A Preliminary Assessment of Knowledge, Attitudes, and Awareness Surrounding Precision Medicine, ESR1 Mutations, and Biomarker Testing Amongst Medical Oncologists

Shari B. Golfarb MD[1], Matthew P. Goetz MD[2], Paul V. Plourde MD[3], Elizabeth Y. Attias ScD[3], David J. Portman MD[3]

[1] Memorial Sloan Kettering, New York, NY; [2] Mayo Clinic, Rochester, MN; [3] Sermonix Pharmaceuticals, Columbus, OH

Presented at the 2018
San Antonio Breast
Cancer Symposium®
December 4-8, 2018

INTRODUCTION

The estrogen receptor has long been known to be a target for hormone receptor sensitive cancers and continues to guide therapy decisions[1]. Since the identification of HER2 as a target, there has been significant progress in the use of biomarkers in breast cancer, and recent advances in treating BRCA germ-line mutations are being translated into clinical development and treatment practice paradigms. However, to date, there have not been significant developments in the ER+HER2- biomarker arena of somatic mutations.

Mutations of the ESR1 gene are recognized as a fundamental mechanism of endocrine therapy resistance, with acquired endocrine resistance having been shown to evolve under selective pressure from endocrine treatments[2]. Recent findings suggest that ESR1 mutations are present in up to 40% of metastatic breast cancer patients treated with previous aromatase inhibitors (AIs), confer constitutive activity to the receptor even in the absence of estrogen, are insensitive to AIs, and have decreased binding affinity to currently available endocrine therapies such as fulvestrant[3,4]. A biomarker to identify ESR1 mutations, especially from a liquid biopsy, in women with ER+HER2- metastatic breast cancer (mBC) would provide the ability to target the mutations with a precision medicine approach.

The objective of this study was to evaluate the perceptions of precision medicine and biomarker testing specific to ESR1 mutations among medical oncologists. In addition, clinicians' perceptions of the future extent and implications of ESR1 mutation testing through the use of a companion diagnostic and treatment implications were explored.

FIGURE 1.

ESR mutations stabilize active conformation of ESR1 ligand binding domain in the absence of estrogen agonists. *Xray crystallographic images courtesy of Dr. Geoffrey Greene, U of Chicago; Also at this conference, see abstract #177/program #PD7-09 for further xray structures relating to ESR mutations.*

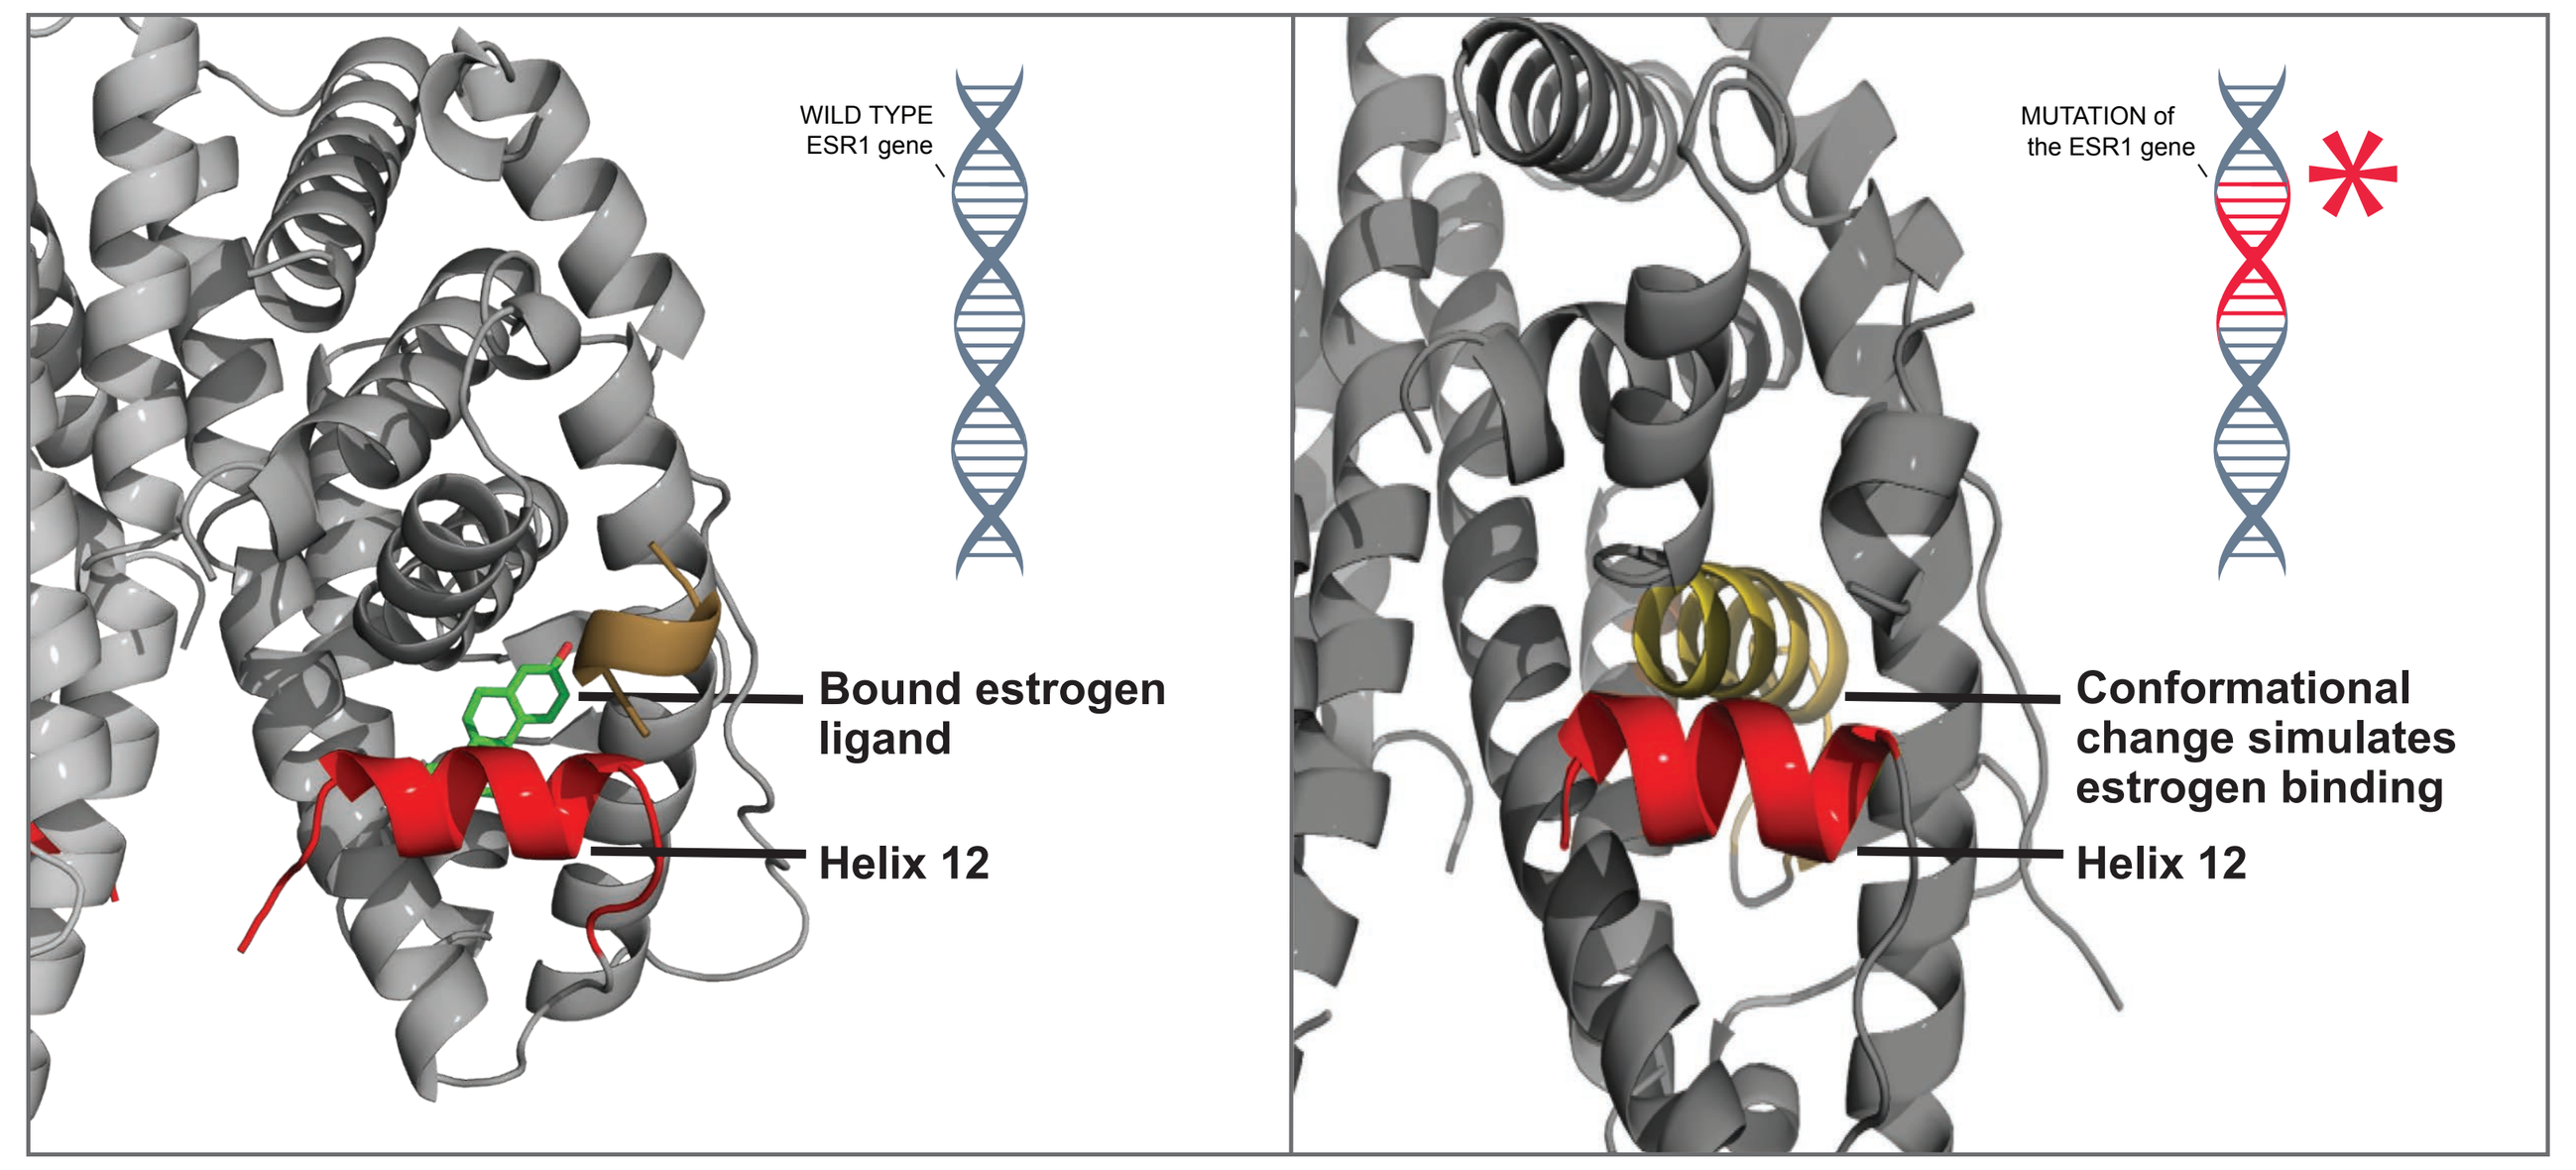
WILD TYPE ESR1 gene
Bound estrogen ligand
Helix 12
MUTATION of the ESR1 gene
Conformational change simulates estrogen binding
Helix 12

E2 Bound to Wild Type Estrogen Receptor

ESR1 D583G Mutation, No Ligand

FIGURE 2.

Precision medicine: Developing custom therapies to treat breast cancer based on the presence of the ESR1 mutation in ER+HER- breast cancer patient population.

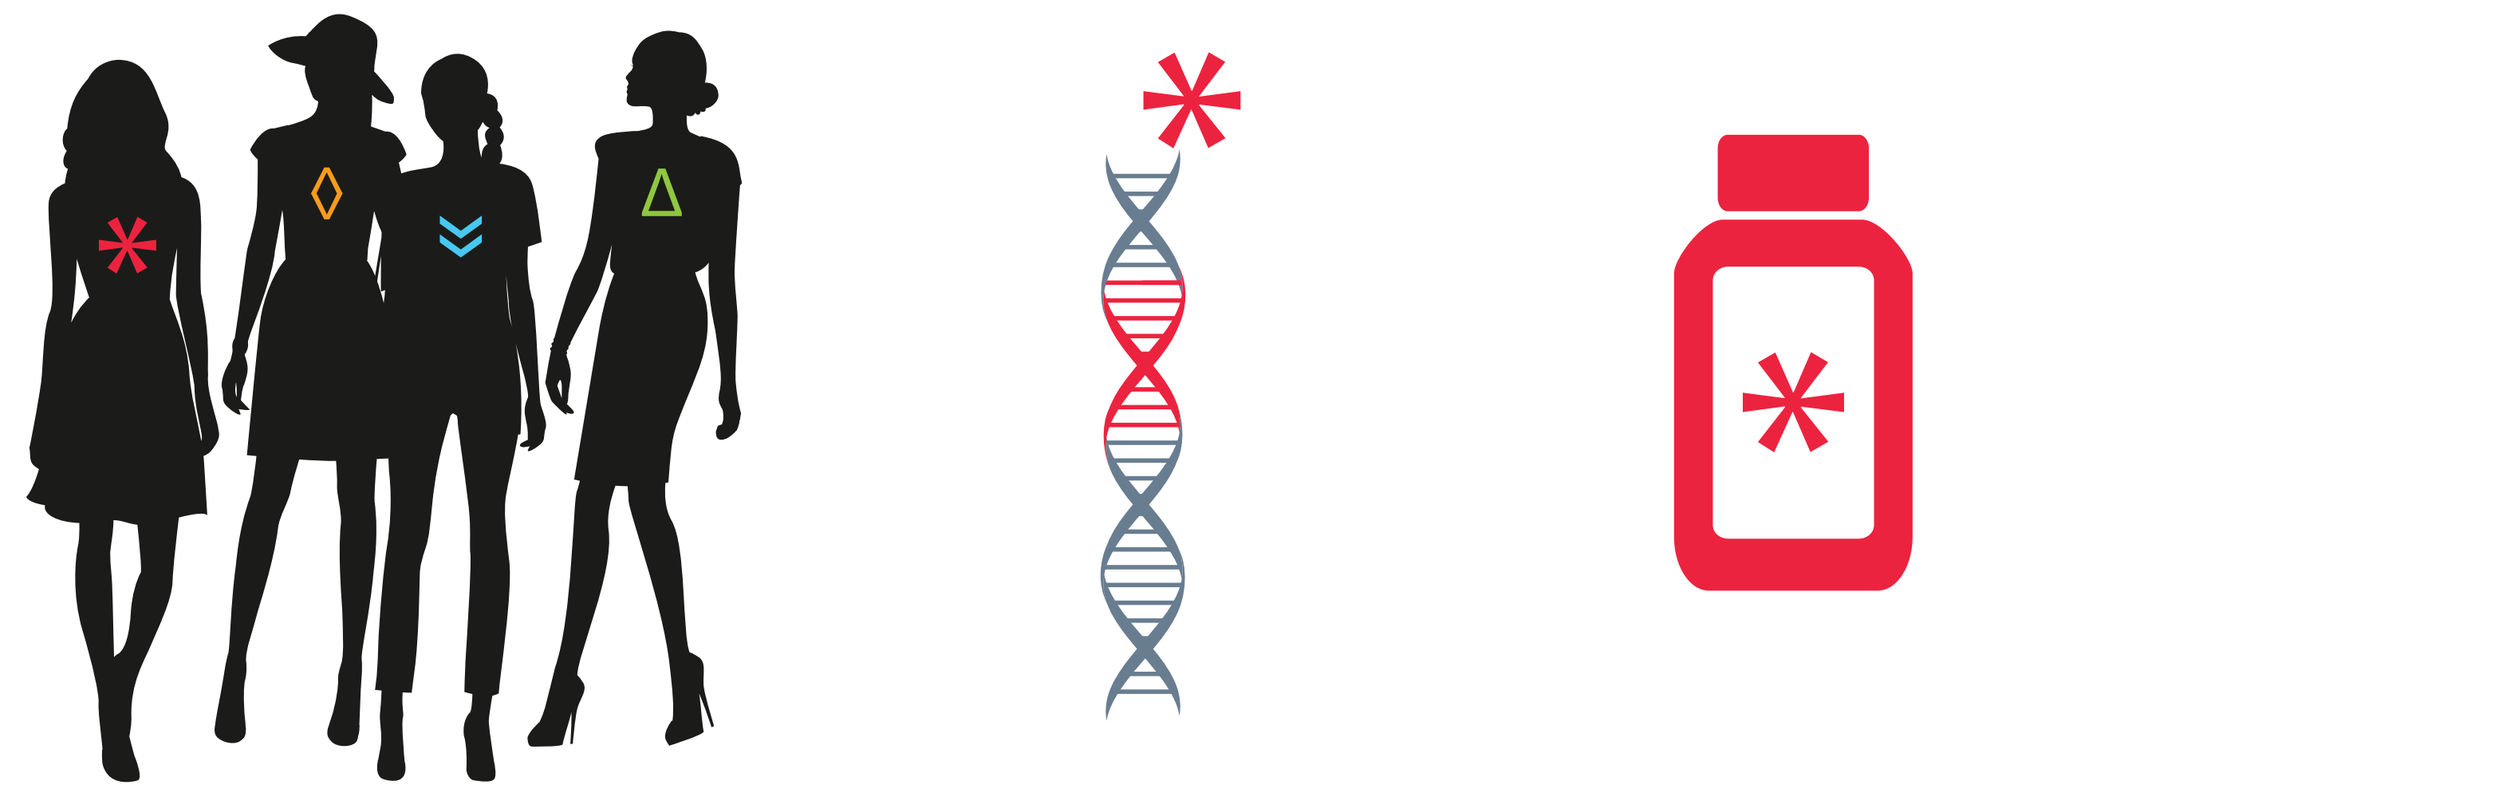

METHODOLOGY

Web-assisted, sixty-minute telephone interviews were conducted with ten non-academic medical oncologists.

Physician were prescreened to ensure board certification and practice experience between 2 and 35 years.

Participant spent >30% of their time on direct patient care and managed the treatment of more than 30 different cancer patients per month, with a minimum of 15 breast cancer patients, including at least 5 with mBC and at least 1 patient with an ESR1 mutation.

Physicians quantitatively assigned treatment allocation for 1st, 2nd and 3rd line therapies, as well as other market-based research questions specific to an investigational product for treating mBC in patients with ESR1 mutations. Quantitative research findings were also captured by an expert interviewer during the web-based portion of the survey.

FIGURE 3.

Interviewed physicians' breast cancer patient cascade.

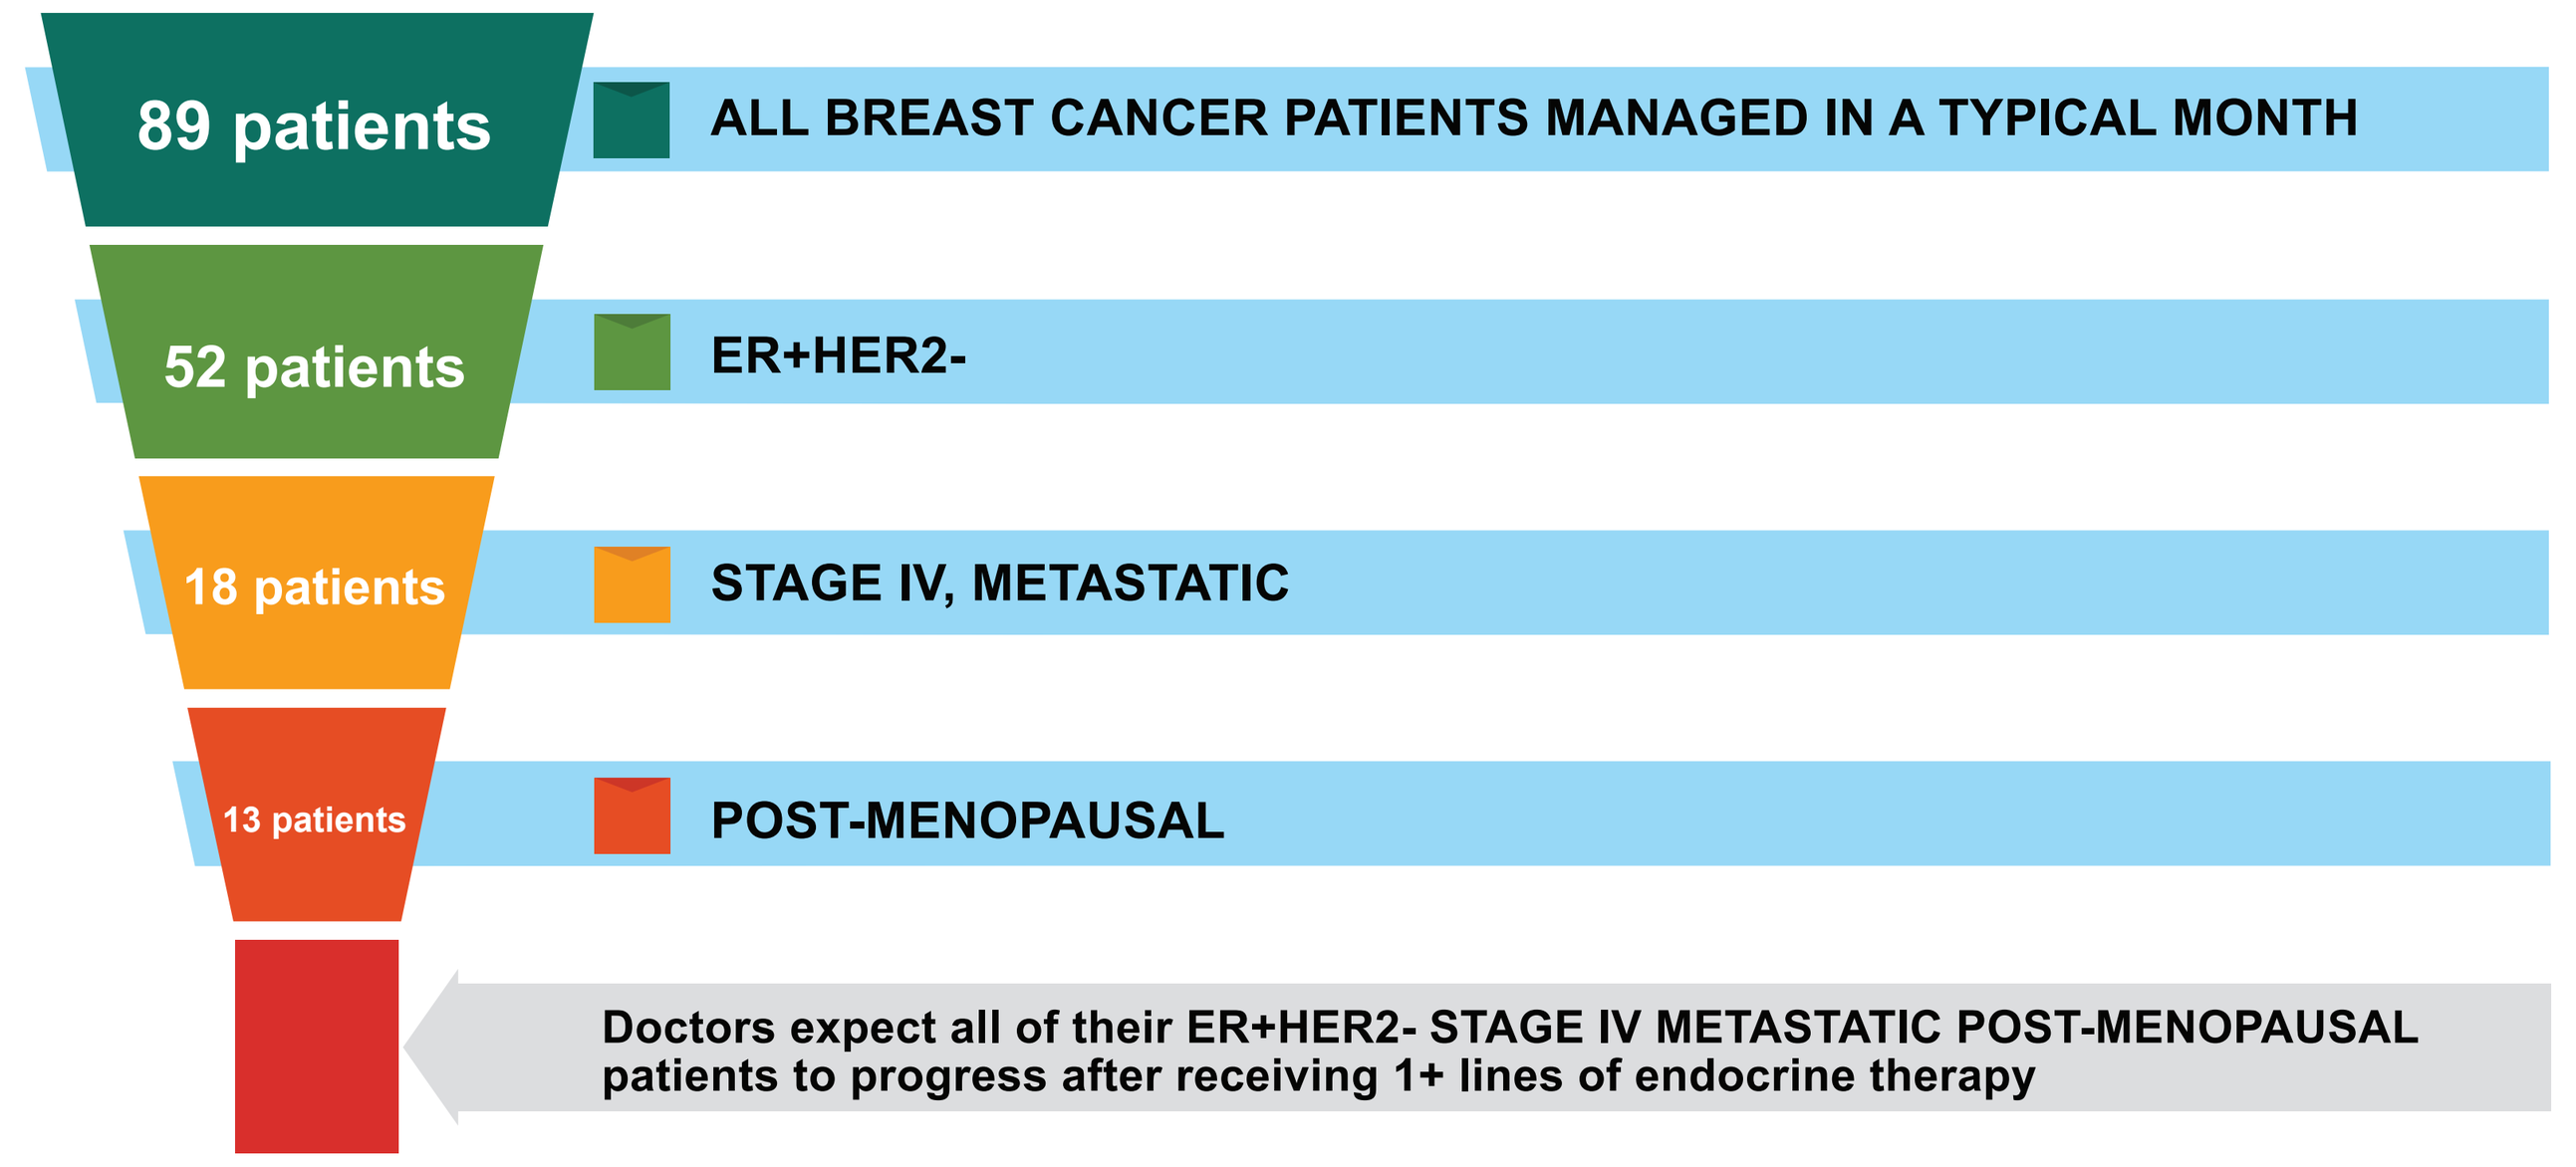
89 patients
ALL BREAST CANCER PATIENTS MANAGED IN A TYPICAL MONTH
52 patients
ER+HER2-
18 patients
STAGE IV, METASTATIC
13 patients
POST-MENOPAUSAL
Doctors expect all of their ER+HER2- STAGE IV METASTATIC POST-MENOPAUSAL patients to progress after receiving 1+ lines of endocrine therapy

PRACTICING CLINICAL ONCOLOGIST INTERVIEW RESULTS

PHYSICIANS BELIEVE PERSONALIZED MEDICINE WILL:
- allow for better efficacy
- help increase overall survival rates by targeting ESR1 mutation

PHYSICIANS HAVE:
- a high desire for personalized treatment in breast cancer
- limited awareness of new somatic genetic mutation markers
- expectation that more genetic markers will emerge in 2-3 years
- some knowledge of ESR1 mutations
- satisfaction with 1st line treatments in ER+HER2- mBC
- dissatisfaction with 2nd and 3rd line treatments in ER+HER2- mBC and with existing treatment options for ESR1 mutation patients

QUESTIONED ABOUT ESR1 MUTATIONS, PHYSICIANS SAID:

"It may have a role in the inability of currently used hormonal therapies to bind to the estrogen receptor"

"It is more common among treatment-experienced patients"

"It may be acquired with selective pressure from AIs or other endocrine therapies"

"It is associated with poorer outcomes"

"Some endocrine therapies should be avoided in patients with ESR1 mutations"

According to Medscape Oncology[5], "in a study of more than 50 oncologists at 5 US cancer centers, genomic tumor profiling of advanced cancers changed clinical practice 23% of the time."

This aligns with our present market research survey results, where we determined an effective treatment for ESR1 mutations would garner significant market share in the 2nd and 3rd line treatments.

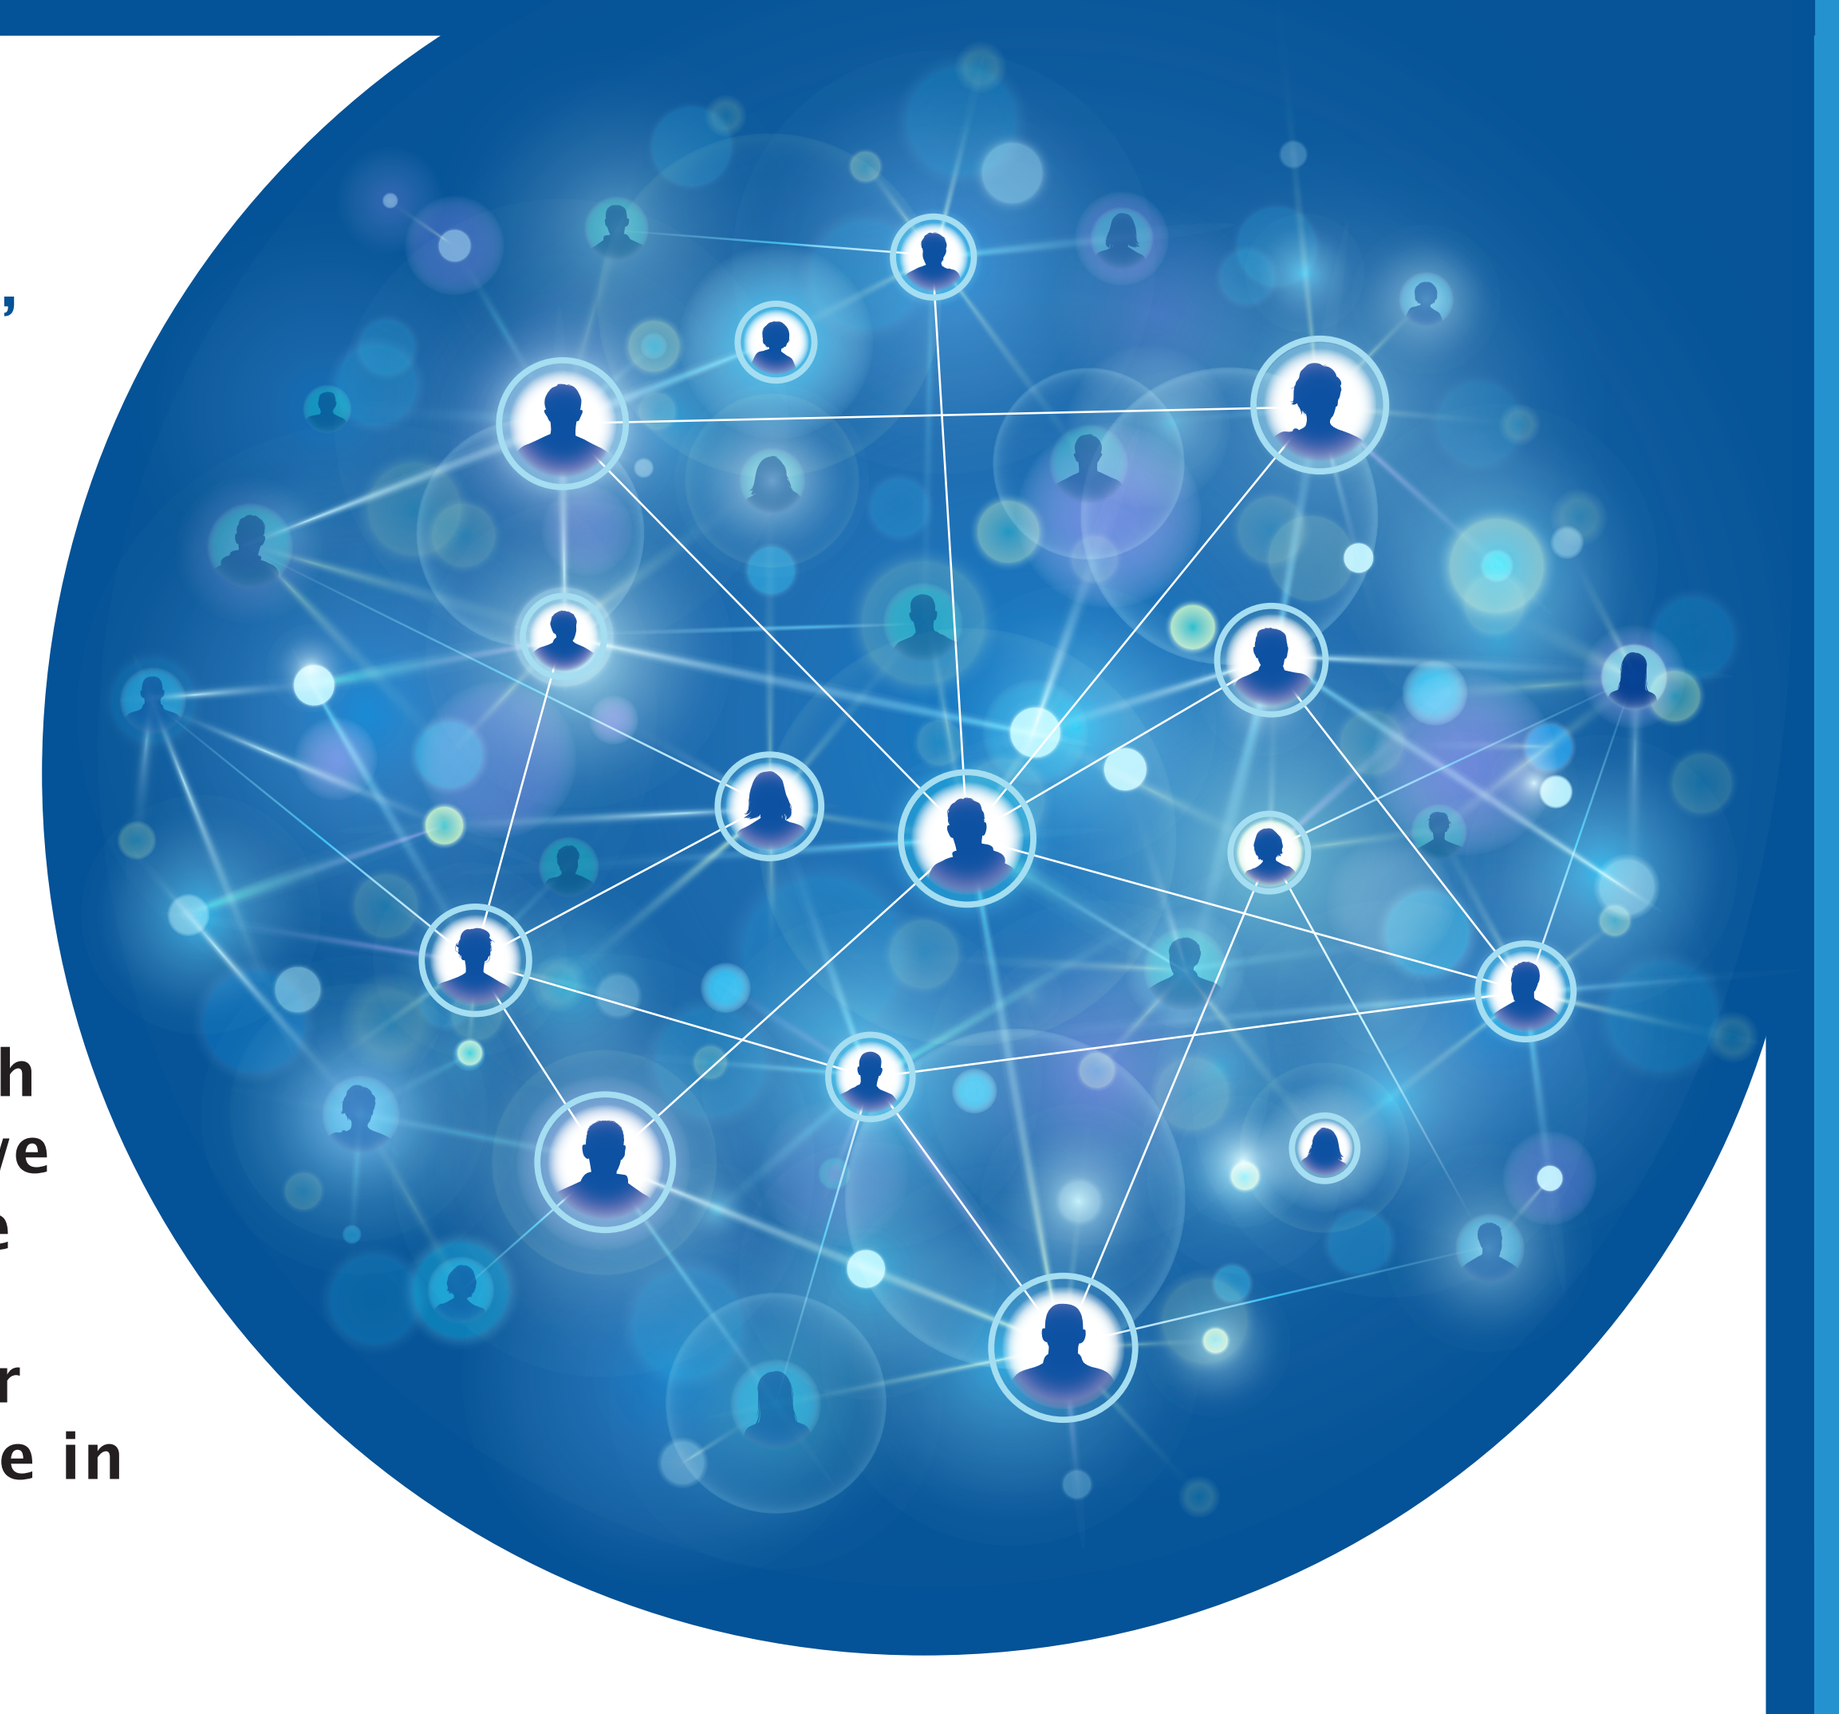

PHYSICIANS SAID REGARDING ESR1 DIAGNOSTIC TESTS:
- highly comfortable ordering them, as long as viable treatment options are available if a mutation is detected
- want to wait until progression of disease before ordering them, with a few physicians suggesting they would test early with metastatic patients to not lose time when the treatment is inevitably needed
- likely to wait for test results rather than treat patients empirically, especially if a liquid biopsy was readily available

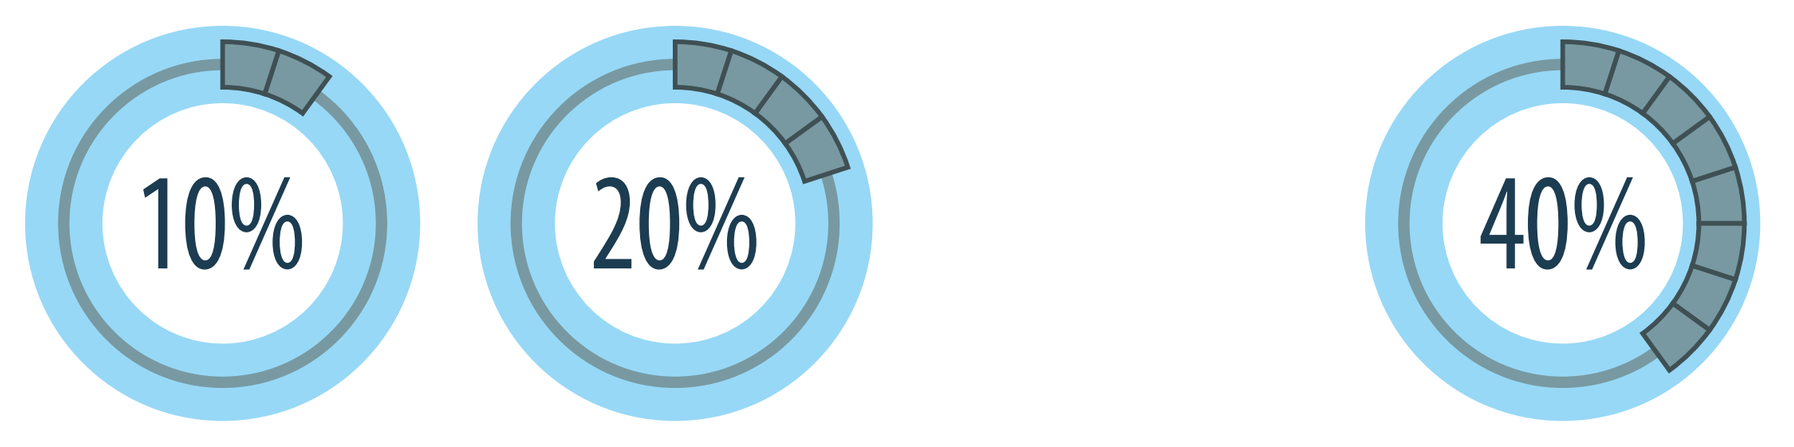
10%
20%
40%

Oncologists think there is a 10-20% ESR1 mutation prevalence

Oncologists accept the possibility that ESR1 mutation prevalence could be as high as 40%

PHYSICIANS WELCOME:
- more efficacious hormonal options for later line therapy
- better durability of remission, with improved progression-free survival and overall survival
- therapies with better tolerability profiles in initial and later line therapy
- lower treatment cost options

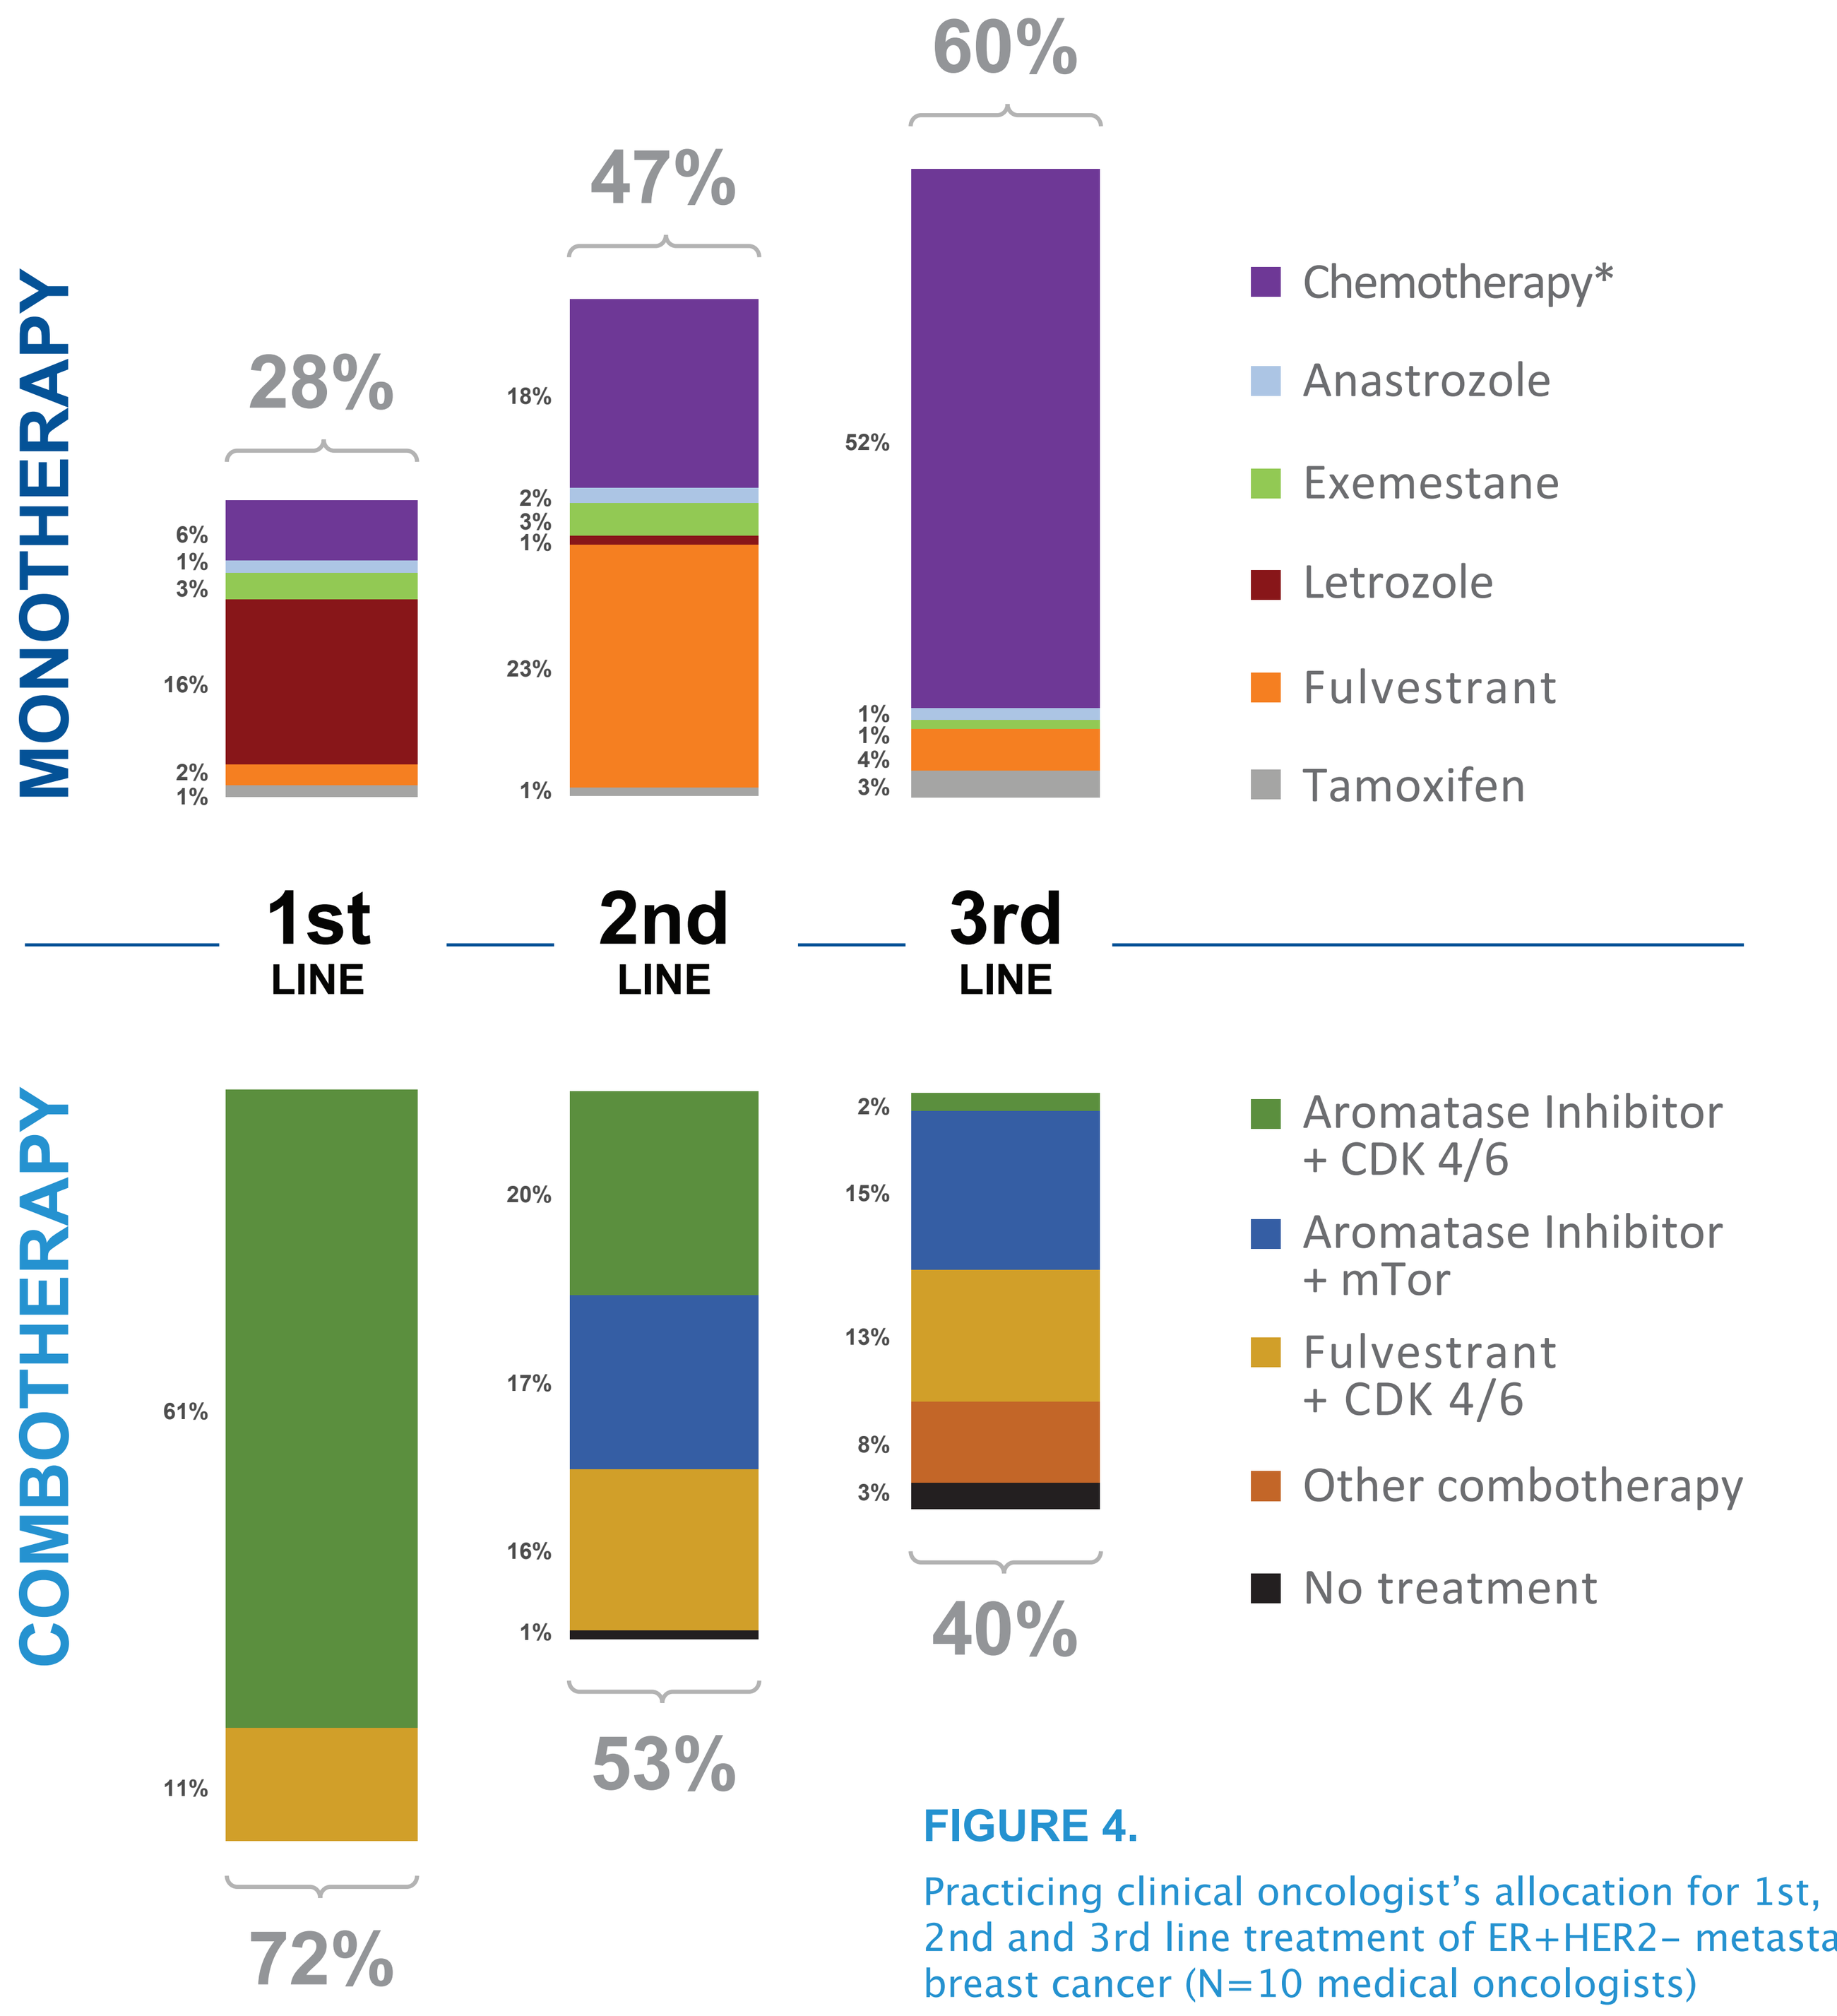
MONOTHERAPY
28%
47%
60%
6%
1%
3%
16%
2%
1%
18%
2%
3%
1%
23%
1%
52%
1%
1%
4%
3%
Chemotherapy*
Anastrozole
Exemestane
Letrozole
Fulvestrant
Tamoxifen
1st LINE
2nd LINE
3rd LINE
COMBOTHERAPY
61%
11%
72%
20%
17%
16%
1%
53%
2%
15%
13%
8%
3%
40%
Aromatase Inhibitor + CDK 4/6
Aromatase Inhibitor + mTor
Fulvestrant + CDK 4/6
Other combotherapy
No treatment

FIGURE 4.

Practicing clinical oncologist's allocation for 1st, 2nd and 3rd line treatment of ER+HER2- metastatic breast cancer (N=10 medical oncologists)

** chemo could be either monotherapy or combotherapy*

CONCLUSIONS

PRACTICING CLINICAL ONCOLOGISTS:

- welcome targeted precision medicine in the ER+HER2- mBC setting
- see no drawbacks to precision medicine from patients, hospital formularies, or payers, especially if a liquid biopsy-based companion diagnostic is available
- believe that detection of mutations and targeting specific tumor genetic signatures will result in improved outcomes
- predict that genetic markers, ESR1 mutations, and diagnostic tests will increase in the future
- welcome the development of new endocrine therapies with documented efficacy, improved tolerability, dosing, and quality of life profiles

This presentation is the intellectual property of the author/presenter. Contact David Portman, Founder and CEO, at DPortman@sermonixpharma.com for permission to reprint and/or distribute.

REFERENCES

1. Jesselsohn, R. Are We Ready to Use ESR1 Mutations in Clinical Practice? Breast Care, 2017;12:309-313.
2. Kuang Y et al. Unraveling the Clinicopathological Features Driving the Emergence of ESR1 Mutations in Metastatic Breast Cancer, Nature Partner Journals (NPJ) Breast Cancer, 2018;22:1-10.
3. Angus L et al. Cancer Treatment Reviews, 2017;52:33-40.
4. Toy W, Shen Y, Woo H, et al. Nat Genet. Dec 2013;45(12):1439-1445.
5. Medscape Oncology @MedscapeOne twitter feed, Oct 2018; presented at the European Society for Medical Oncology (ESMO).