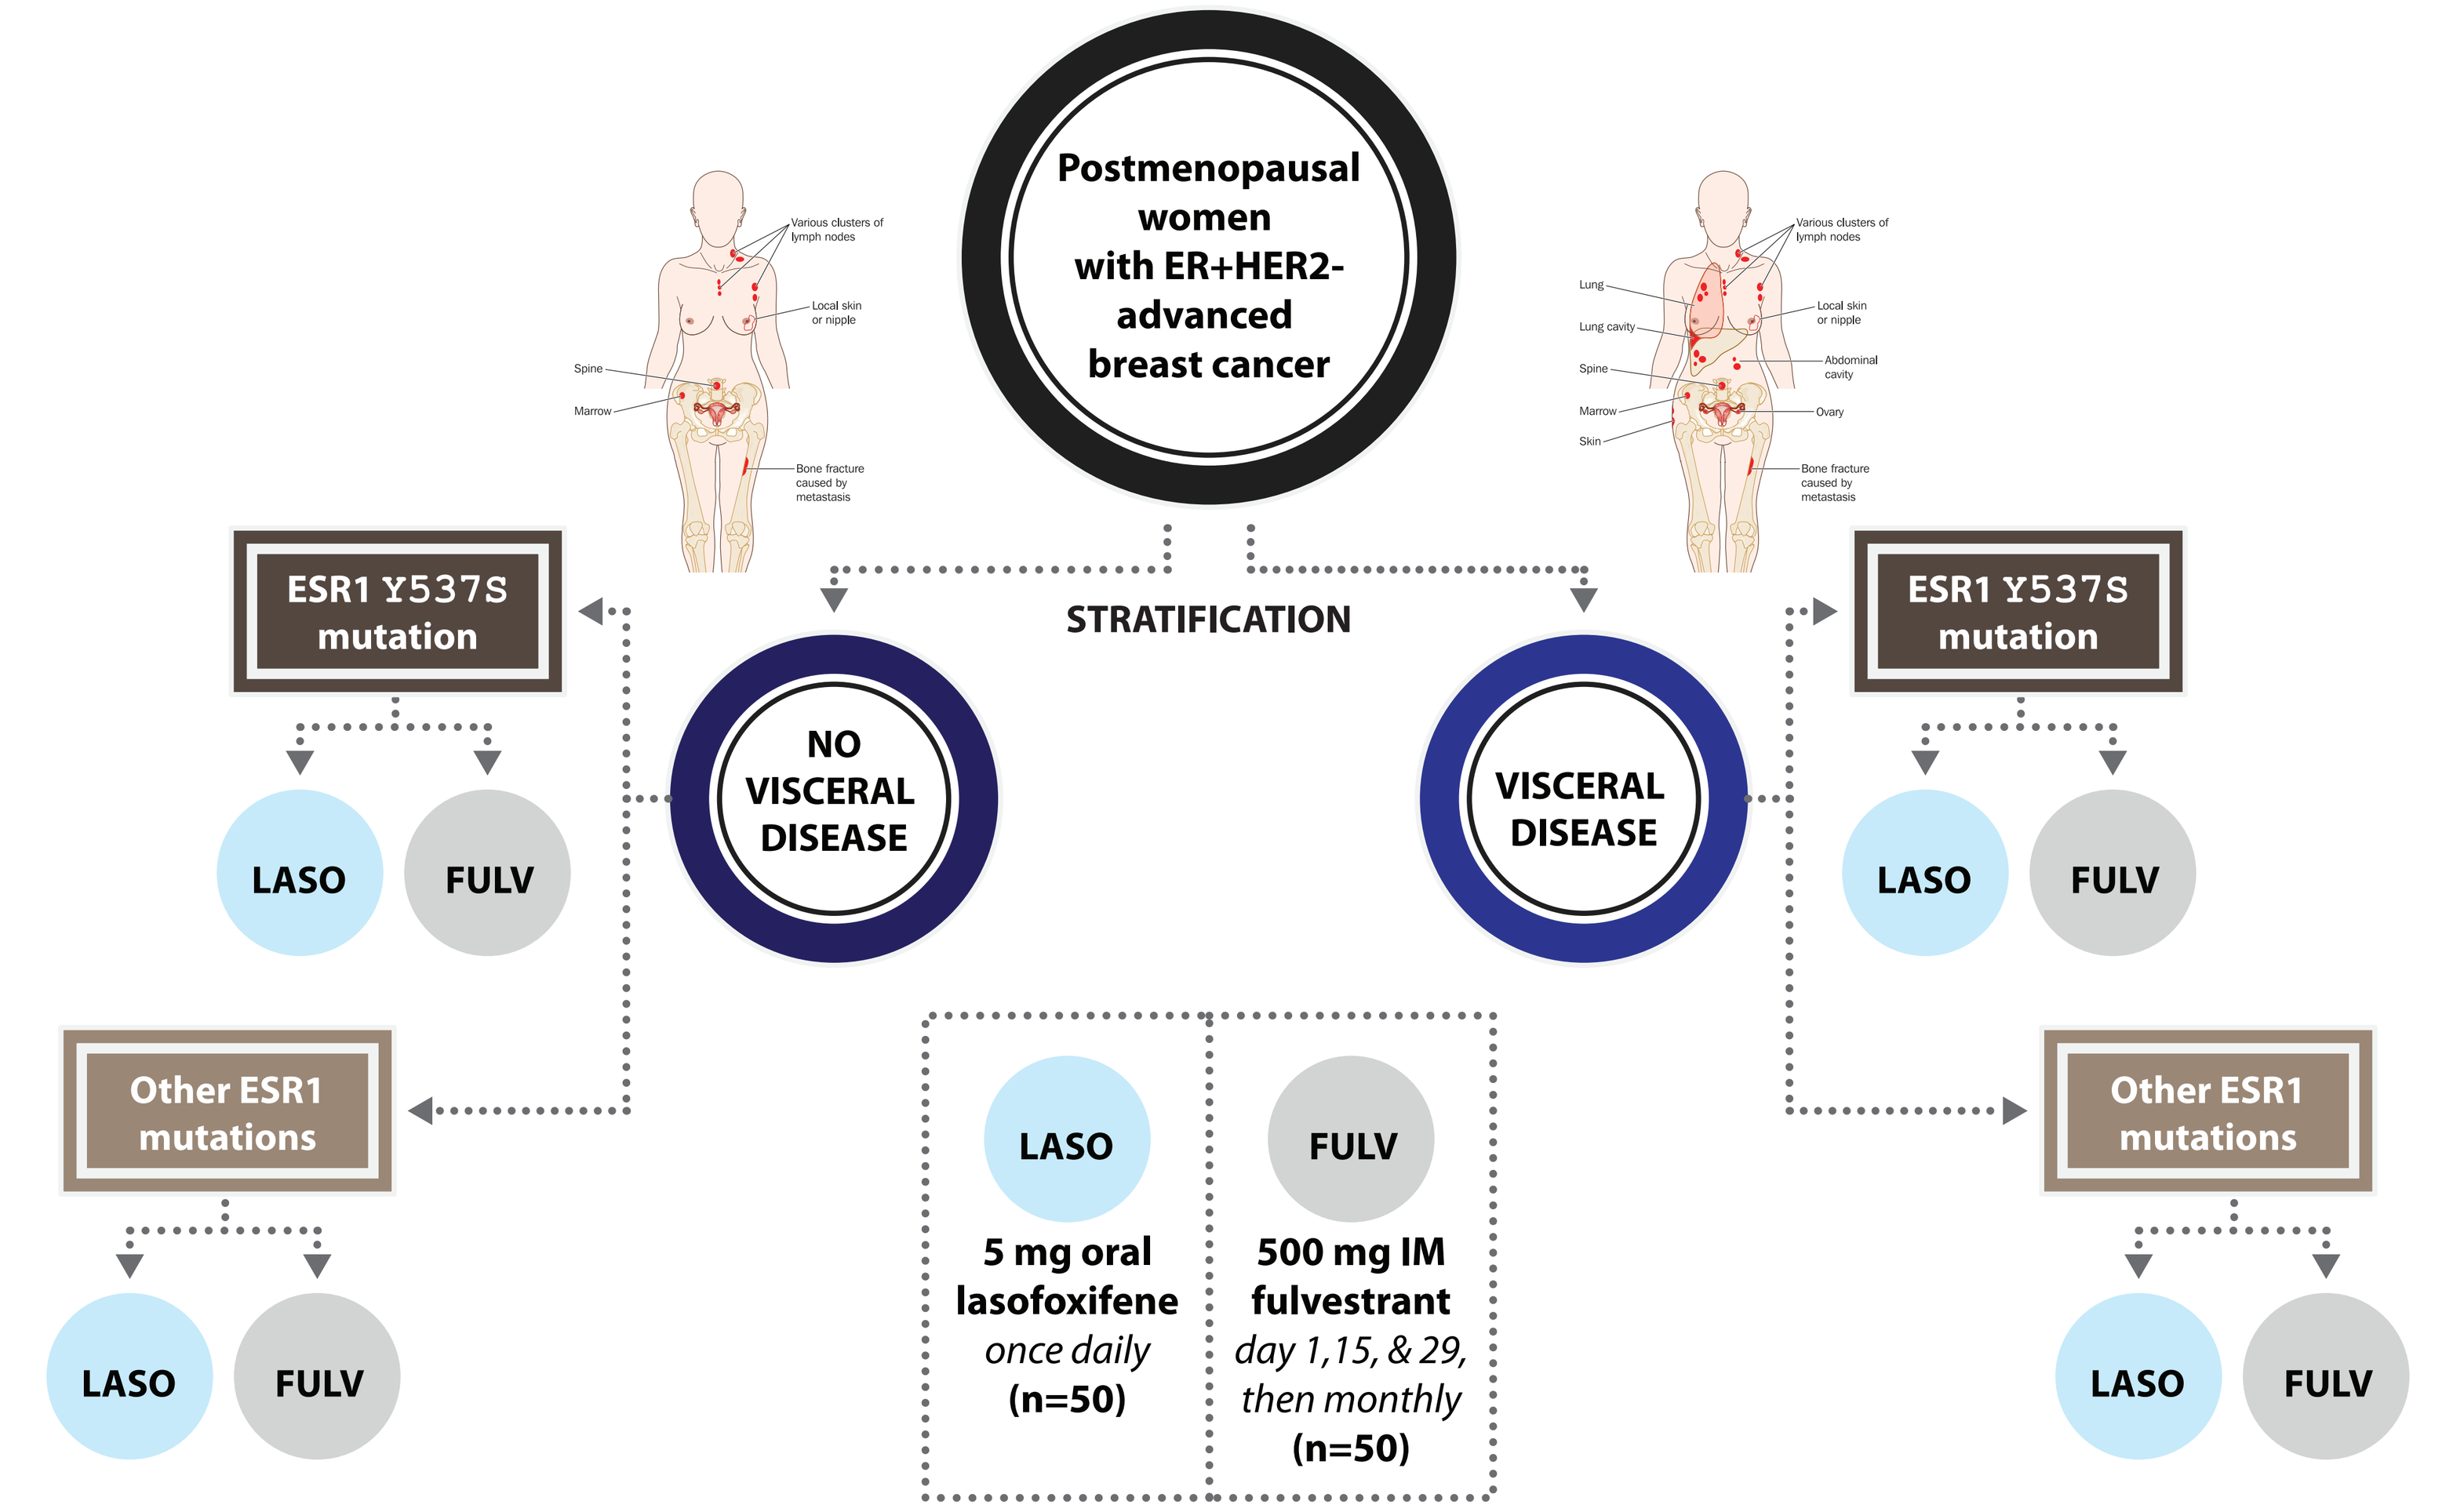
Postmenopausal women with ER+HER2- advanced breast cancer
STRATIFICATION
NO VISCERAL DISEASE
VISCERAL DISEASE
ESR1 Y537S mutation
Other ESR1 mutations
LASO
FULV
5 mg oral lasofoxifene
once daily
(n=50)
500 mg IM fulvestrant
day 1,15, & 29, then monthly
(n=50)

PRIMARY ENDPOINT

The progression free survival (PFS) of 5 mg lasofoxifene relative to fulvestrant

SECONDARY ENDPOINTS

- Clinical benefit rate (CBR)
- Duration of response
- Objective response rate (ORR)
- Quality of life (QoL)
- Safety of lasofoxifene
- Time to response

TREATMENT

- Open label, randomized, multicenter study
- Screening tests within thirty days of enrollment
- Subjects treated until documented disease progression
- Full recruitment into study estimated to occur within twelve to sixteen months, with another twelve months of follow up before the primary outcome measure is analyzed

VISIT SCHEDULE

- Vital signs, height, weight, physical exam, and adverse event (AE) assessment at each visit
- Bone scan every two months if metastatic bone lesions present at baseline
- MRI or CT of the chest, abdomen and pelvis every two months or sooner if clinically indicated
- CBC with differential, routine chemistries, and sample for PK analysis every visit

INCLUSION CRITERIA

1. Postmenopausal women with:
 a. locally advanced or metastatic breast cancer with either measurable or non-measurable lesions
 b. progression on an AI in combination with a CDK 4/6 inhibitor for advanced breast cancer
2. If possible, a biopsy of metastatic breast cancer tissue will be obtained to confirm ER+ and HER2- disease
3. At least one or more of the following ESR1 mutations: Y537S, Y537N, Y537C, D538G, L536Q, E380Q, or S463P assessed from tumor-free DNA in blood
4. Received one chemotherapy regimen in the neo-adjuvant or adjuvant setting prior to entry into the trial
5. ECOG performance score of 0 or 1

EXCLUSION CRITERIA

Subjects who meet any of the following criteria will be excluded from entering the trial:

1. Prior use of any SERM in the adjuvant or metastatic setting
2. Presence of brain metastasis or lymphangitic carcinomatosis involving the lung
3. Impending visceral crisis as assessed by the investigator
4. Radiotherapy within 30 days prior to randomization
5. History of long QT_C syndrome or a QT_C of >480 ms
6. History of a pulmonary embolus (PE) or deep vein thrombosis (DVT) within the last 6 months, or any known thrombophilia
7. History of a positive human immunodeficiency virus (HIV), hepatitis B virus (HBV) or hepatitis C virus (HCV) at screening

STATISTICAL PLAN

PFS will be compared between the lasofoxifene and fulvestrant arms using the stratified log-rank test. Median PFS for each treatment arm will be estimated by the Kaplan-Meier method and the lasofoxifene over fulvestrant hazard ratio will be estimated by the Cox proportional hazards model.

It is assumed that lasofoxifene will double the median PFS compared to fulvestrant in this ESR1 mutation population, i.e., from four months to eight months, for a lasofoxifene over fulvestrant hazard ratio of 0.5.

For ITT subjects, fifty for each of the two arms will be targeted for enrollment. Conservatively assuming forty evaluable subjects in each arm and a one-year accrual period with one additional year follow-up, the power is 0.89 using 1-sided stratified log-rank $p < 0.05$ defining a positive study. The same set-up has a power of 0.82 for reaching a 1-sided $p < 0.025$ for a conclusive outcome. However, should there be few lost-to-follow cases and the total evaluable N be close to one hundred, the power for reaching a 1-sided $p < 0.05$ and a 1-sided $p < 0.025$ would be ~0.94 and 0.89, respectively.

CONCLUSIONS

- Lasofoxifene has a well-characterized safety profile from Phase 1-3 studies in over 10,000 women in non-oncology indications
- Lasofoxifene significantly reduced the incidence of ER+ breast cancer by 83% in a large five-year osteoporosis study
- In non-clinical models of breast cancer, lasofoxifene demonstrated superior efficacy to fulvestrant in inhibiting tumor growth and metastases to liver and lung, particularly in Y537S mutations[8]
- Non-clinical finding may be explained by lasofoxifene's binding affinity to mutated estrogen receptors and its favorable pK characteristics
- These aspects, coupled with lasofoxifene-specific conformational changes to the receptor, effectively block receptor mediated proliferation
- These data clearly support further investigation of this molecule in a clinical setting
- This trial will be the first prospective controlled randomized study investigating a SERM for the targeted treatment of ER+ advanced breast cancers with an ESR1 mutation
- If successful, lasofoxifene will confer a major benefit to women with metastatic breast cancer and thereby delay the need for toxic chemotherapy

Study centers are now being considered and trial to be initiated in January 2019.

Investigators interested in participating should contact our CRO partner Linical-Accelovance, Laura Smoral, lsmoral@linical.accelovance.com.

This presentation is the intellectual property of the author/presenter. Contact David Portman, Founder and CEO, at DPortman@sermonixpharma.com for permission to reprint and/or distribute.

REFERENCES

1. Toy W, Shen Y, Won H, et al. ESR1 ligand-binding domain mutations in hormone-resistant breast cancer. Nat Genet. 2013;45:1439-45.
2. Kuang, Y, Siddiqui B, Hu J, et al. Unraveling the clinicopathological features driving the emergence of ESR1 mutations in metastatic breast cancer. Nature Partner Journal Breast Cancer Mar 2018;22:1-10.
3. Ke HZ, Qi H, Crawford RT, et al. Lasofoxifene (CP 336,156), a selective estrogen receptor modulator prevents bone loss induced by aging and orchidectomy in the adult rat. Endocrinology: 2000;141:1338-44.
4. Wardell SE, Nelson ER, McDonnell DP. From empirical to mechanism-based discover of clinically useful selective estrogen receptor modulators (SERMs). Steroids: 2014;90:30-38.
5. Lainé M, Greene M, Chang Y, et al. Lasofoxifene efficacy in a mammary intraductal (MIND) xenograft model of ERα+ breast cancer. American Society of Clinical Oncology 2018; Abstract E13054.
6. LaCroix AZ, Powles T, Osborne CK, et al. Breast cancer incidence in the randomized PEARL trial of lasofoxifene in postmenopausal osteoporotic women. J Natl Cancer Inst. 2010;102:1706-15.
7. Katzenellenbogen JA, Mayne CG, Katzenellenbogen BS, Greene GL, Chandarlapaty S. Structural underpinnings of oestrogen receptor mutations in endocrine therapy resistance. Nat Rev Cancer. 2018 Jun;18(6):377-388.
8. Laine et al., poster at this conference: abstract #177/program #PD7-09